Identifying Targets to Inhibit Dystrophic Adipogenic Differentiation of Fibro-Adipogenic Progenitors via a Small Molecule Screen

Aidan Bailey, Genevieve Wilson, Richard Lieberman, Johnny Lucas, Joseph Maglio, Erin Valentine, Anthony Accorsi
Fulcrum Therapeutics, Inc. 26 Landsdowne Street, Cambridge, Massachusetts, USA.

Abstract

- Gradual replacement of contractile myofibers with non-contractile fibro-fatty infiltration is a hallmark of muscular dystrophies and other chronic myopathies
- Muscle-resident fibro-adipogenic progenitors (FAPs) are directly implicated in the development of these infiltrates due to their ability to differentiate into fibroblasts and adipocytes
- Although transient fibrosis is necessary to some extent for muscle repair, fatty infiltration only impedes muscle function and health
- Targeting pathological adipogenic differentiation of FAPs represents an opportunity to slow the course of disease progression for those living with muscular dystrophies
- No therapies exist yet to treat this major detriment to muscle function and quality of life

Objective:

- Develop a scalable and robust cellular assay that enables a small molecule screen to identify potential targets to prevent or reverse dystrophic fatty infiltration

1. Fatty infiltration in muscular dystrophies and the role of FAPs

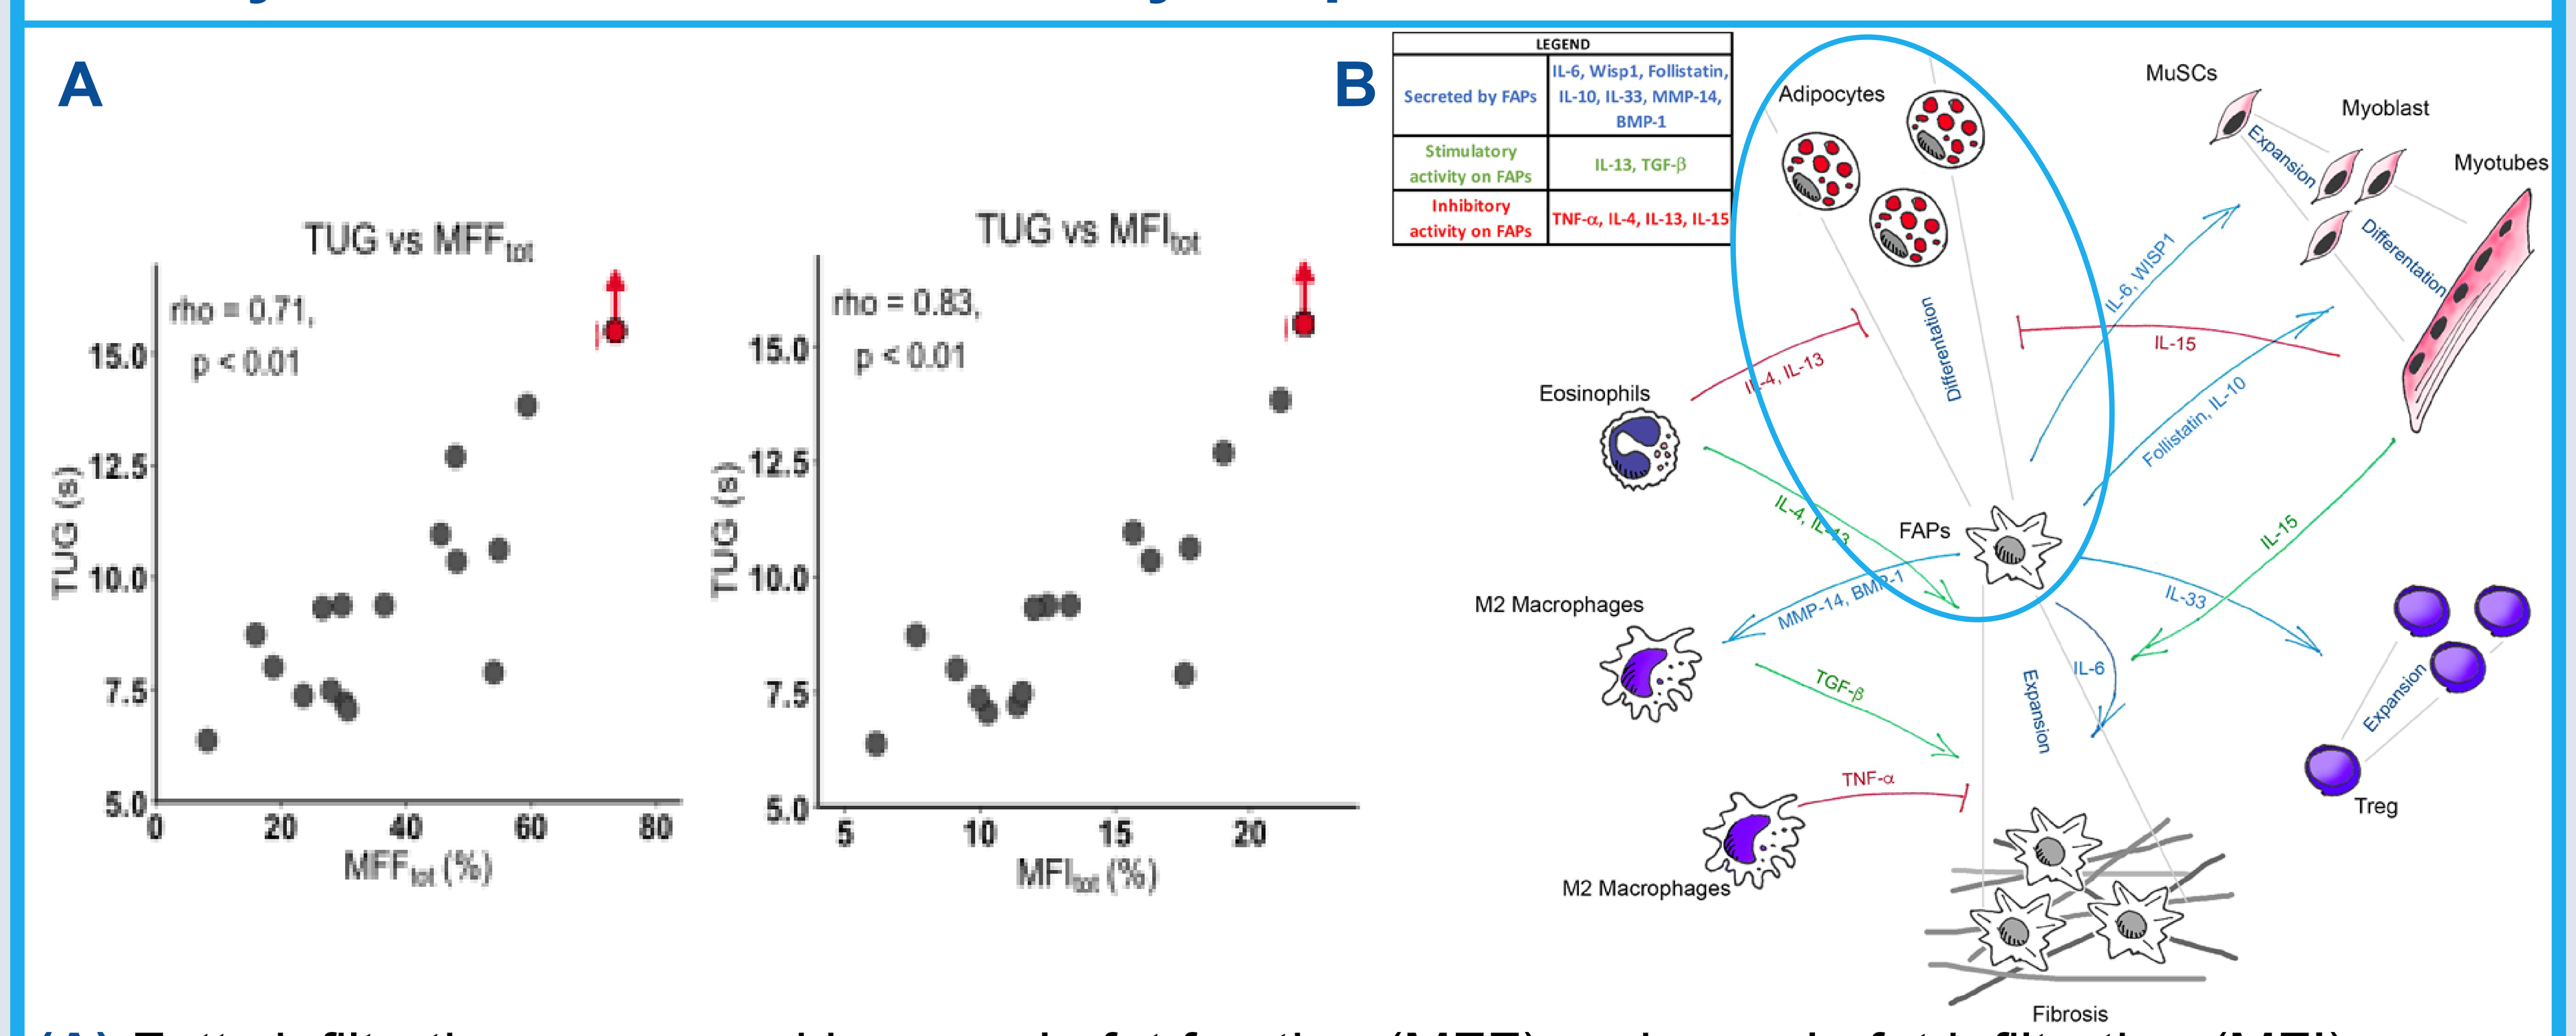
A
TUG vs MFFtot
rho = 0.71,
p < 0.01
TUG (s)
MFFtot (%)
TUG vs MFItot
rho = 0.83,
p < 0.01
MFItot (%)
B
LEGEND
Secreted by FAPs
IL-6, Wisp1, Follistatin, IL-10, IL-33, MMP-14, BMP-1
Stimulatory activity on FAPs
IL-13, TGF-β
Inhibitory activity on FAPs
TNF-α, IL-4, IL-13, IL-15
Adipocytes
MuSCs
Myoblast
Myotubes
Expansion
Differentiation
Eosinophils
FAPs
M2 Macrophages
Treg
Fibrosis

(A) Fatty infiltration, measured by muscle fat fraction (MFF) and muscle fat infiltration (MFI), strongly correlates with impairment of functional outcomes in FSHD, for example. TUG = timed up and go.
(B) Pathological FAP expansion and adipogenic differentiation in the dystrophic microenvironment represents a critical node for therapeutic targets.[1]

2. Fulcrum's approach to targeting dystrophic FAP adipogenesis

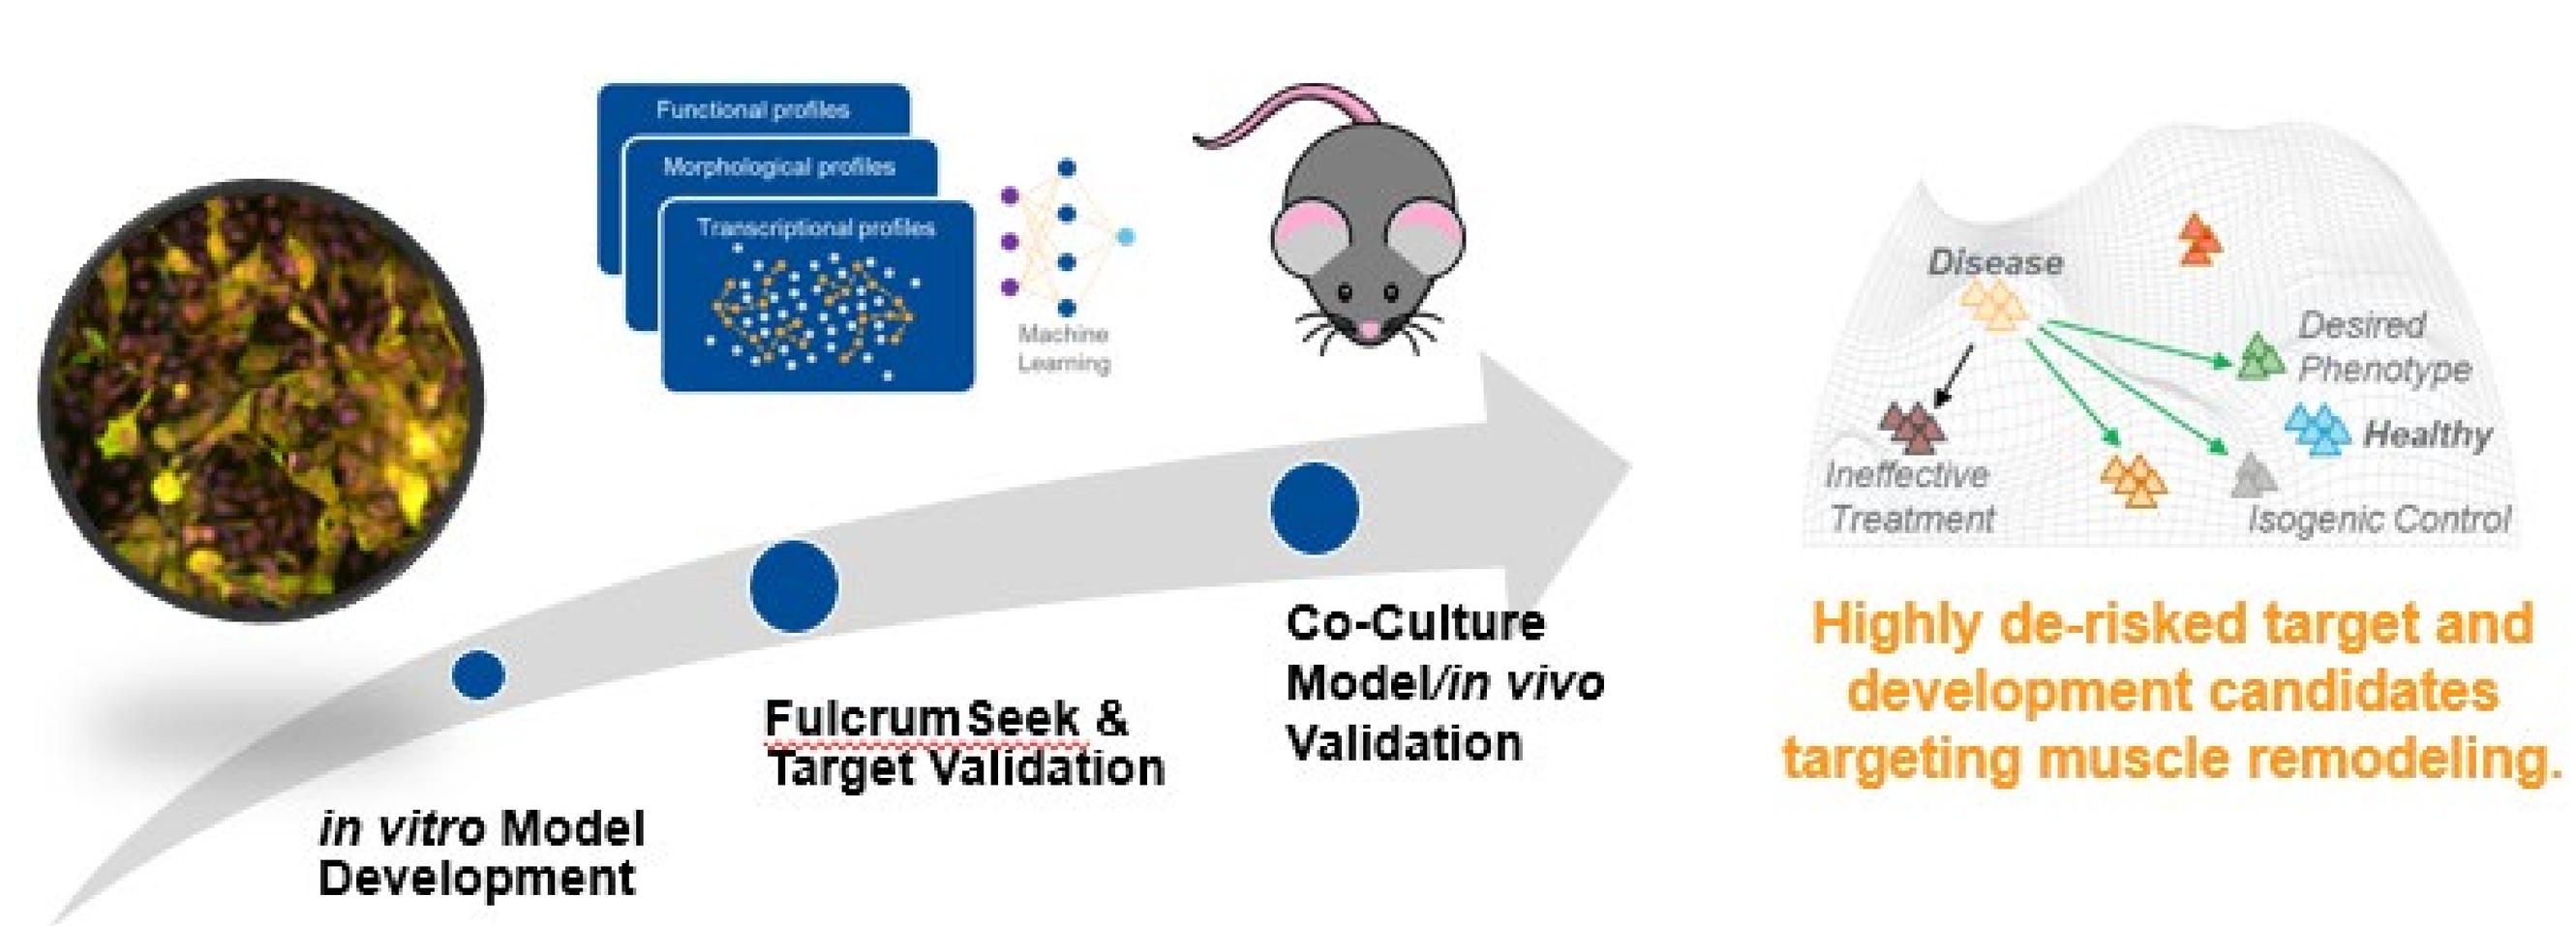
Functional profiles
Morphological profiles
Transcriptional profiles
Machine Learning
in vitro Model Development
FulcrumSeek & Target Validation
Co-Culture Model/in vivo Validation
Disease
Desired Phenotype
Healthy
Ineffective Treatment
Isogenic Control
Highly de-risked target and development candidates targeting muscle remodeling.

Developing a robust and reproducible cellular assay for adipogenic differentiation is the first step in enabling a high-throughput screen for identifying druggable targets. Here, we develop an assay using adipose-derived mesenchymal stem cells (A-MSCs), harnessing their scalability, similarities to FAPs in gene expression and multipotency, and ability to be passaged without losing these features.

3. Differentiation paradigm and optimization

A. Optimized timeline for differentiation and screening

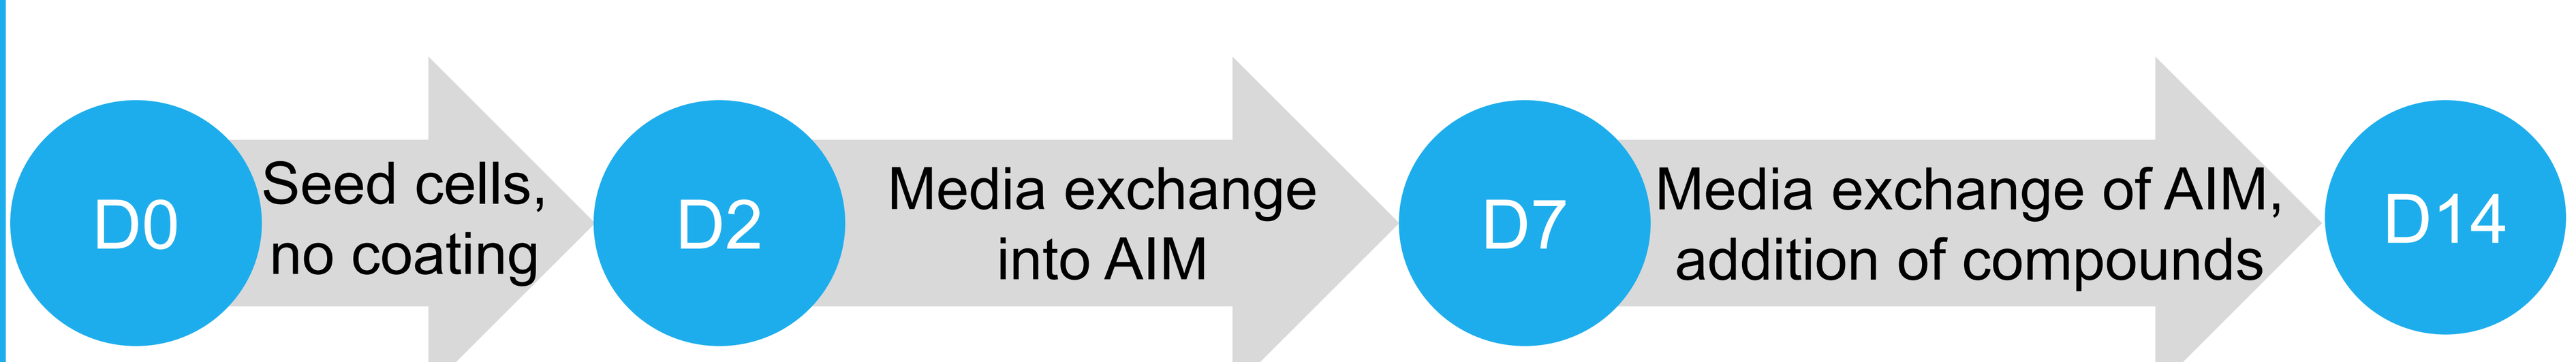
D0
Seed cells, no coating
D2
Media exchange into AIM
D7
Media exchange of AIM, addition of compounds
D14

B. Selection of imaging panel to visualize adipogenic differentiation

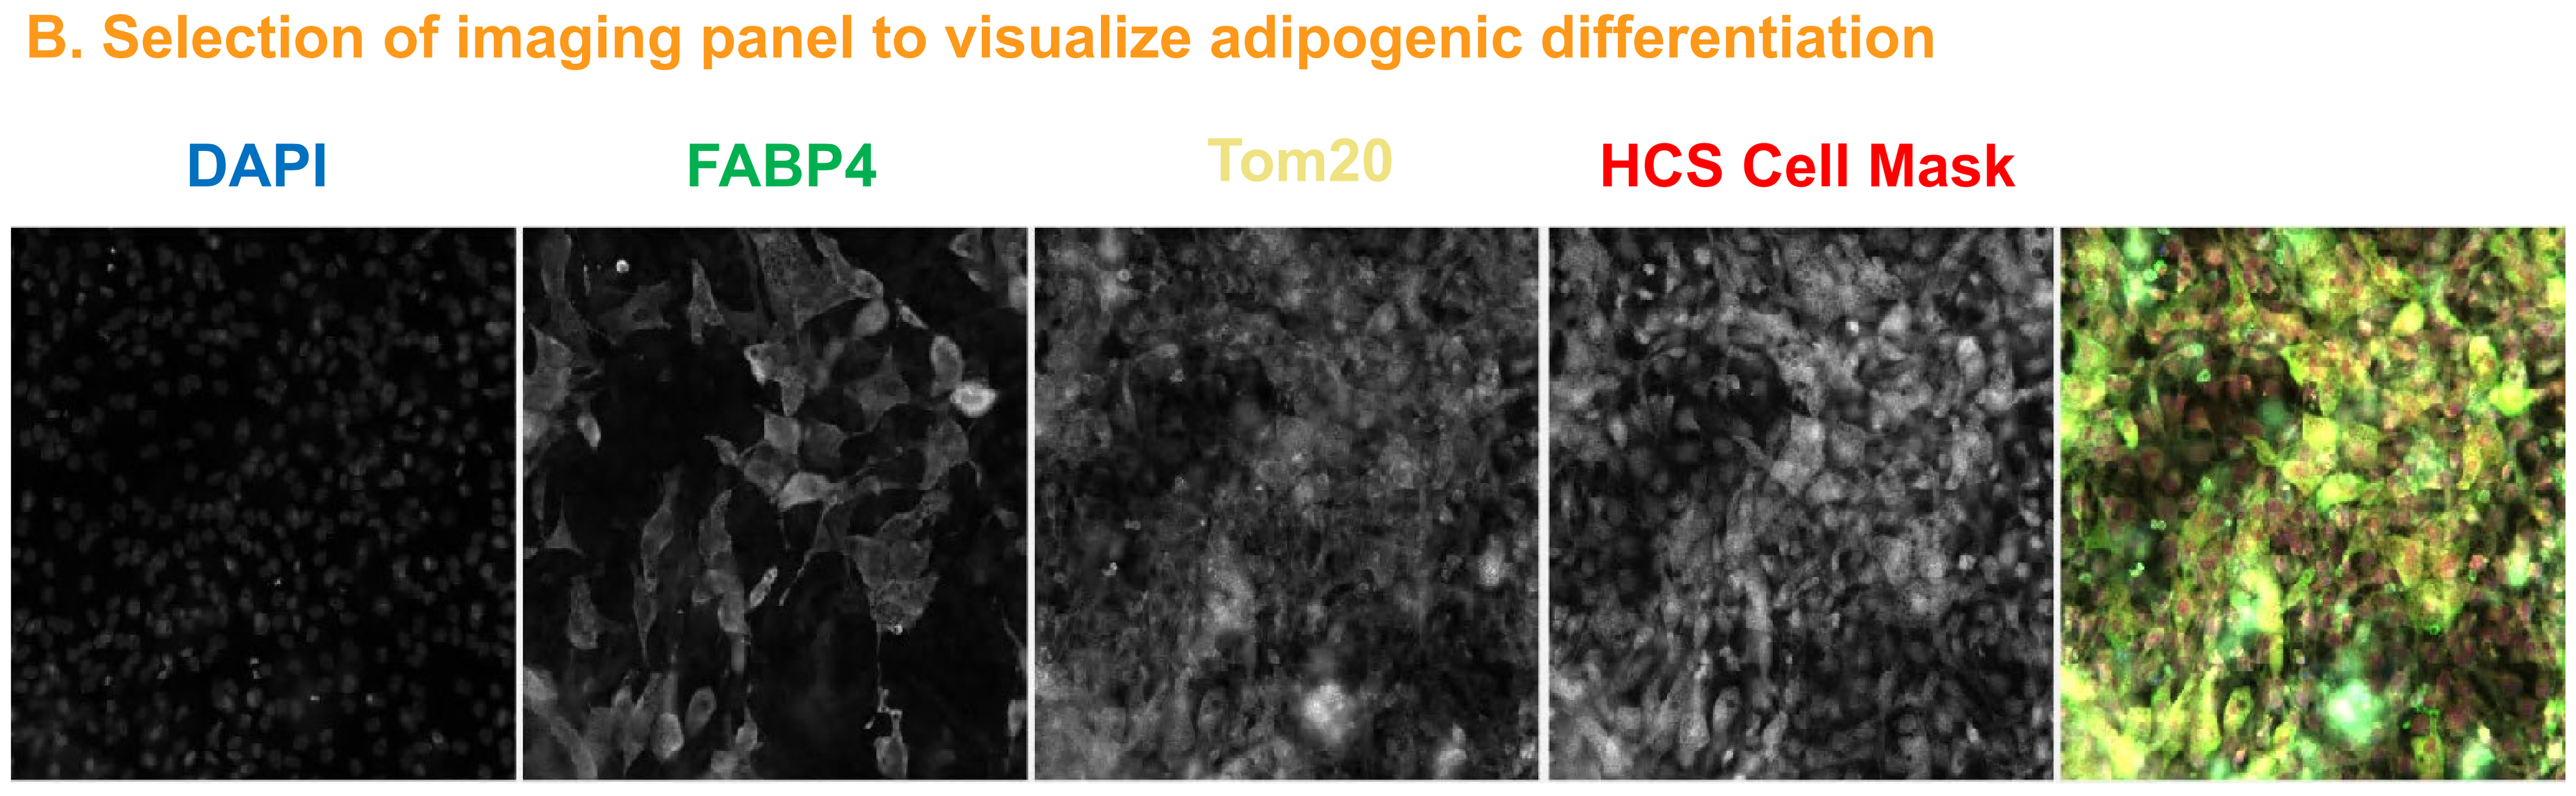
DAPI
FABP4
Tom20
HCS Cell Mask

(A) Optimized timeline for differentiation of A-MSCs into adipocytes, in light of screening parameters. On Day 14 of differentiation, cells are fixed or lysed for downstream assays. AIM = adipogenic induction media.

(B) Selection of an imaging panel for eventual high-content imaging analysis. DAPI identifies cell nuclei, fatty acid binding protein 4 (FABP4) is an adipocyte-specific marker, Tom20 marks mitochondria for cell agnostic feature extraction, and HCS Cell Mask is a morphological marker. Images taken at 20x magnification.

4. FAPs vs. A-MSCs Characterization

A. Gene expression profiling of FAPs vs. A-MSCs undergoing adipogenic differentiation

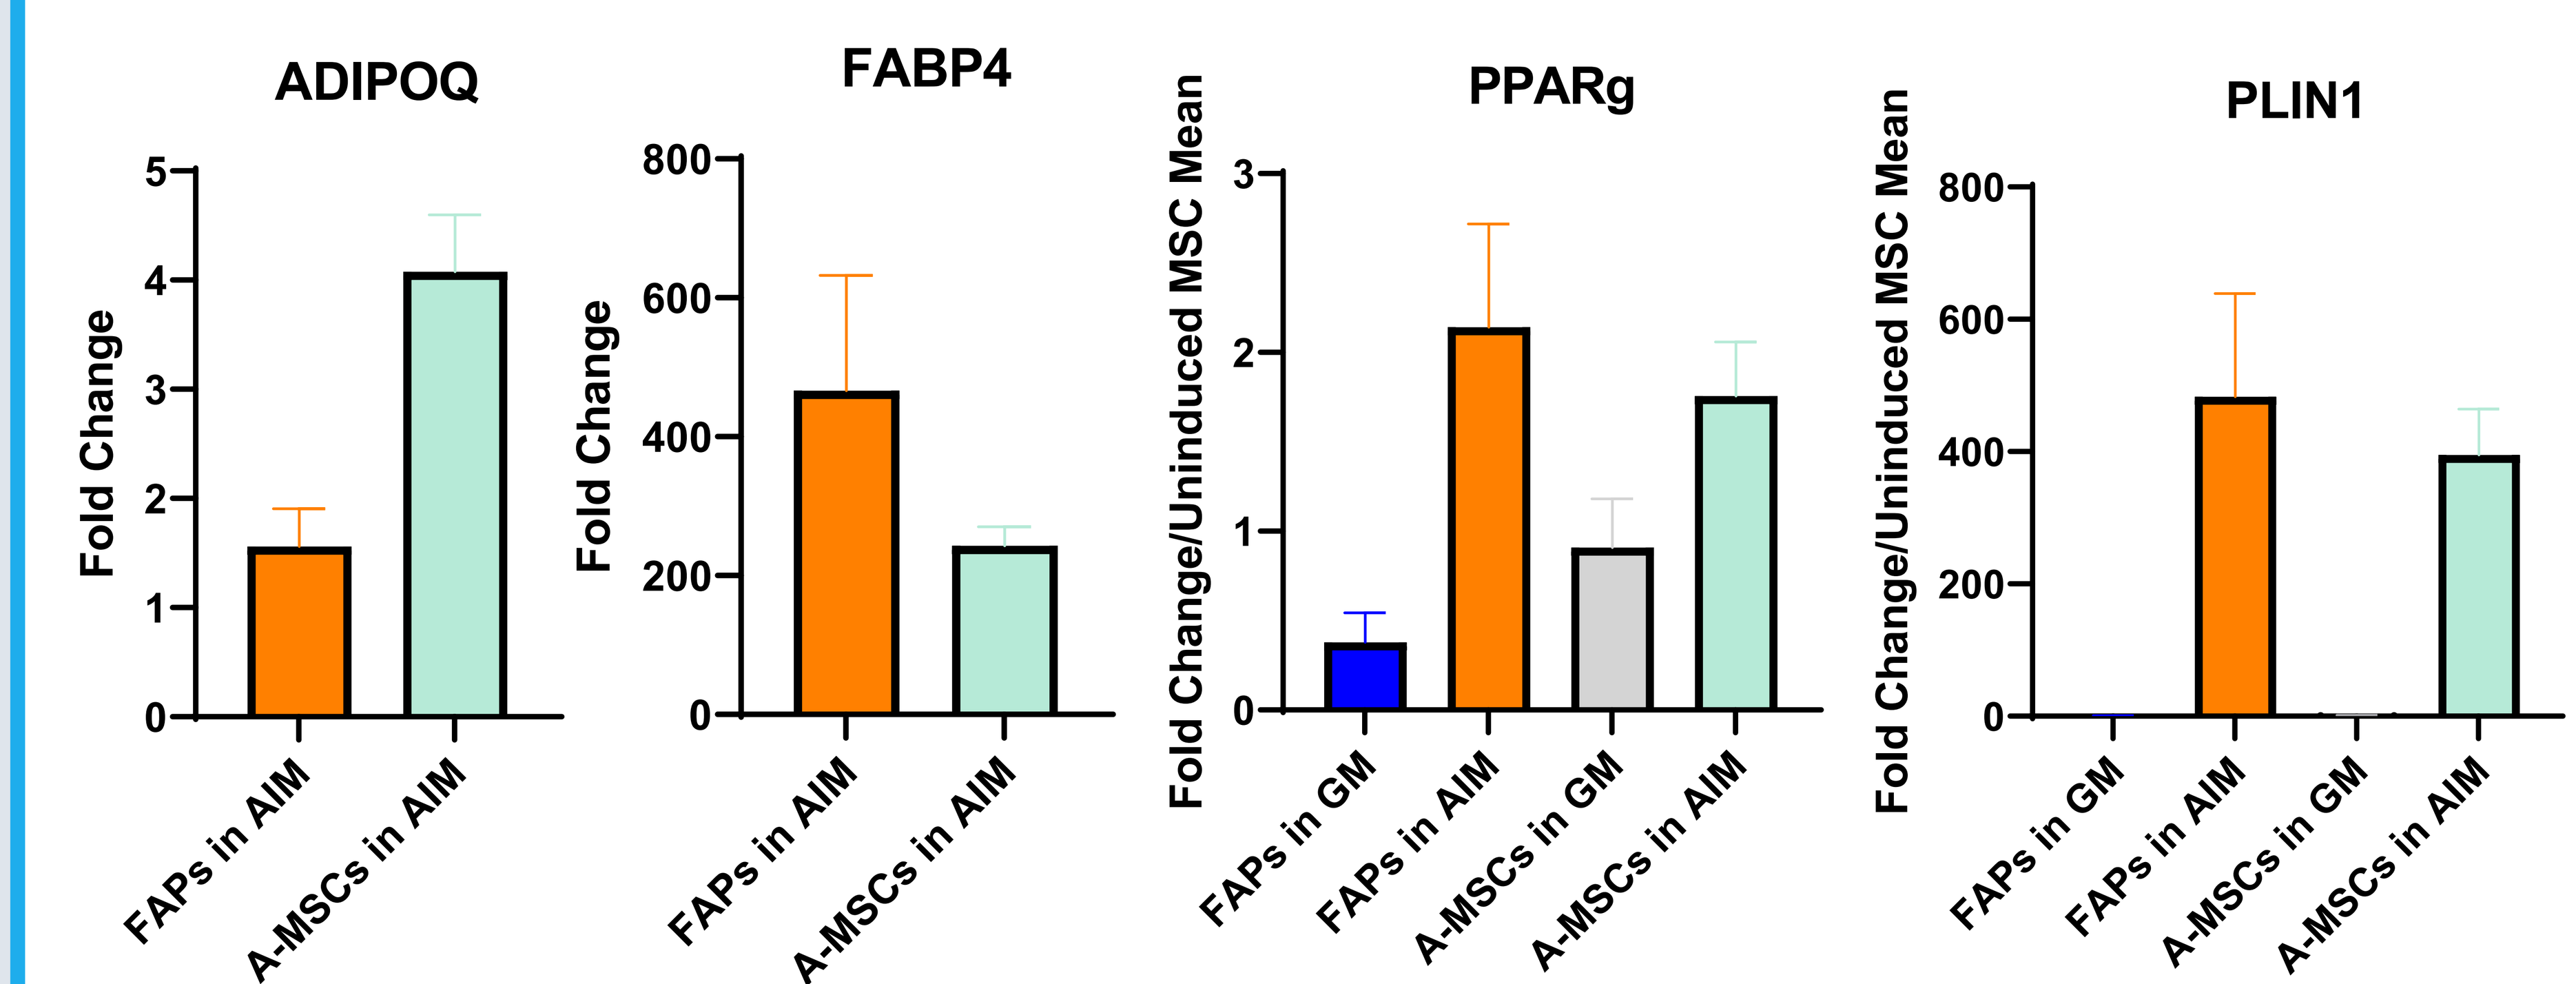
ADIPOQ
Fold Change
FAPs in AIM
A-MSCs in AIM
FABP4
PPARg
Fold Change/Uninduced MSC Mean
FAPs in GM
A-MSCs in GM
PLIN1

B. ICC comparison of FAPs vs. A-MSCs after adipogenic differentiation

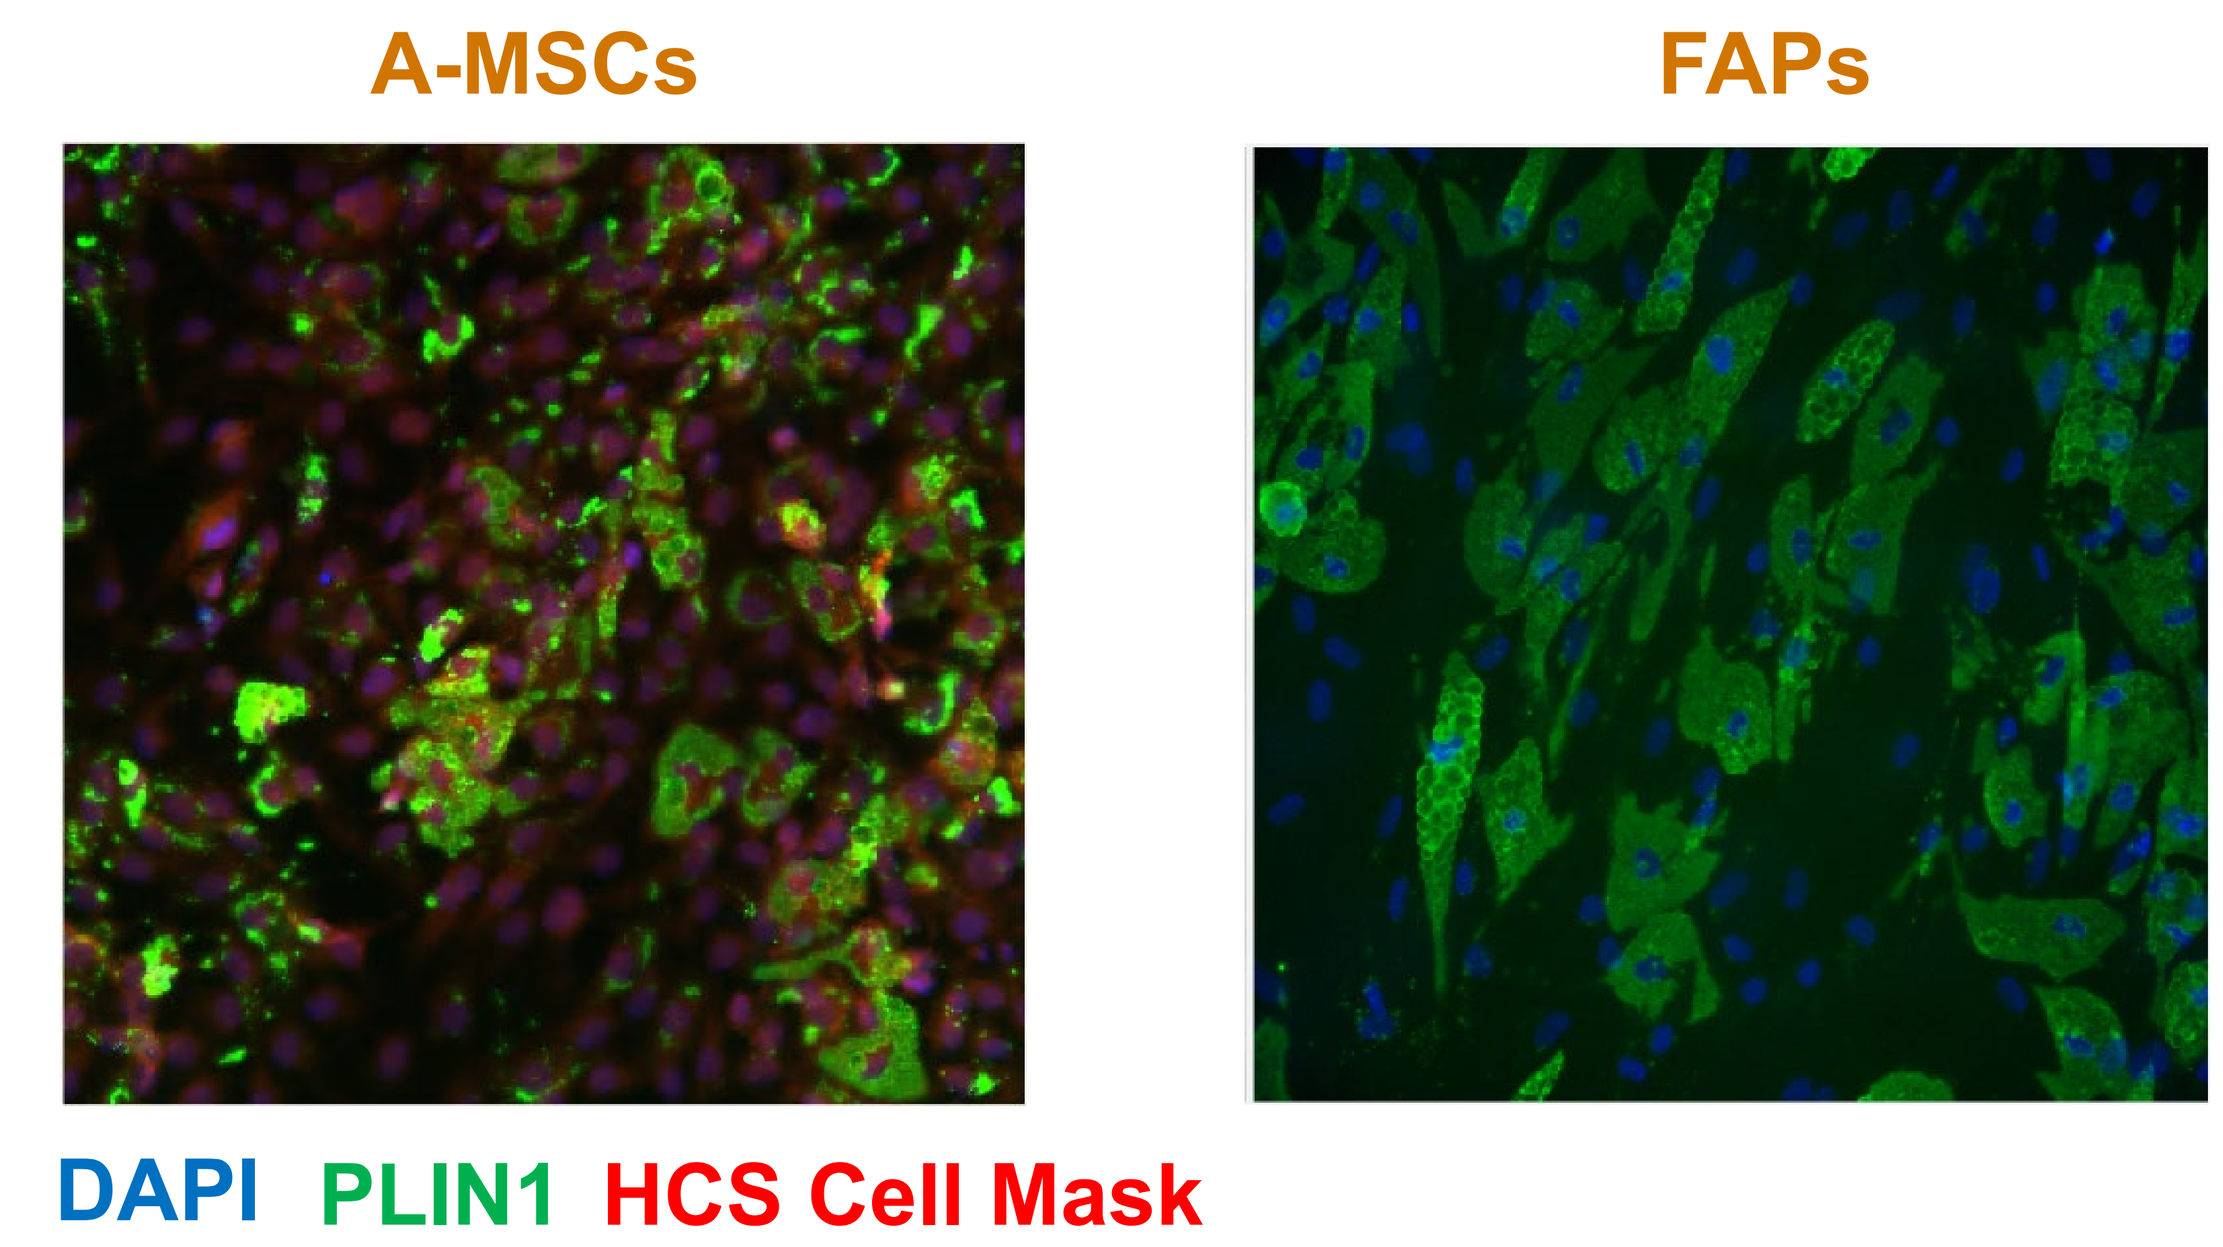
A-MSCs
FAPs
DAPI
PLIN1
HCS Cell Mask

(A) Gene expression profiling of FAPs and A-MSCs after 14 days of adipogenic differentiation. Using qPCR to measure transcript production, we show that for four adipogenic markers, A-MSCs experience similar changes in gene expression to FAPs, justifying their use as a proxy cell type for screening purposes. ADIPOQ = adiponectin, FABP4 = fatty acid binding protein 4, PPARg = peroxisome proliferator-activated receptor gamma, PLIN1 = perilipin-1.
(B) ICC comparison of FAPs and A-MSCs after 14 days of adipogenic differentiation. Using PLIN1 as a basic marker of adipocytes, these representative images show that A-MSCs stain positive in a similar manner to FAPs, further justifying their use as a proxy cell type for screening purposes.

5. Endpoint development: Imaging classification

Imaging classifier tool to quantify adipogenic response

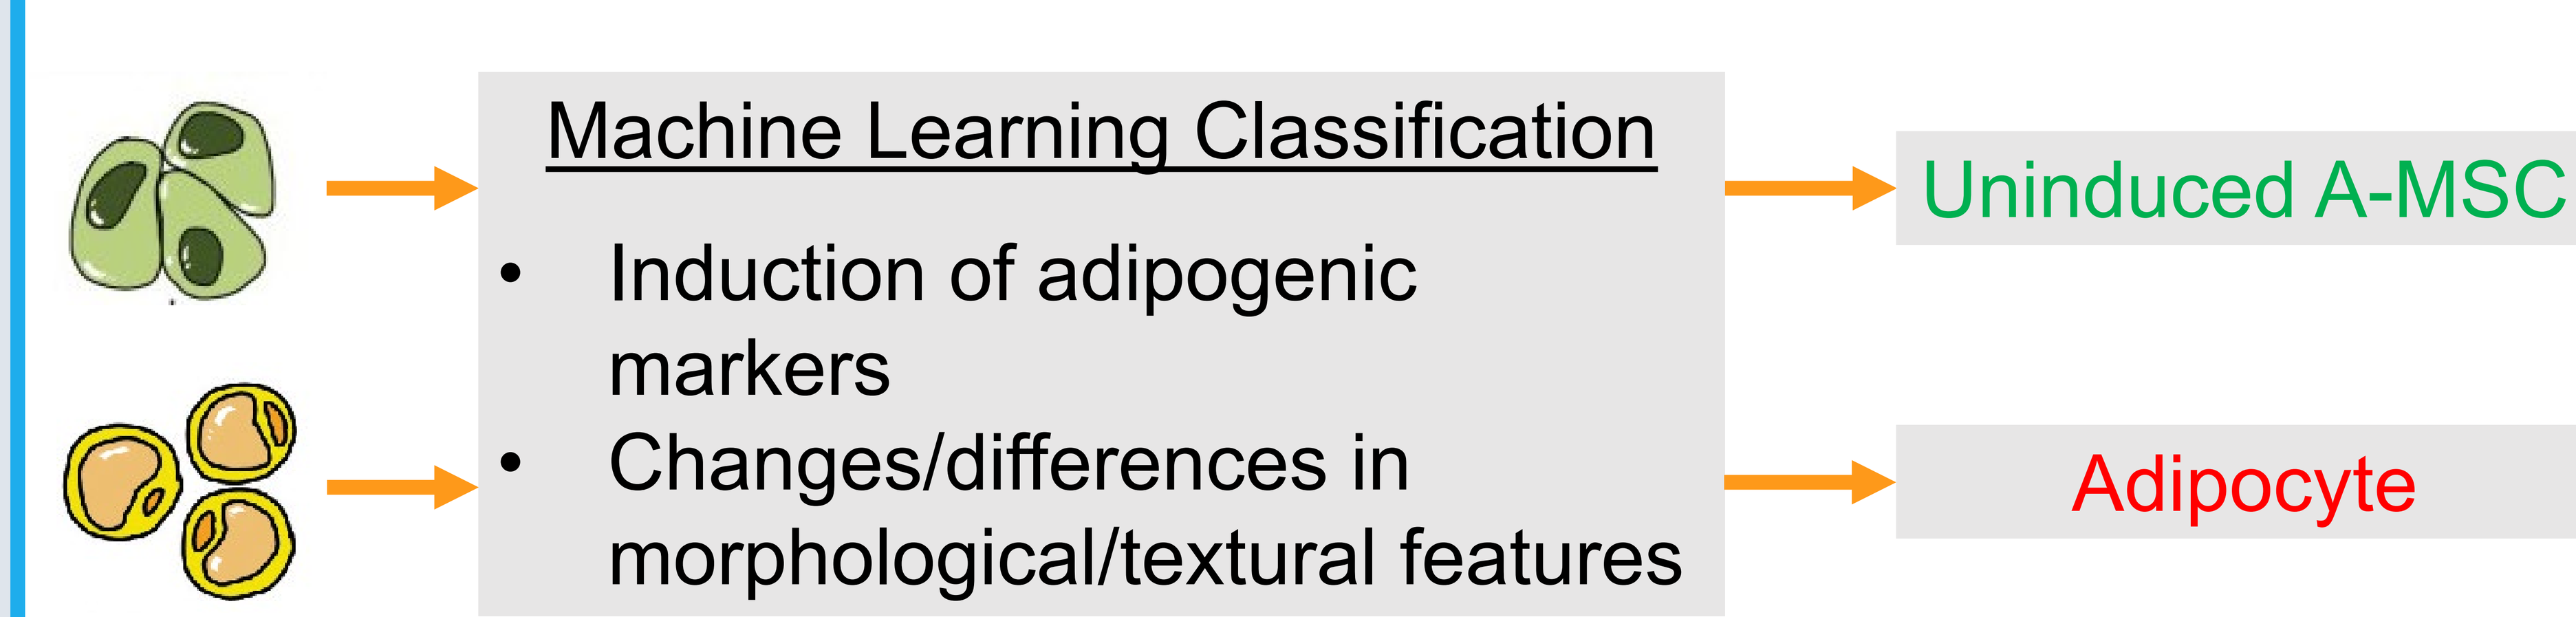
Machine Learning Classification
Induction of adipogenic markers
Changes/differences in morphological/textural features
Uninduced A-MSC
Adipocyte

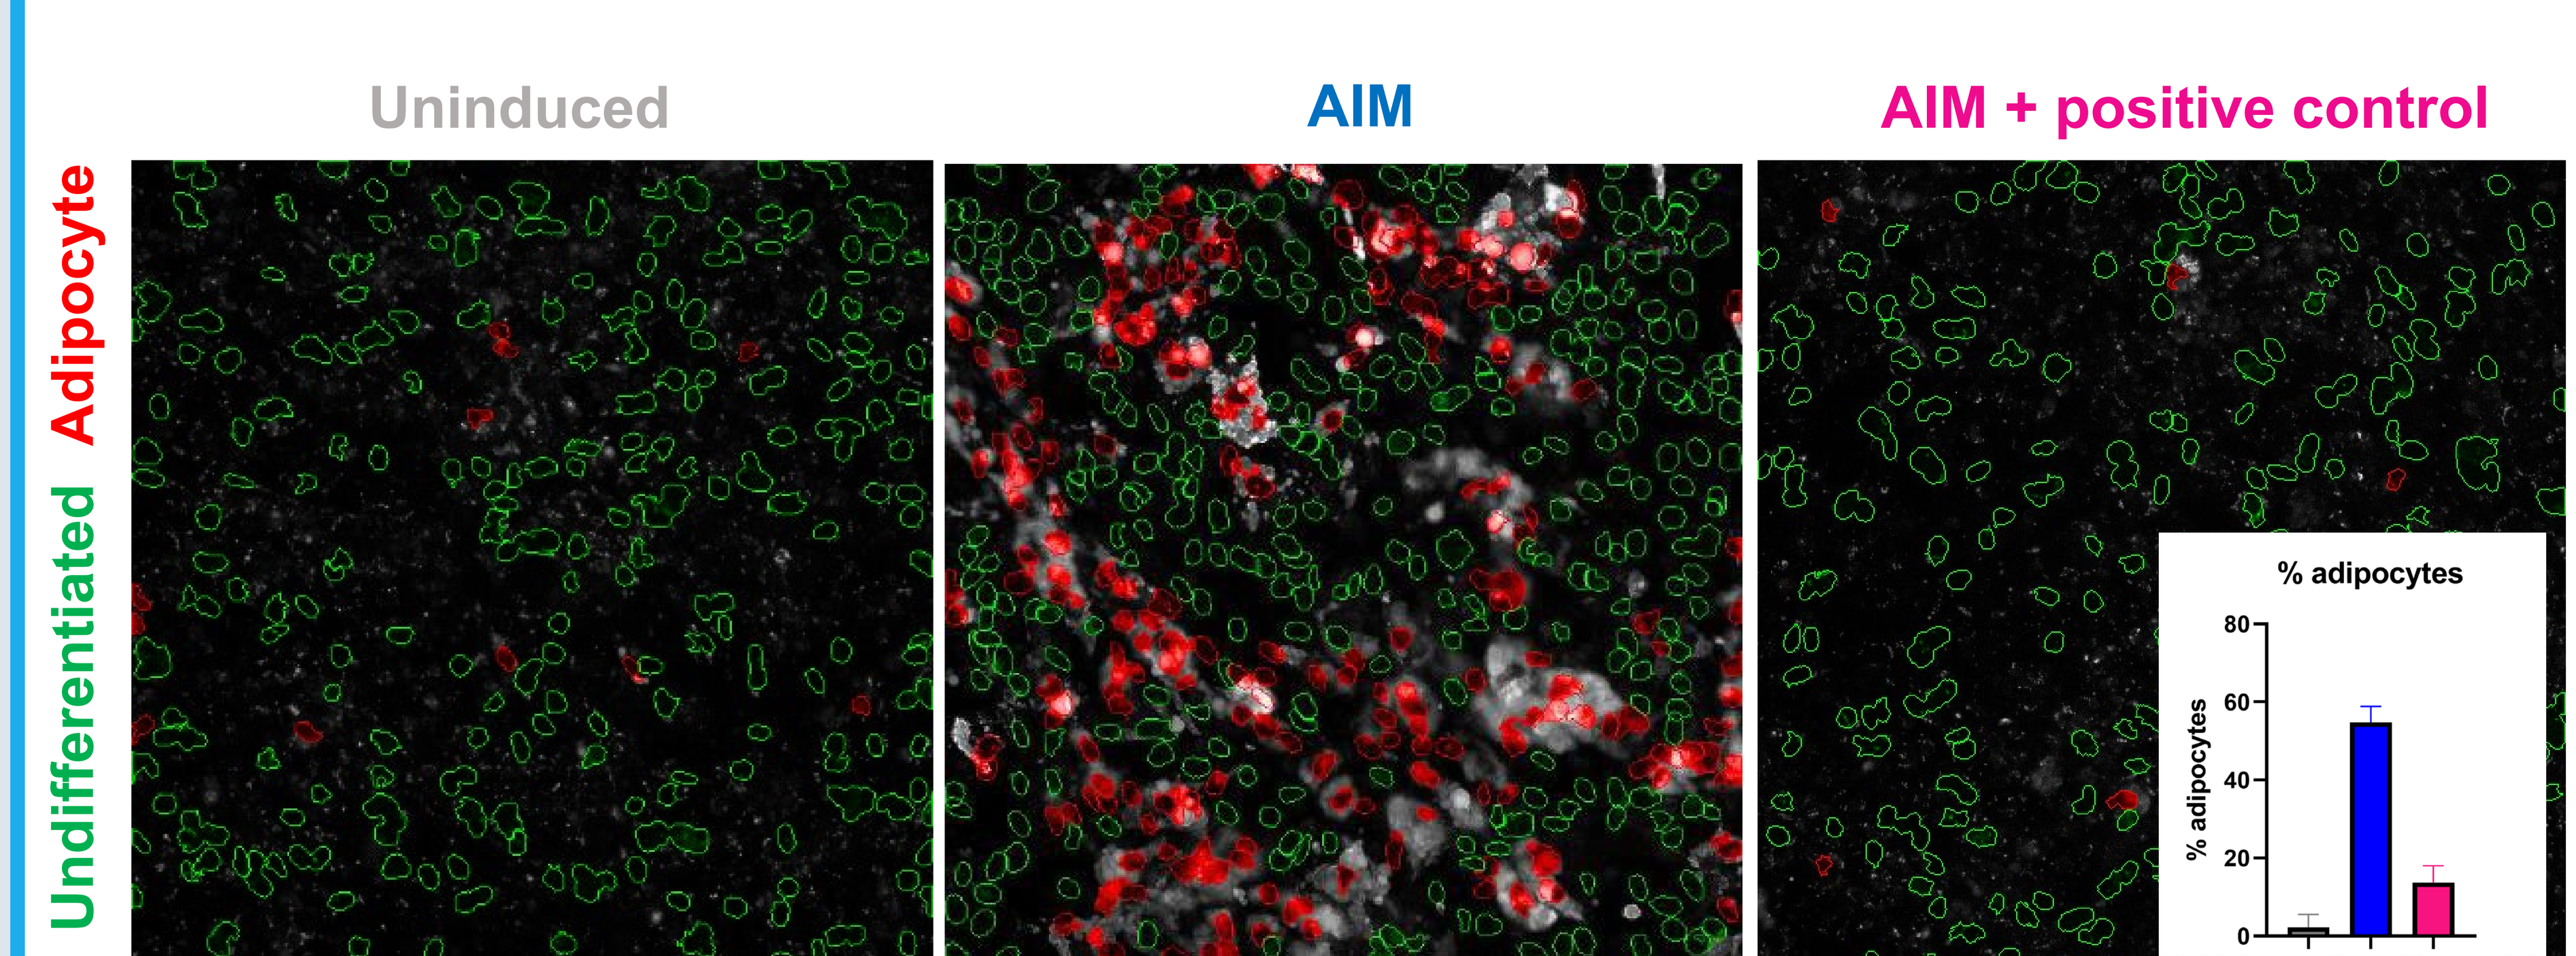
Uninduced
AIM
AIM + positive control
Undifferentiated
Adipocyte
% adipocytes

We developed a linear classifier that can be trained to distinguish between undifferentiated cells and adipocytes using images from our ICC analysis. The readout quantifies what is observed in each of the different conditions.

6. Next Steps

Next, we plan to execute a small molecule probe screen to identify modulators of adipogenic cell fate after further scaling up activities, including adaptation of automation protocols and pressure-testing assay validity and reproducibility.

References

(1) Biferali et al. Fibro-adipogenic progenitors cross-talk in skeletal muscle: The social network. *Front. Physiol.* 10:1074 (2019).