Design of a biomarker of DUX4 activity to evaluate losmapimod treatment effect in FSHD Phase 2 trials

Fulcrum Therapeutics

Lucienne Ronco[1], Diego Cadavid[1], Jayashree Chadchankar[1] Aaron Chang[1], Michelle Mellion[1], Alan Robertson[1],, Alejandro Rojas[1], Ning Shen[1], Rabi Tawil[2], Stephen Tapscott[3], Leo Wang[4], Owen Wallace[1]

[1]Fulcrum Therapeutics. 26 Landsdowne Street, Cambridge, Massachusetts, USA. [2]Neuromuscular Unit, Department of Neurology, University of Rochester Medical Center, Rochester, NY, USA
[3]Human Biology Division, Fred Hutchinson Research Center, Seattle, Washington, USA [4]Department of Neurology, University of Washington, Seattle, Washington, USA

Abstract

Both genetic types of facioscapulohumeral muscular dystrophy (FSHD) are caused by the aberrant expression of the *homeobox transcription factor* DUX4. Ectopic DUX4 expression in skeletal muscle is the root cause of FSHD. Aberrant DUX4 expression in maturing myofibers activates a complex transcriptional program that leads to myofiber death and the replacement of muscle with fat. DUX4 expression results in profound transcriptional dysregulation and a characteristic DUX4-regulated signature has been described by many laboratories. Clinical evaluation of the p38α/β inhibitor losmapimod is ongoing including the assessment of drug-exposure, target engagement and inhibition of the DUX4 transcriptional program in muscle biopsies. Reliable detection of DUX4 protein and mRNA in affected FSHD skeletal muscle biopsies is challenging as the levels are very low and likely short lived. Conversely, levels of DUX4-regulated transcripts are much higher and longer lasting. Here we used RNA-seq profiling of repeated FSHD skeletal muscle biopsies to help design a biomarker of DUX4 activity to evaluate losmapimod treatment effect in FSHD trials.

Objectives:

- Identify DUX4-regulated gene transcripts to measure DUX4 activity in skeletal muscle needle biopsies in preparation for ongoing FSHD clinical trials with losmapimod (Ph1 study FIS 001-2018, placebo-controlled ReDUX4 phase 2b study FIS 002-2019 and phase 2 open label study FIS 001-2019).
- Assess the longitudinal variability under natural history conditions of DUX4-regulated gene transcripts in MRI-affected skeletal muscle needle biopsy samples from FSHD patients with moderate disability (Ricci score 2-4).
- Identify a subset of these transcripts that exhibit higher expression and lower variability in repeated biopsies.

1. Background

A.

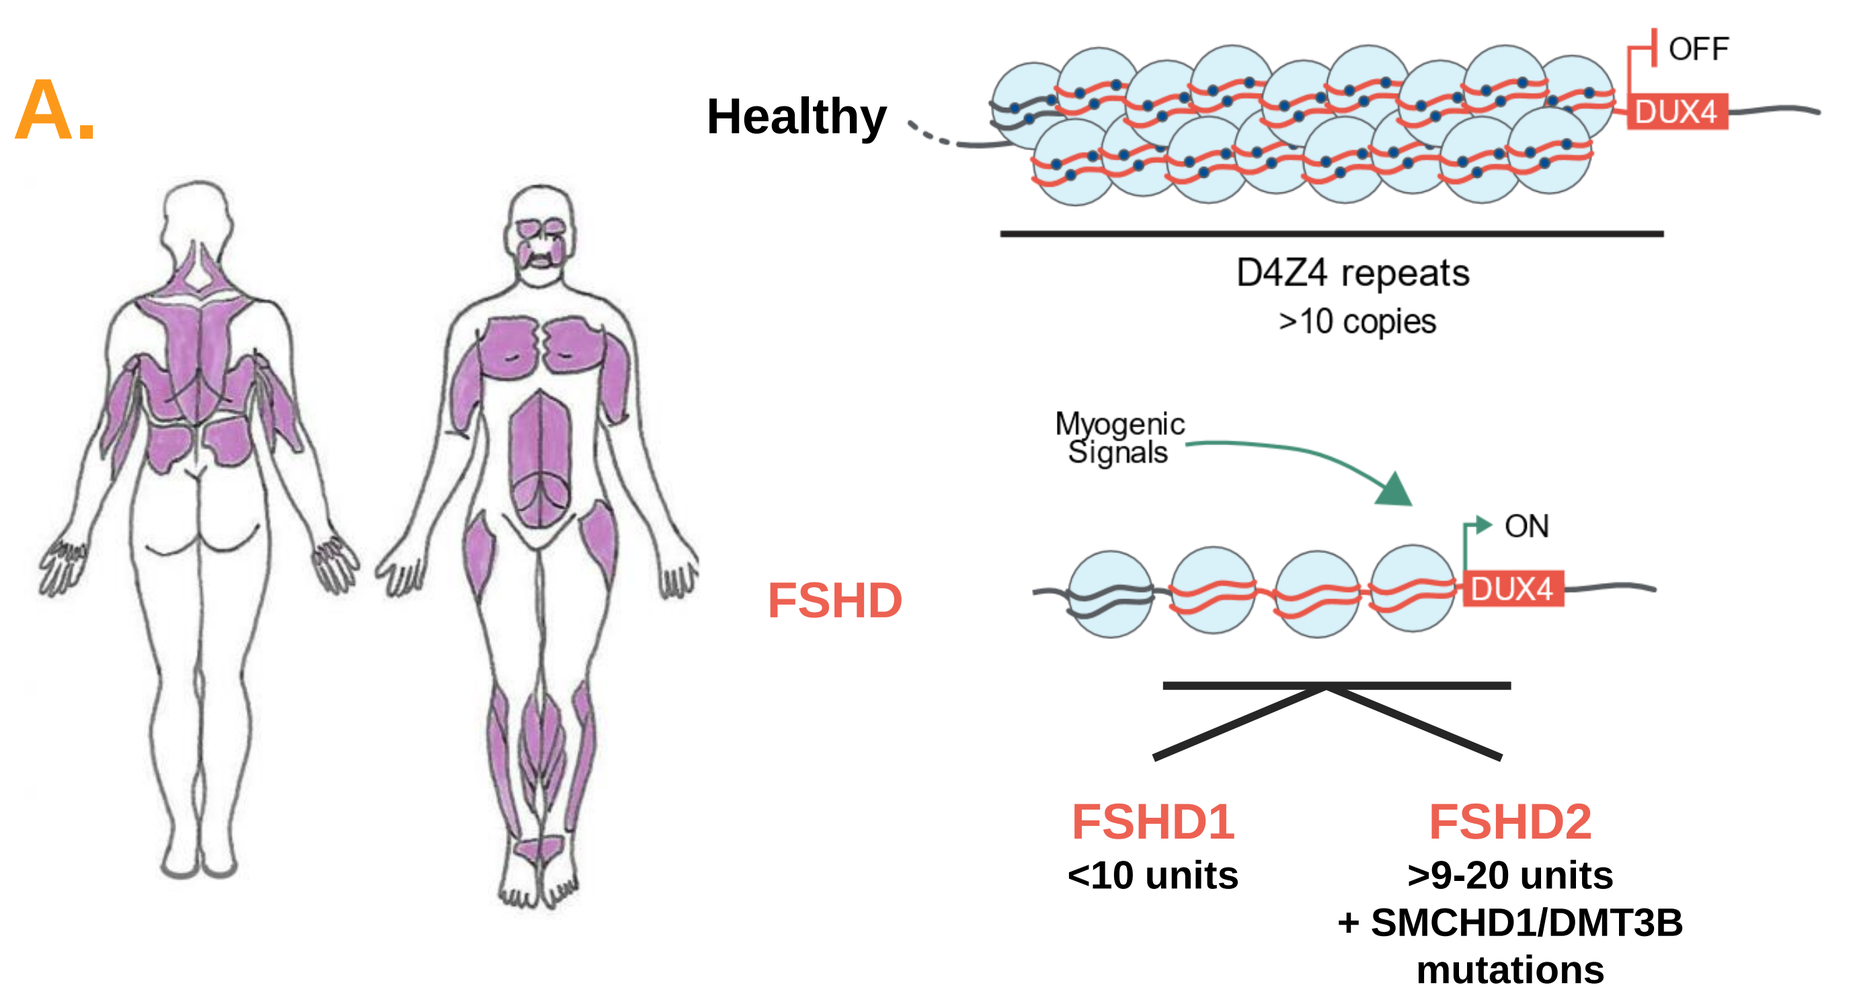
Healthy
OFF
DUX4
D4Z4 repeats
>10 copies
Myogenic Signals
ON
DUX4
FSHD
FSHD1
<10 units
FSHD2
>9-20 units
+ SMCHD1/DMT3B mutations

B.

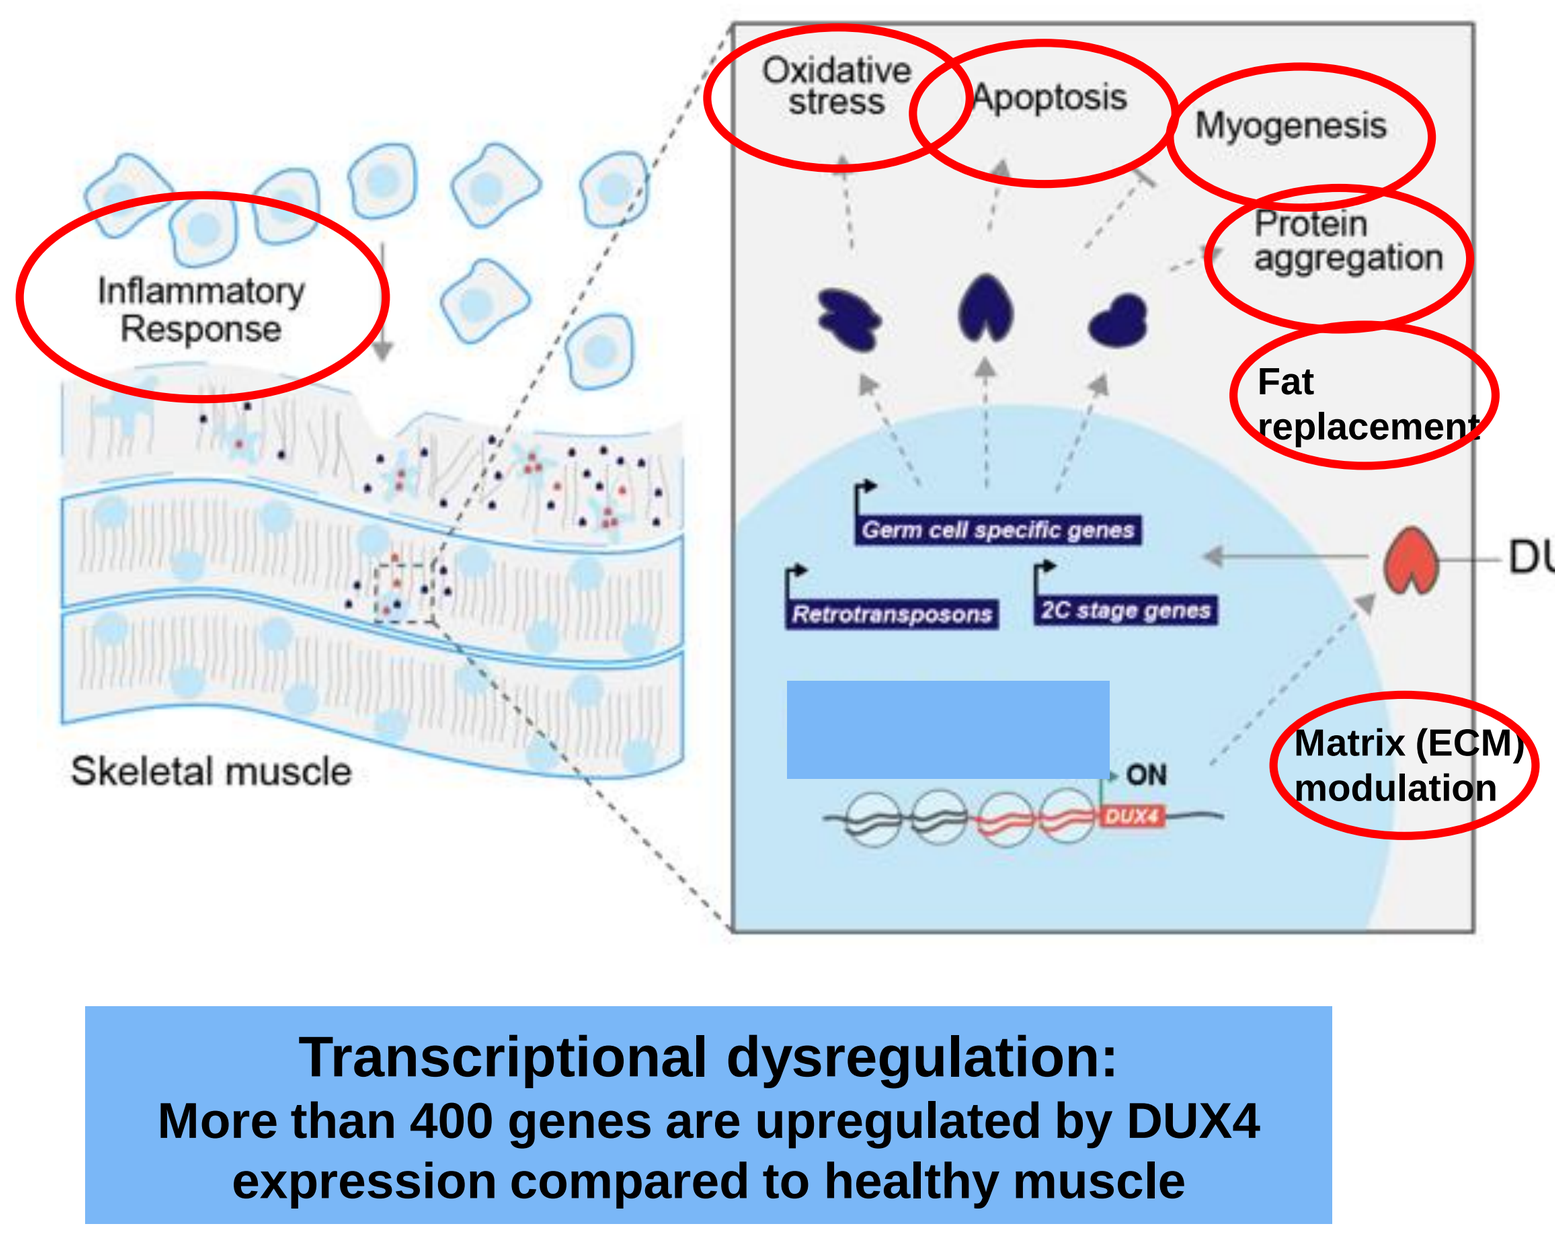
Oxidative stress
Apoptosis
Myogenesis
Protein aggregation
Inflammatory Response
Fat replacement
Germ cell specific genes
Retrotransposons
2C stage genes
DUX4
Skeletal muscle
ON
Matrix (ECM) modulation
Transcriptional dysregulation:
More than 400 genes are upregulated by DUX4 expression compared to healthy muscle

(A) Schematic of the loss in gene repression caused by contraction of D4Z4 repeats that leads to DUX4 expression
(B) Schematic describing the downstream transcriptional consequences in the muscle of FSHD subjects.

2. MRI guided skeletal muscle biopsies collected and analyzed for DUX4 transcriptional signature

A.

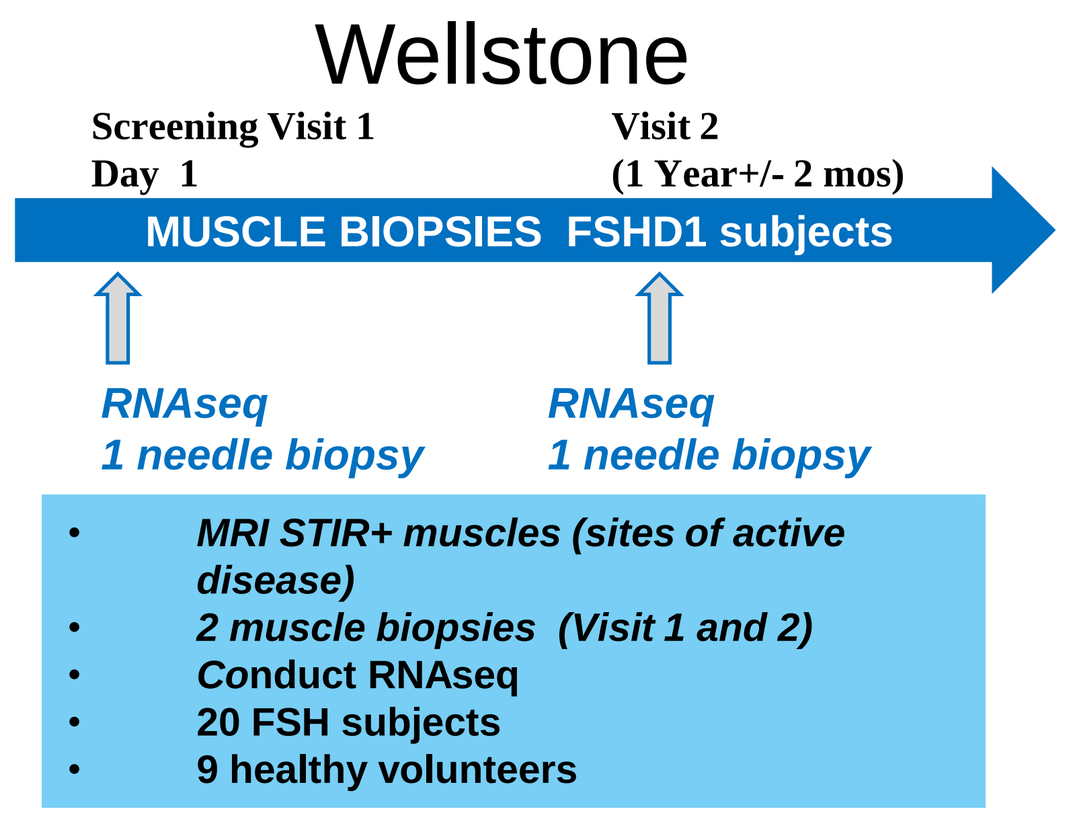
Wellstone
Screening Visit 1
Day 1
Visit 2
(1 Year+/- 2 mos)
MUSCLE BIOPSIES FSHD1 subjects
RNAseq
1 needle biopsy
RNAseq
1 needle biopsy
MRI STIR+ muscles (sites of active disease)
2 muscle biopsies (Visit 1 and 2)
Conduct RNAseq
20 FSH subjects
9 healthy volunteers

B.

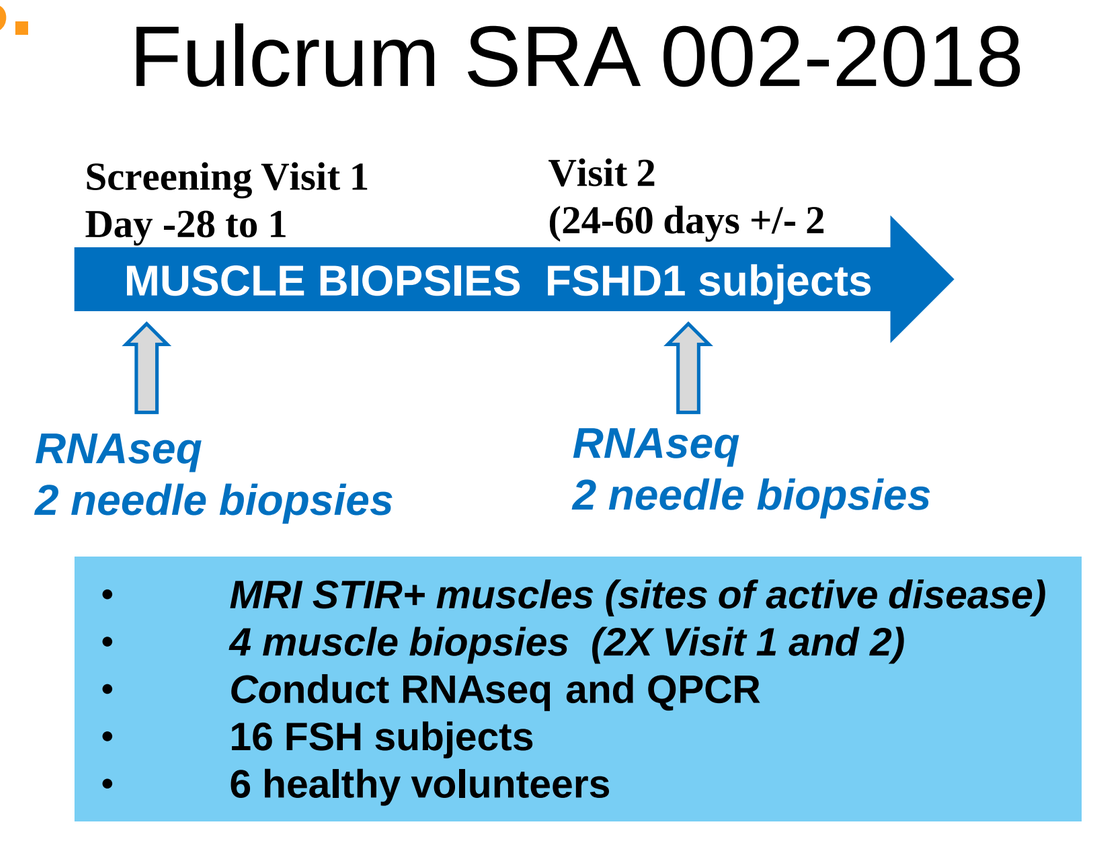
Fulcrum SRA 002-2018
Screening Visit 1
Day -28 to 1
Visit 2
(24-60 days +/- 2
MUSCLE BIOPSIES FSHD1 subjects
RNAseq
2 needle biopsies
RNAseq
2 needle biopsies
MRI STIR+ muscles (sites of active disease)
4 muscle biopsies (2X Visit 1 and 2)
Conduct RNAseq and QPCR
16 FSH subjects
6 healthy volunteers

C.

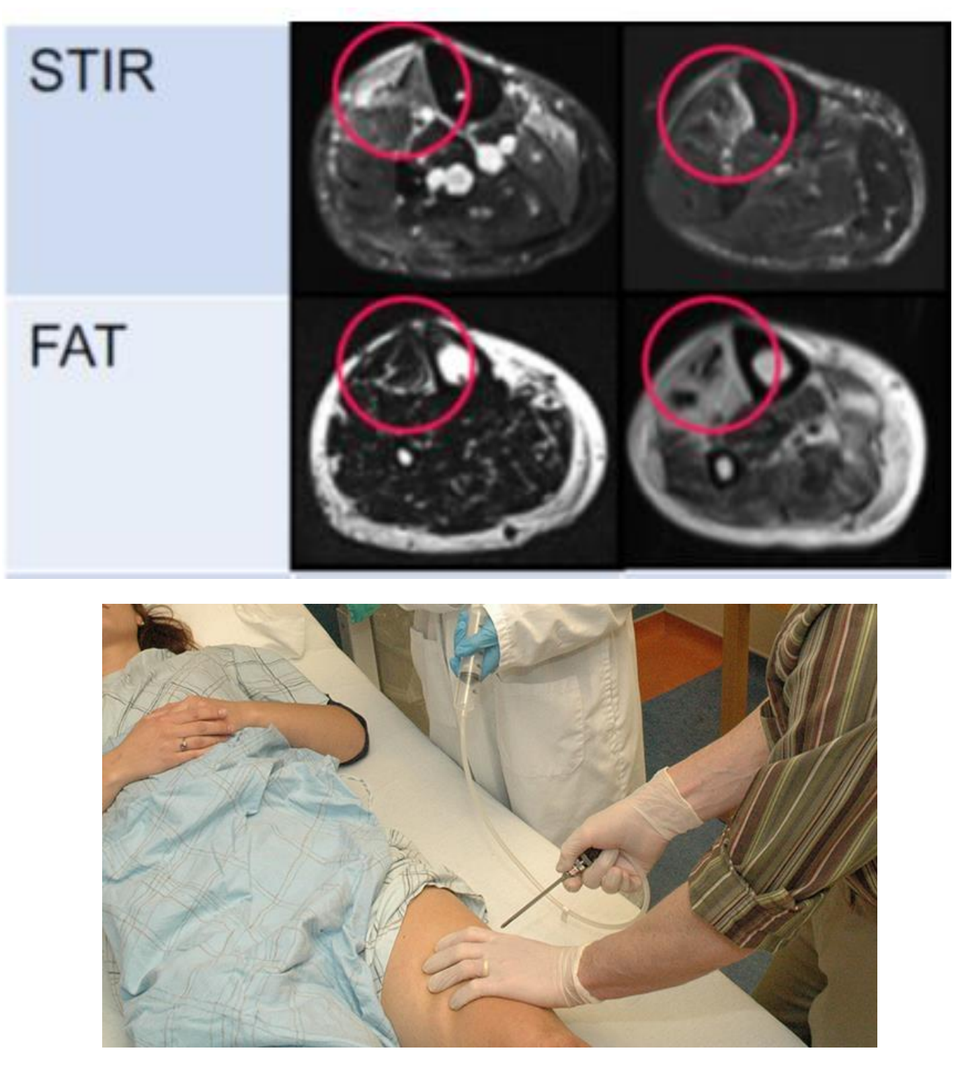
STIR
FAT

(A) Schematics of the design of the Wellstone natural history study in FSHD subjects (Wang et al., 2018)
(B) Schematic of the design of Fulcrum Therapeutics natural history preparatory study SRA-002-2018
(C) Representative images of Magnetic Resonance Imaging. Presence of short tau inversion recovery (STIR) positive signal with intermediate degree of fat replacement in at least one leg muscle was required for eligibility.

3. Analytical Approach

A comparative differential analysis of transcriptomes from control and affected FSHD muscle biopsies was conducted to identify a subset of DUX4-regulated gene transcripts that could serve as the proof of concept efficacy biomarker of DUX4 activity

1. Identified DUX4 regulated genes using literature, expert advice, and Fulcrum data from FSH myotubes in culture

2. Observed differentially expressed transcripts in RNAseq analysis of FSHD and healthy skeletal muscle biopsies

3. Observed losmapimod inhibition of these transcripts in myotubes in culture

4. Observed DUX4 dependent regulation using DUX4 antisense oligonucleotide (ASO)

Quantitative assay (Fluidigm) PCR confirmation of up to 30 DUX4 regulated and exploratory genes from biopsy samples

Criteria for identification of DUX4-regulated gene transcripts

(1) Transcripts demonstrate little to no expression in healthy muscle
(2) Transcripts are consistently detected in a majority of FSHD affected muscles
(3) Transcripts demonstrate small variation of within patient expression observed over time in short (1-2 month apart; Fulcrum study) and long (1 year apart; Wellstone) term longitudinal studies

4. Results: A stable subset of DUX4-regulated gene transcripts can be detected in skeletal muscle biopsies taken 1 year apart

A. At visit 1, 47 transcripts fulfilled selection criteria

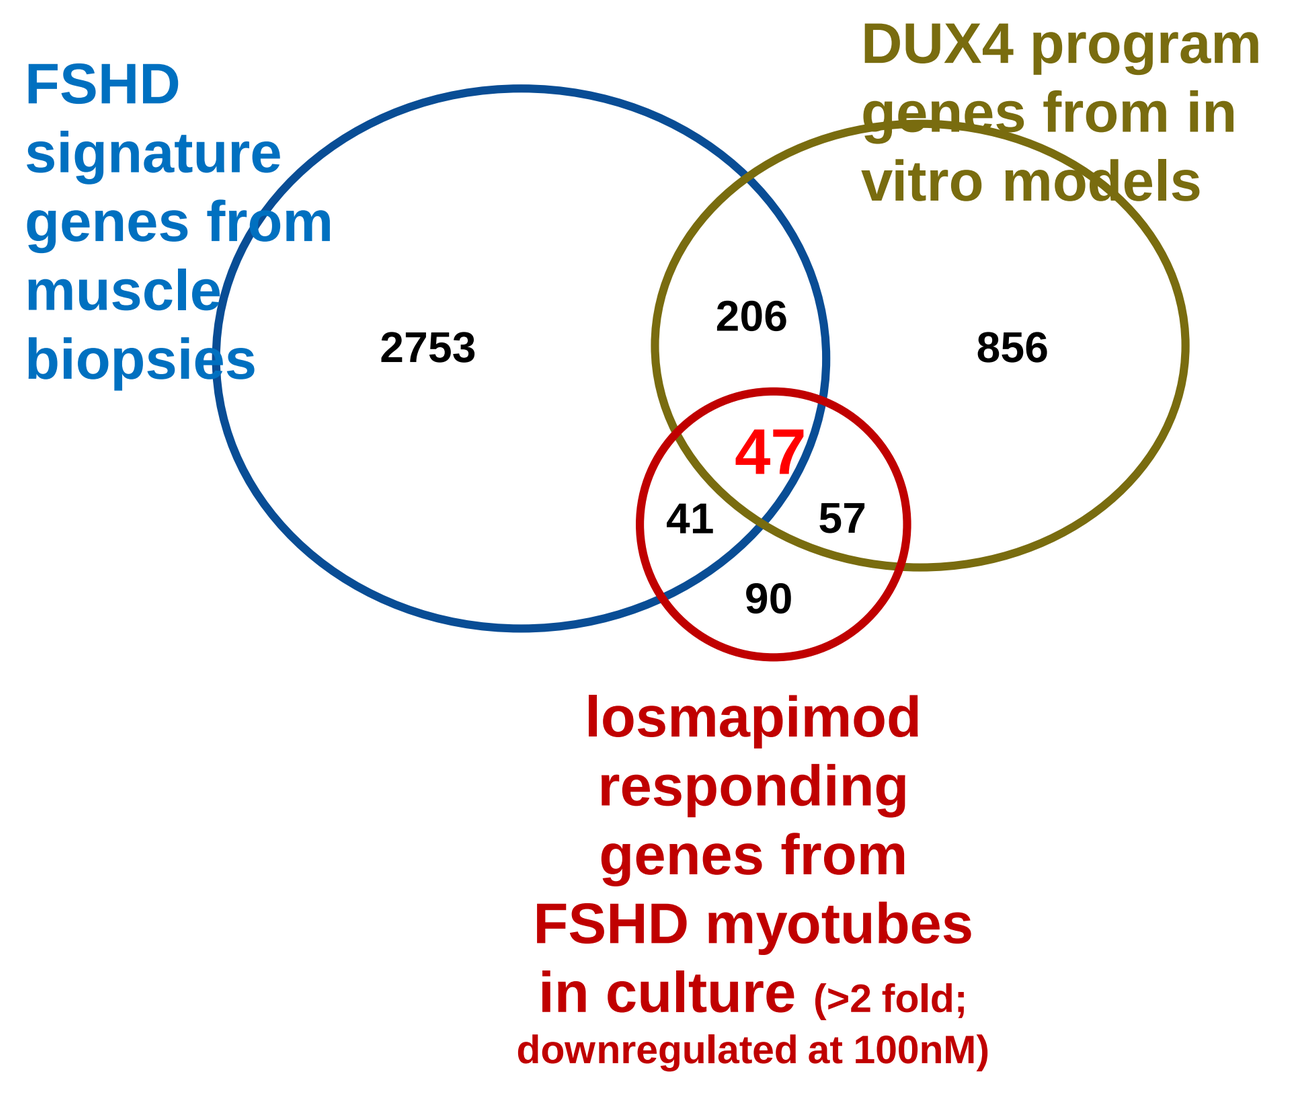
FSHD signature genes from muscle biopsies
DUX4 program genes from in vitro models
2753
206
856
47
41
57
90
losmapimod responding genes from FSHD myotubes in culture (>2 fold; downregulated at 100nM)

B. 35 transcripts are stably detected after one year

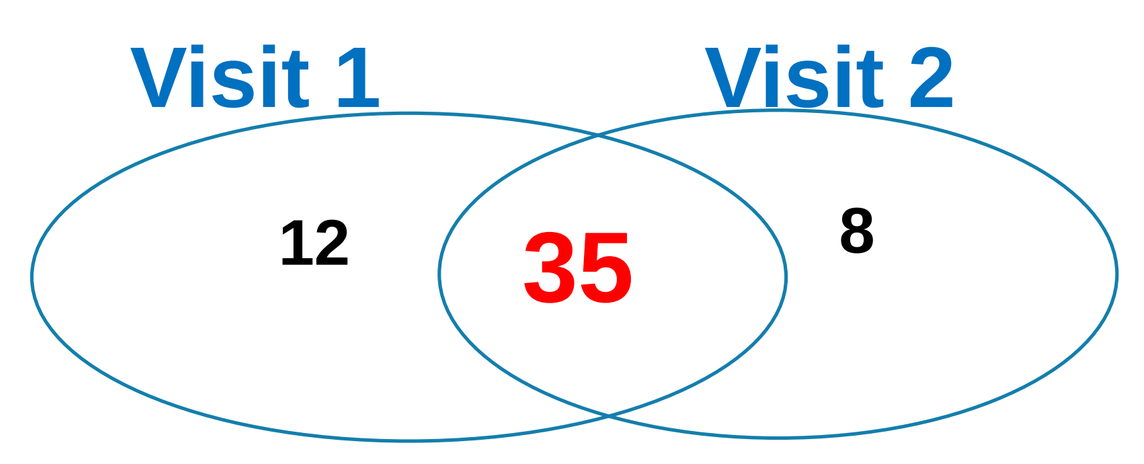
Visit 1
Visit 2
12
35
8

C. A subset of DUX4 regulated gene transcripts found to be stably expressed in FSHD muscle biopsies taken 1 year apart

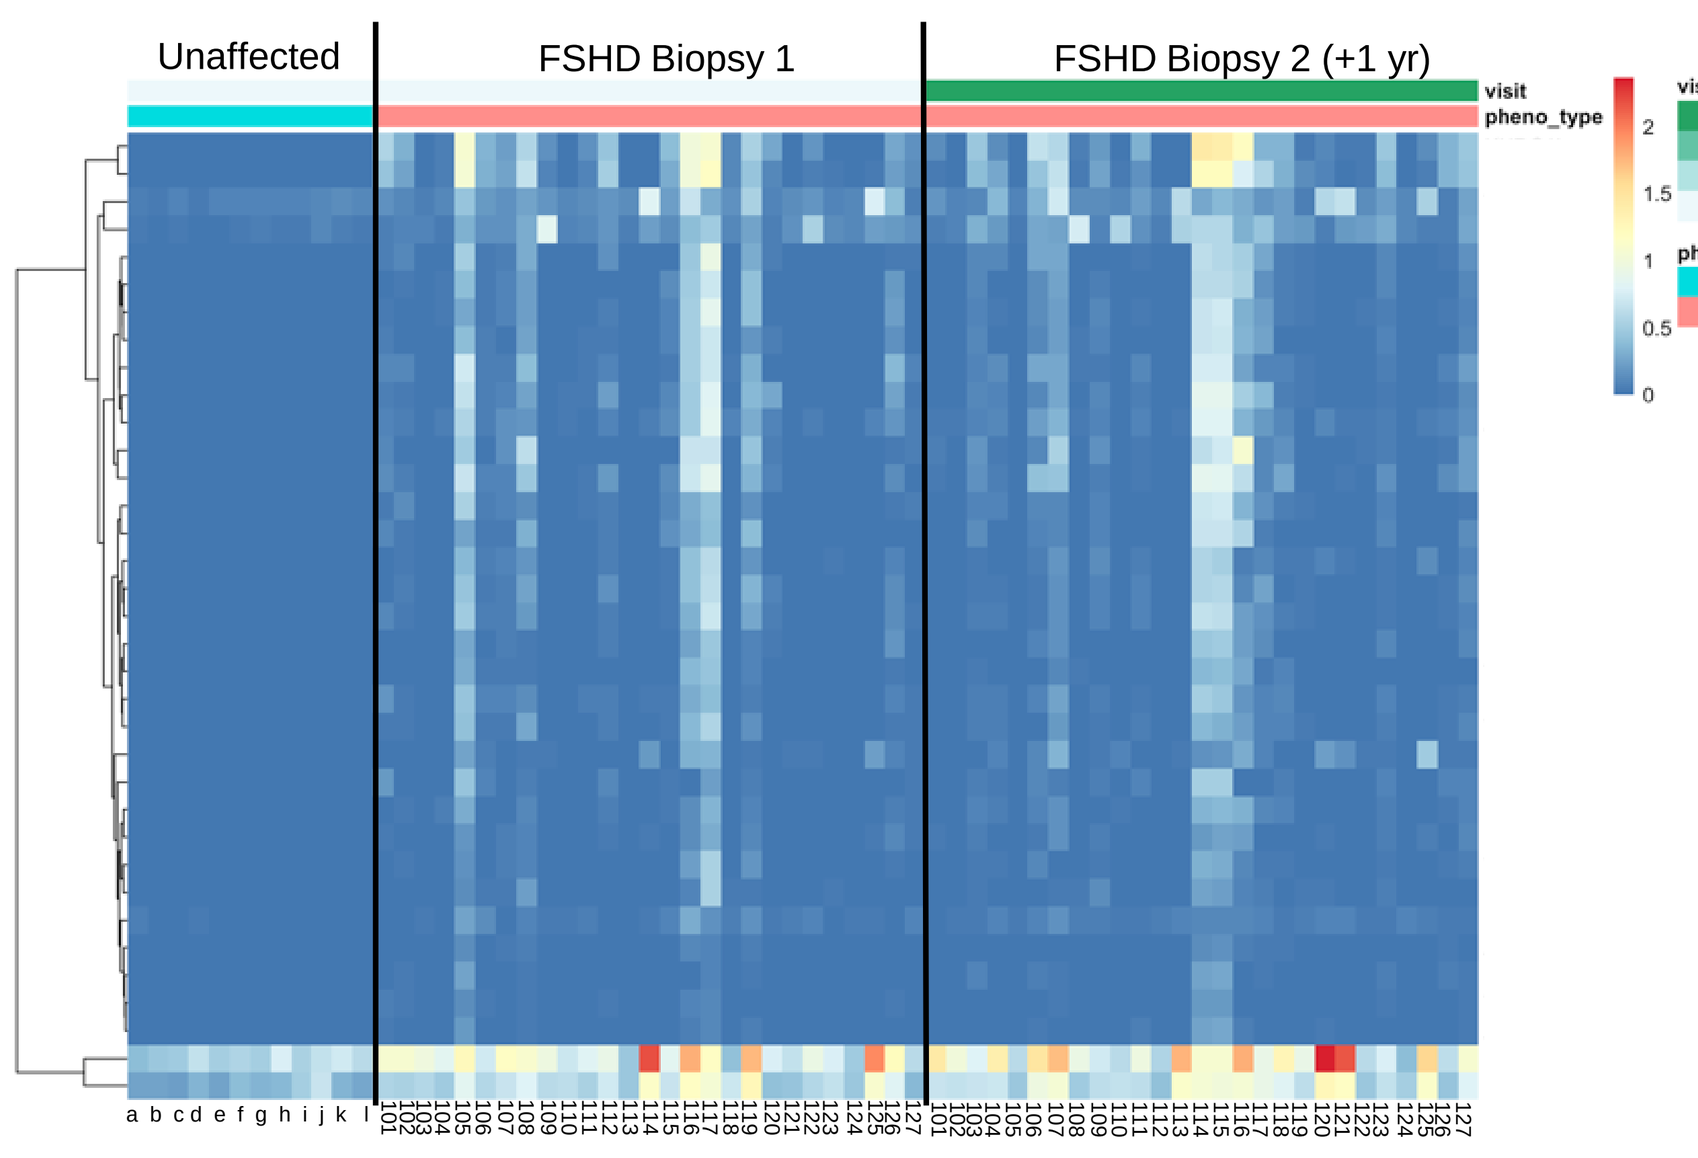
Unaffected
FSHD Biopsy 1
FSHD Biopsy 2 (+1 yr)
visit
pheno_type
Control
FSHD

D. Quantification of expression of the subset of DUX4-regulated gene transcripts from affected FSHD muscles biopsied twice 1 year apart

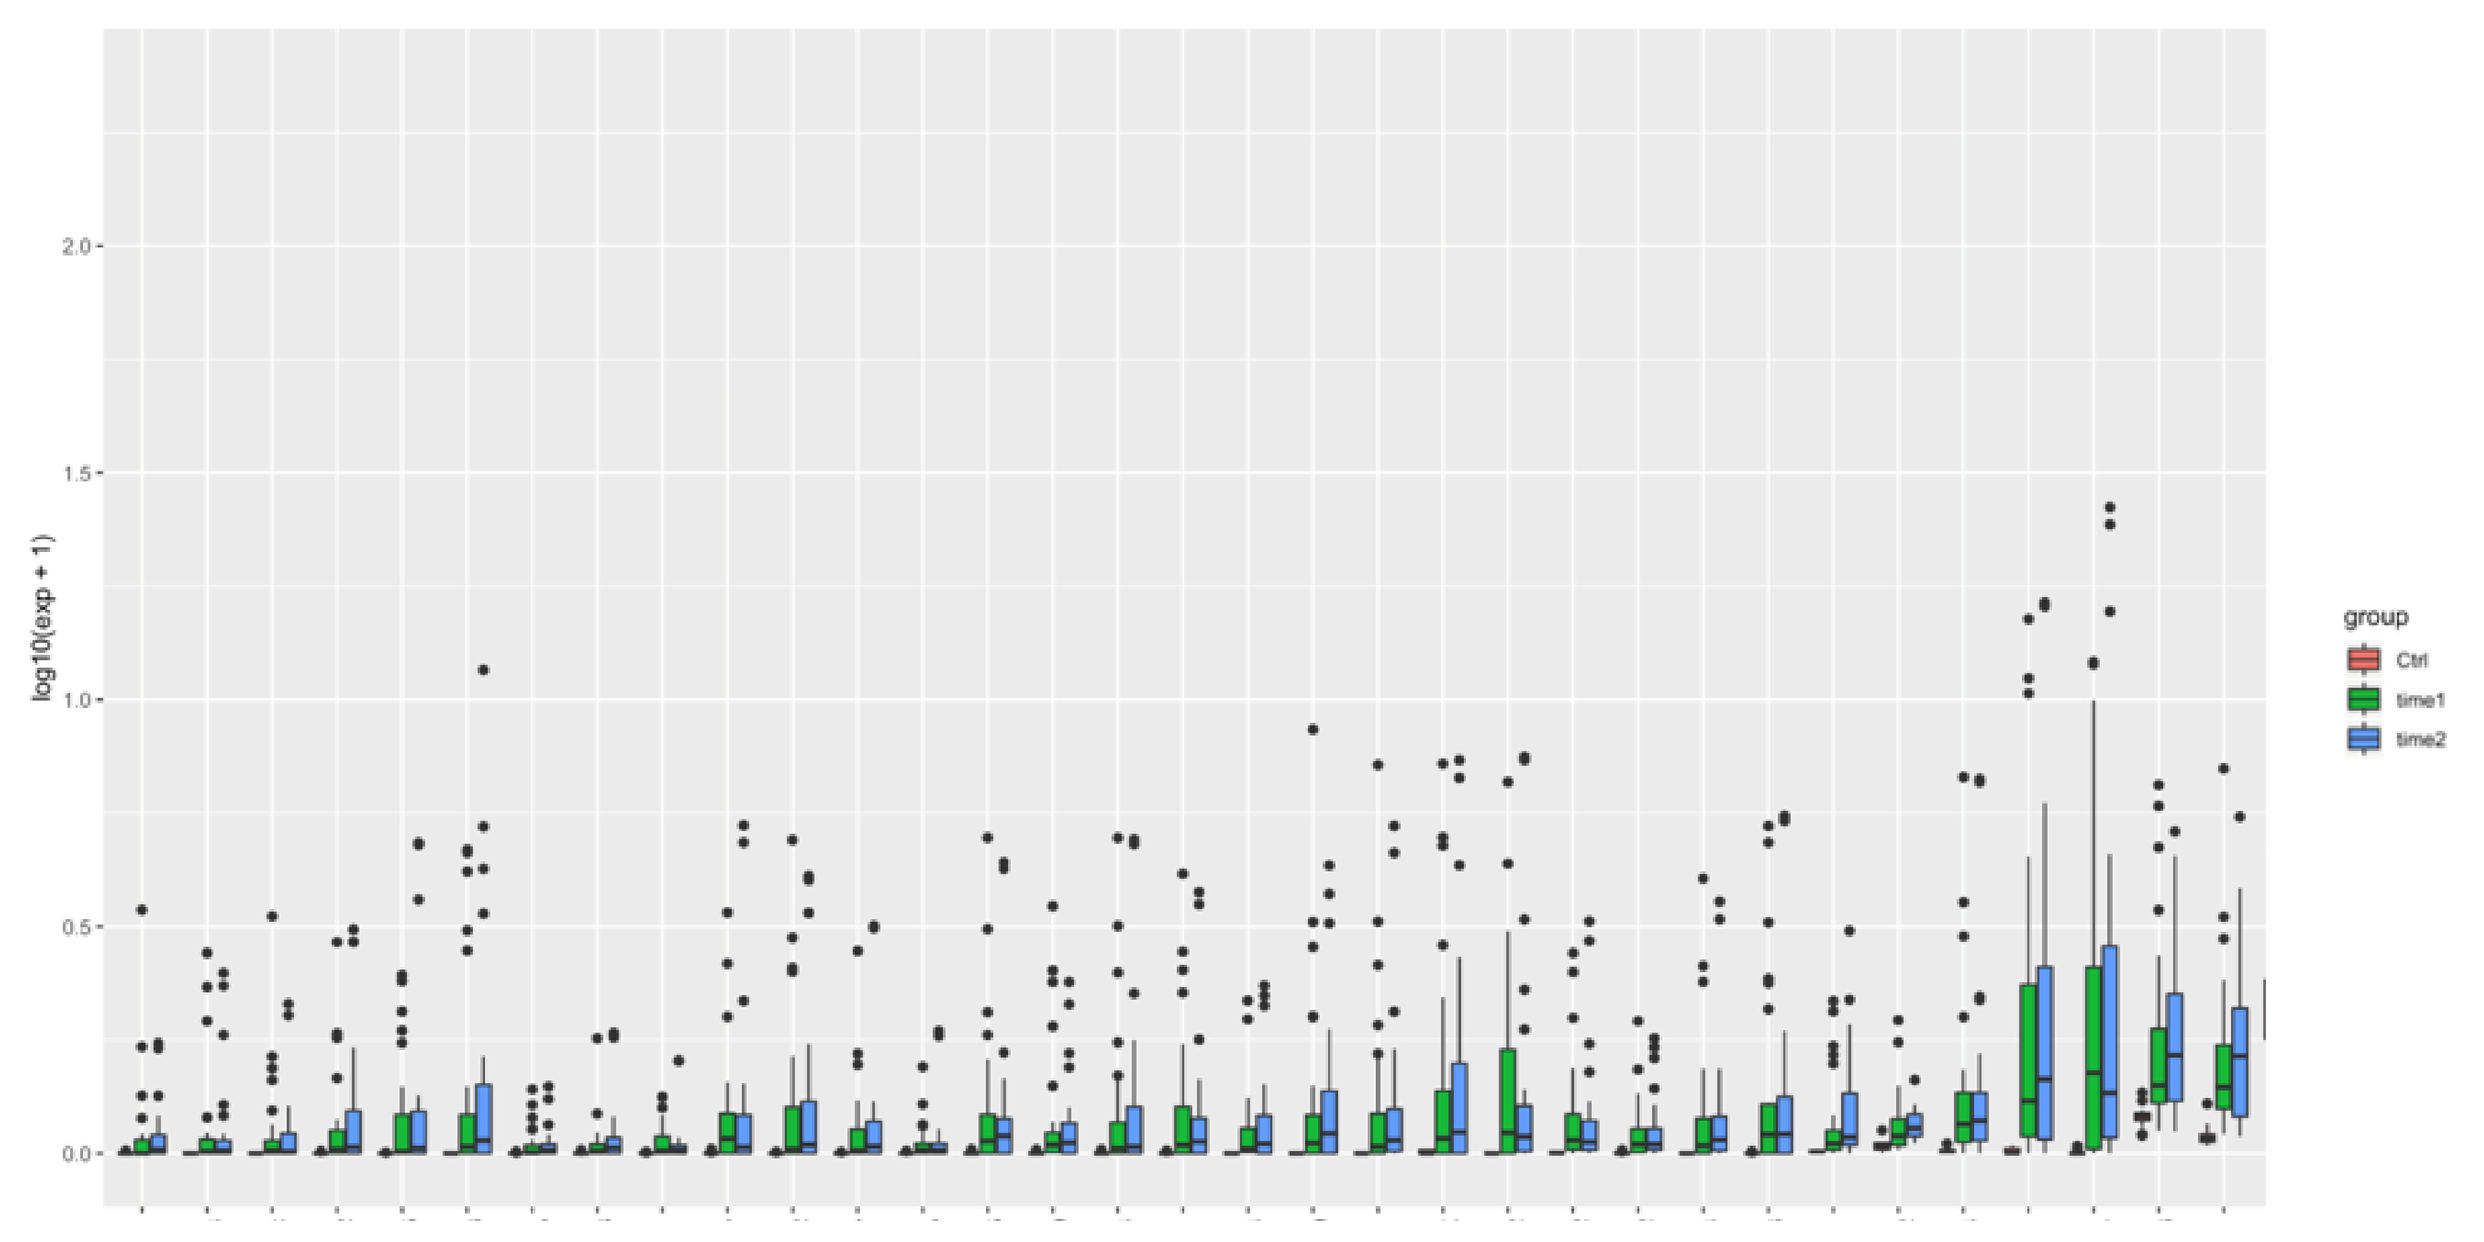
log10(exp + 1)
group
Ctrl
time1
time2

(C) The RNAseq transcript heatmap displays Gene Fragments Per Kilobase of transcript per Million (FPKM) mapped reads for all genes in the gene subset. FPKM value is log2(FPKM). Control and FSHD samples (visits 1 and 2) are included in the data set.
(D) Boxplot of DUX4-regulated and differentially expressed transcripts identified in FSHD skeletal muscle compared to healthy biopsies. Transcripts are sorted by mean abundance in expression across all groups. Abundance is calculated as the log2 of FPKM.

5. A subset of genes was confirmed to be directly regulated by DUX4.

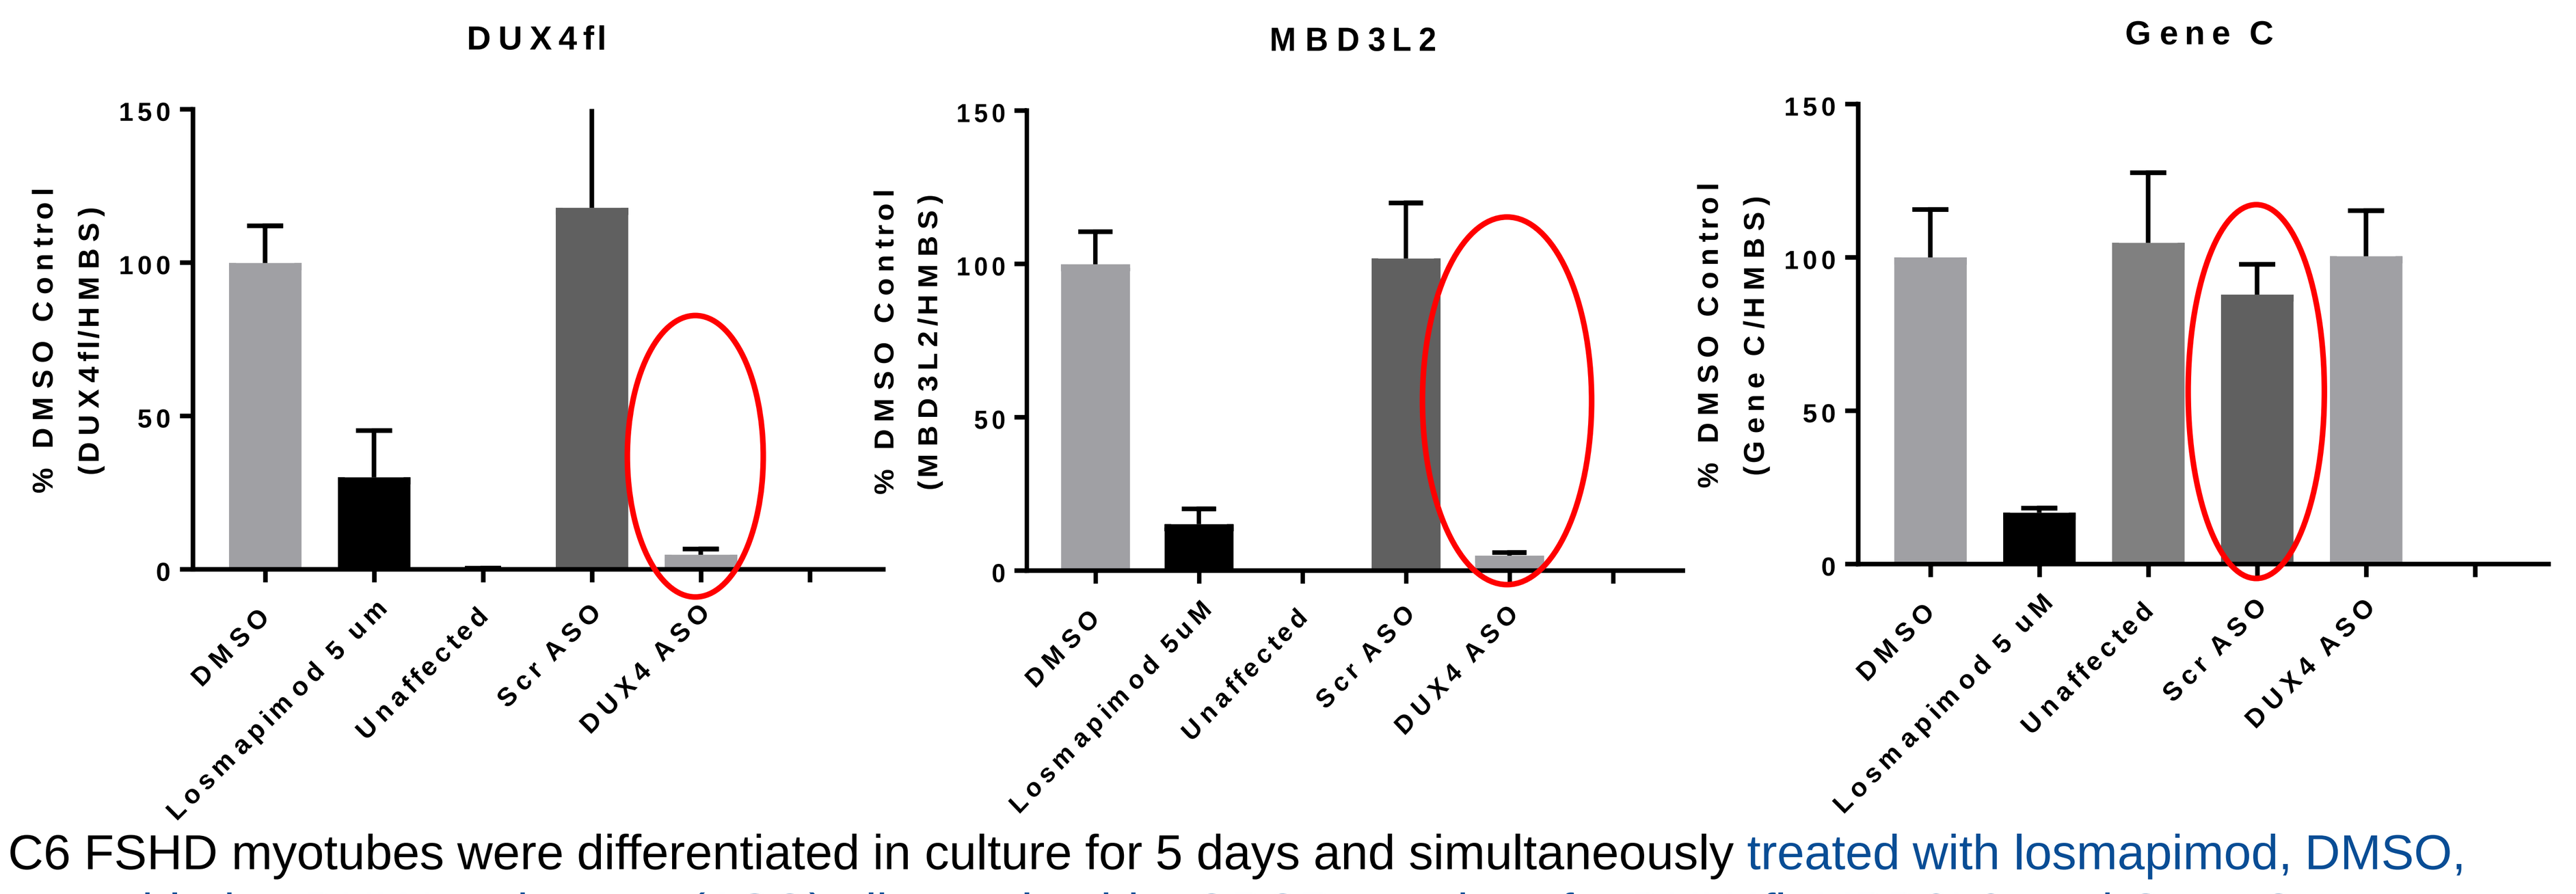
DUX4fl
MBD3L2
Gene C
% DMSO Control (DUX4fl/HMBS)
% DMSO Control (MBD3L2/HMBS)
% DMSO Control (Gene C/HMBS)
DMSO
Losmapimod 5 um
Losmapimod 5uM
Losmapimod 5 uM
Unaffected
Scr ASO
DUX4 ASO

C6 FSHD myotubes were differentiated in culture for 5 days and simultaneously treated with losmapimod, DMSO, scrambled or DUX4 anti-sense (ASO) oligonucleotide. QPCR reactions for DUX4fl, MBD3L2, and Gene C were carried out.

6. Conclusion

This study identified DUX4 regulated gene transcripts that are stably expressed for at least one year in affected muscle biopsies from FSHD subjects. An initial subset of DUX4 regulated gene transcripts to use as biomarker of DUX4 activity to evaluate losmapimod treatment effect in FSHD trials has been identified. The analysis of Fulcrum study SRA-002-2018 is still ongoing.

References

(1) Wang et al., 2018
(2) Jagannathan et al., 2016
(3) Rojas et al., 2019; P38α Regulates Expression of DUX4 in Facioscapulohumeral Muscular Dystrophy