Targeting DUX4 Expression, the Root Cause of FSHD: Identification of a Drug Target and Development Candidate

Owen B. Wallace, Anthony Accorsi, Richard Barnes, Angela Cacace, Diego Cadavid, Aaron Chang, David Eyerman, Robert Gould, Steven Kazmirski, Joseph Maglio, Michelle Mellion, Peter Rahl, Alan Robertson, Alejandro Rojas, Lucienne Ronco, Ning Shen, Lorin A. Thompson and Erin Valentine

Fulcrum Therapeutics. 26 Landsdowne Street, Cambridge, Massachusetts, USA.

Abstract

- FSHD is caused by the loss of repression at the D4Z4 locus leading to aberrant DUX4 expression in skeletal muscle, activation of its early embryo transcriptional program and muscle fiber death.
- While some progress toward understanding the signals driving DUX4 expression has been made, the factors and pathways involved in the transcriptional activation of this gene remain largely unknown.
- Using optimized myotube culture conditions, we identified p38 MAPK as a key regulator of DUX4 expression.
- We observed that treatment with the p38α/β inhibitor losmapimod results in reduction of DUX4 expression, activity and cell death in FSHD patient-derived myotubes with minimal impact on myogenesis.
- RNA-seq studies revealed that only a small number of genes were differentially expressed after treatment with losmapimod, ~90% of these are targets of DUX4.
- Fulcrum Therapeutics has selected losmapimod, a specific p38α/β inhibitor, for clinical development in FSHD.

1. Facioscapulohumeral Muscular Dystrophy (FSHD)

A. B.

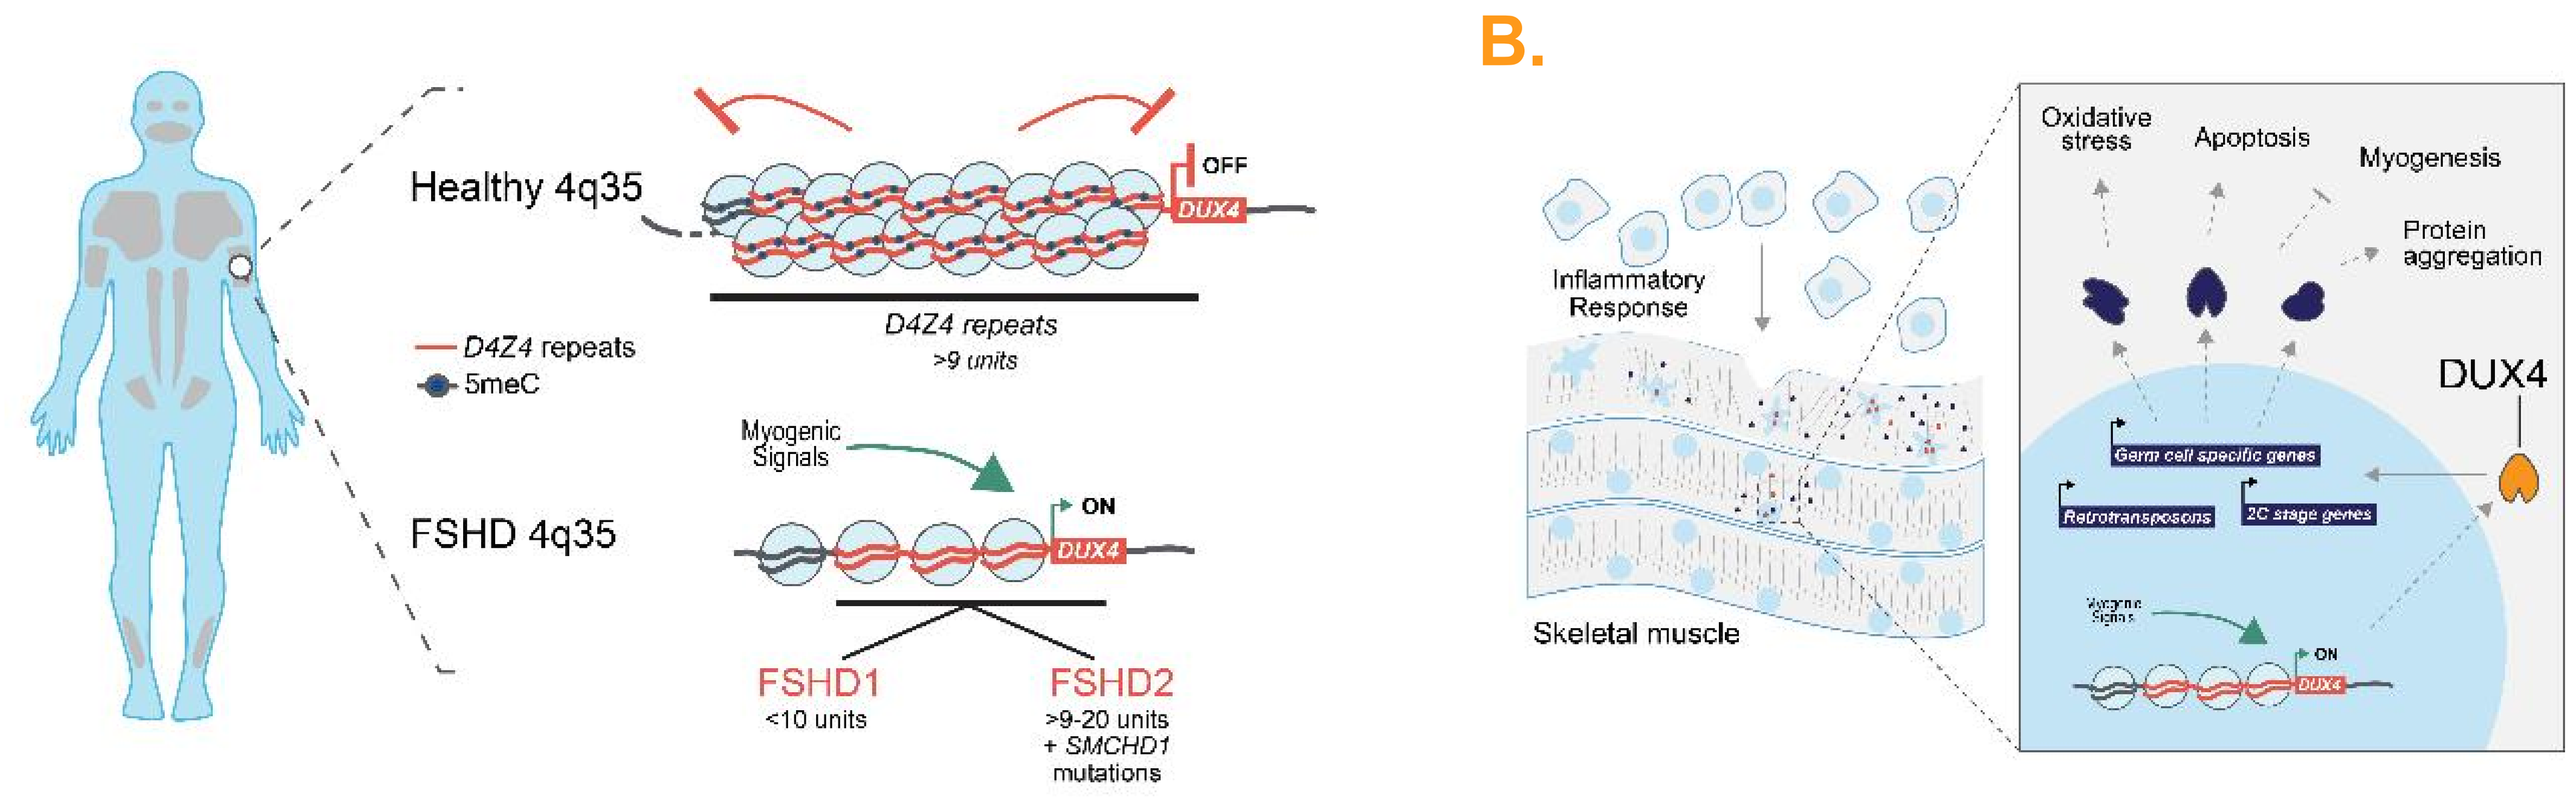
Healthy 4q35
OFF
DUX4
D4Z4 repeats
>9 units
D4Z4 repeats
5meC
Myogenic Signals
ON
FSHD 4q35
FSHD1
<10 units
FSHD2
>9-20 units
+ SMCHD1 mutations
Oxidative stress
Apoptosis
Myogenesis
Protein aggregation
Inflammatory Response
DUX4
Germ cell specific genes
Retrotransposons
2C stage genes
Skeletal muscle

(A) Schematic of the loss in gene repression caused by contraction of D4Z4 repeats that leads to DUX4 expression in the muscle of FSHD1 patients.
(B) Schematic describing the downstream consequences of DUX4 expression in skeletal muscle.

2. Fulcrum's approach to target identification and validation

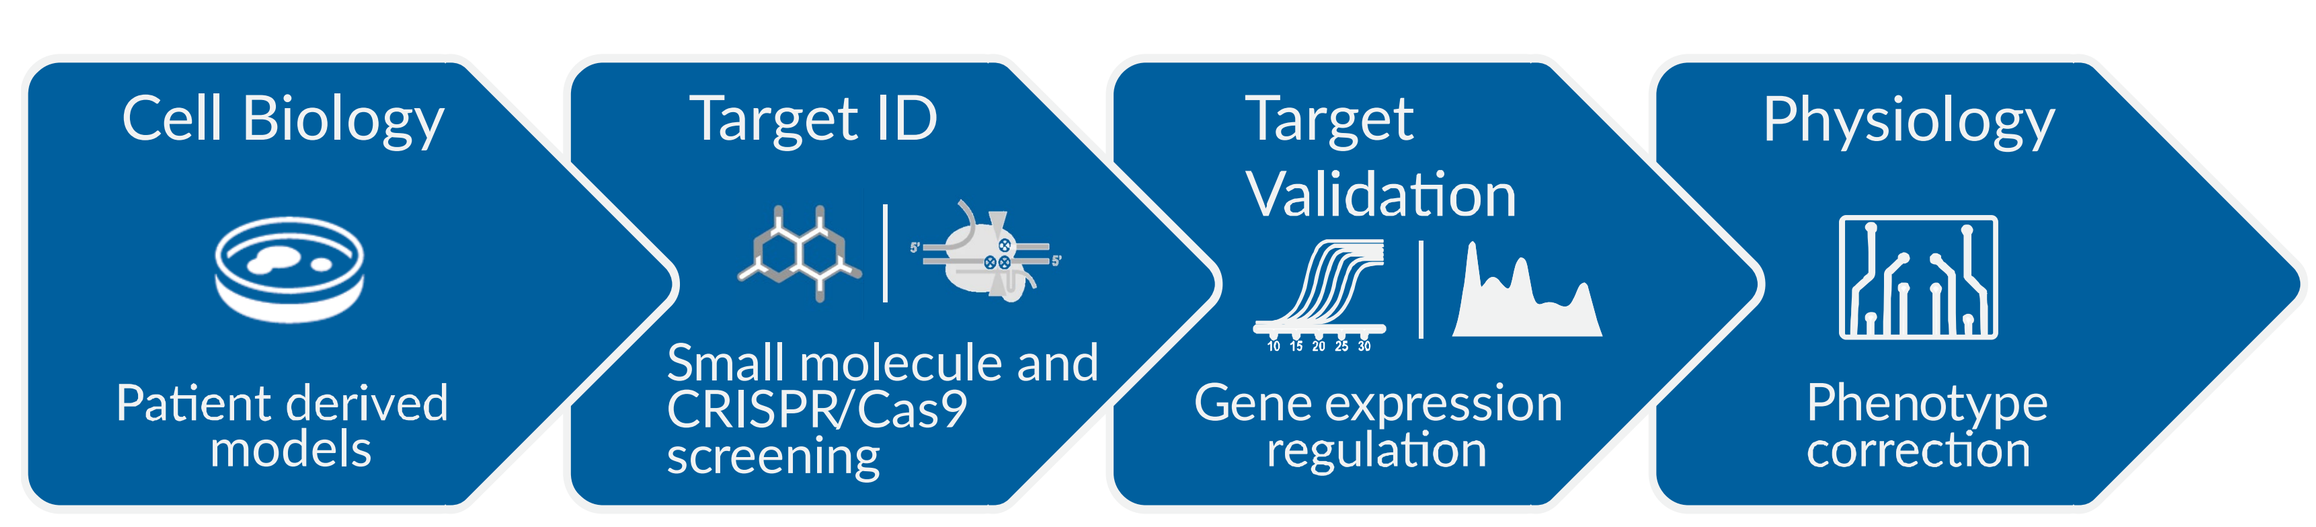
Cell Biology
Patient derived models
Target ID
Small molecule and CRISPR/Cas9 screening
Target Validation
Gene expression regulation
Physiology
Phenotype correction

Cell-based screening drives Fulcrum's target identification engine

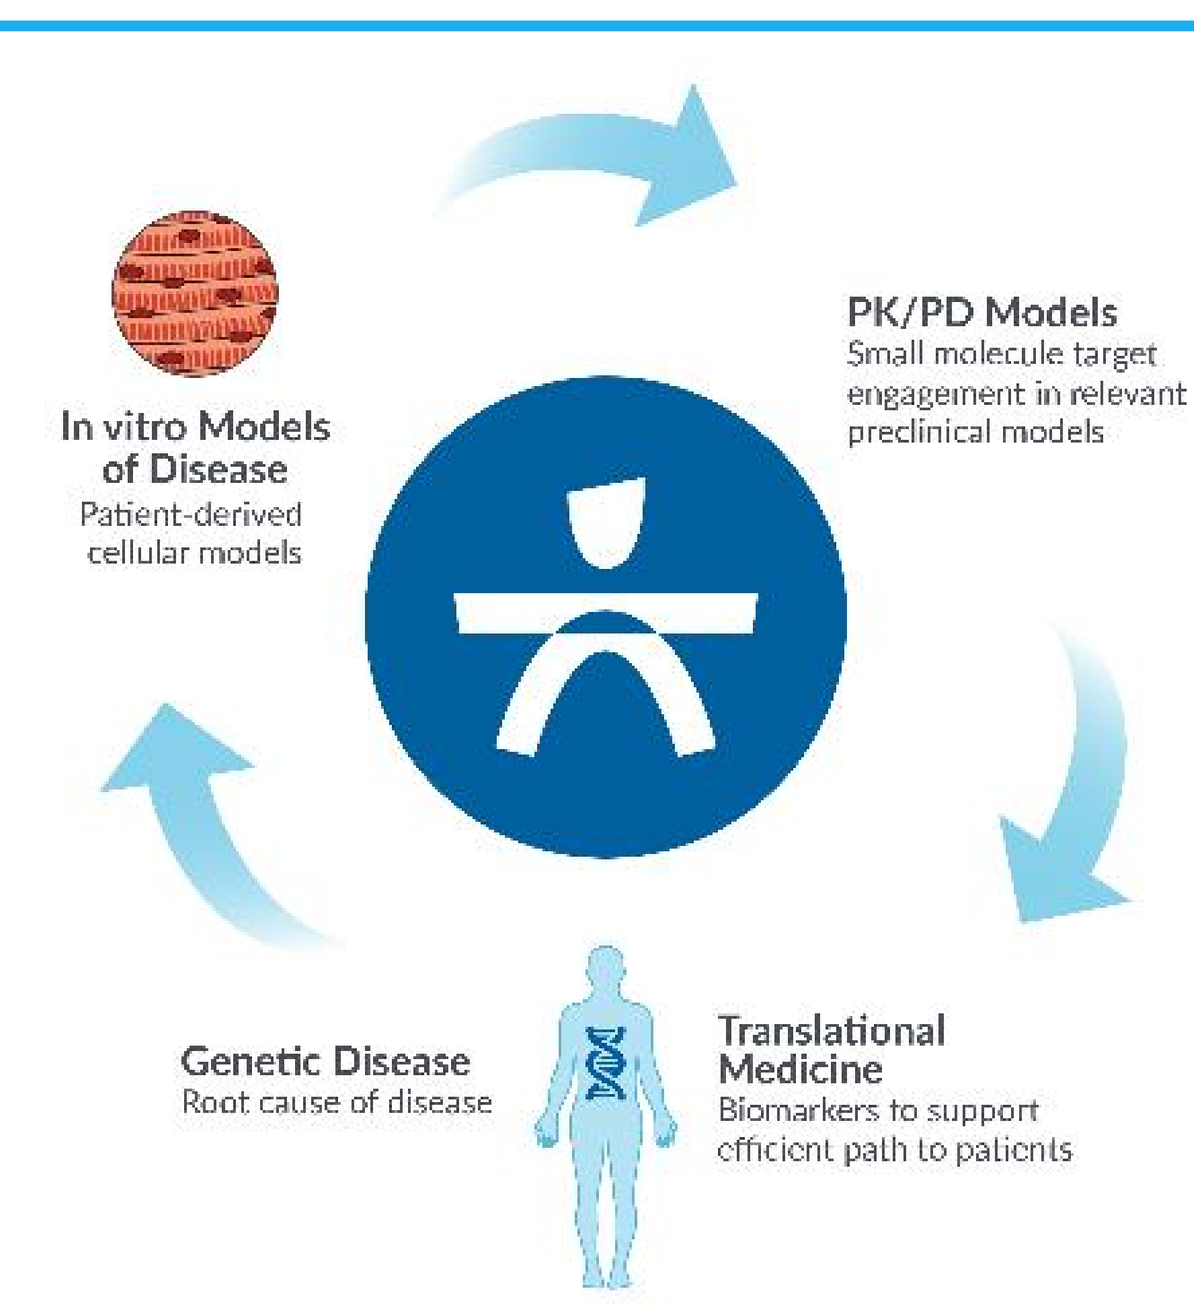
In vitro Models of Disease
Patient-derived cellular models
PK/PD Models
Small molecule target engagement in relevant preclinical models
Translational Medicine
Biomarkers to support efficient path to patients
Genetic Disease
Root cause of disease

3. Identification of a drug target that inhibits DUX4 expression

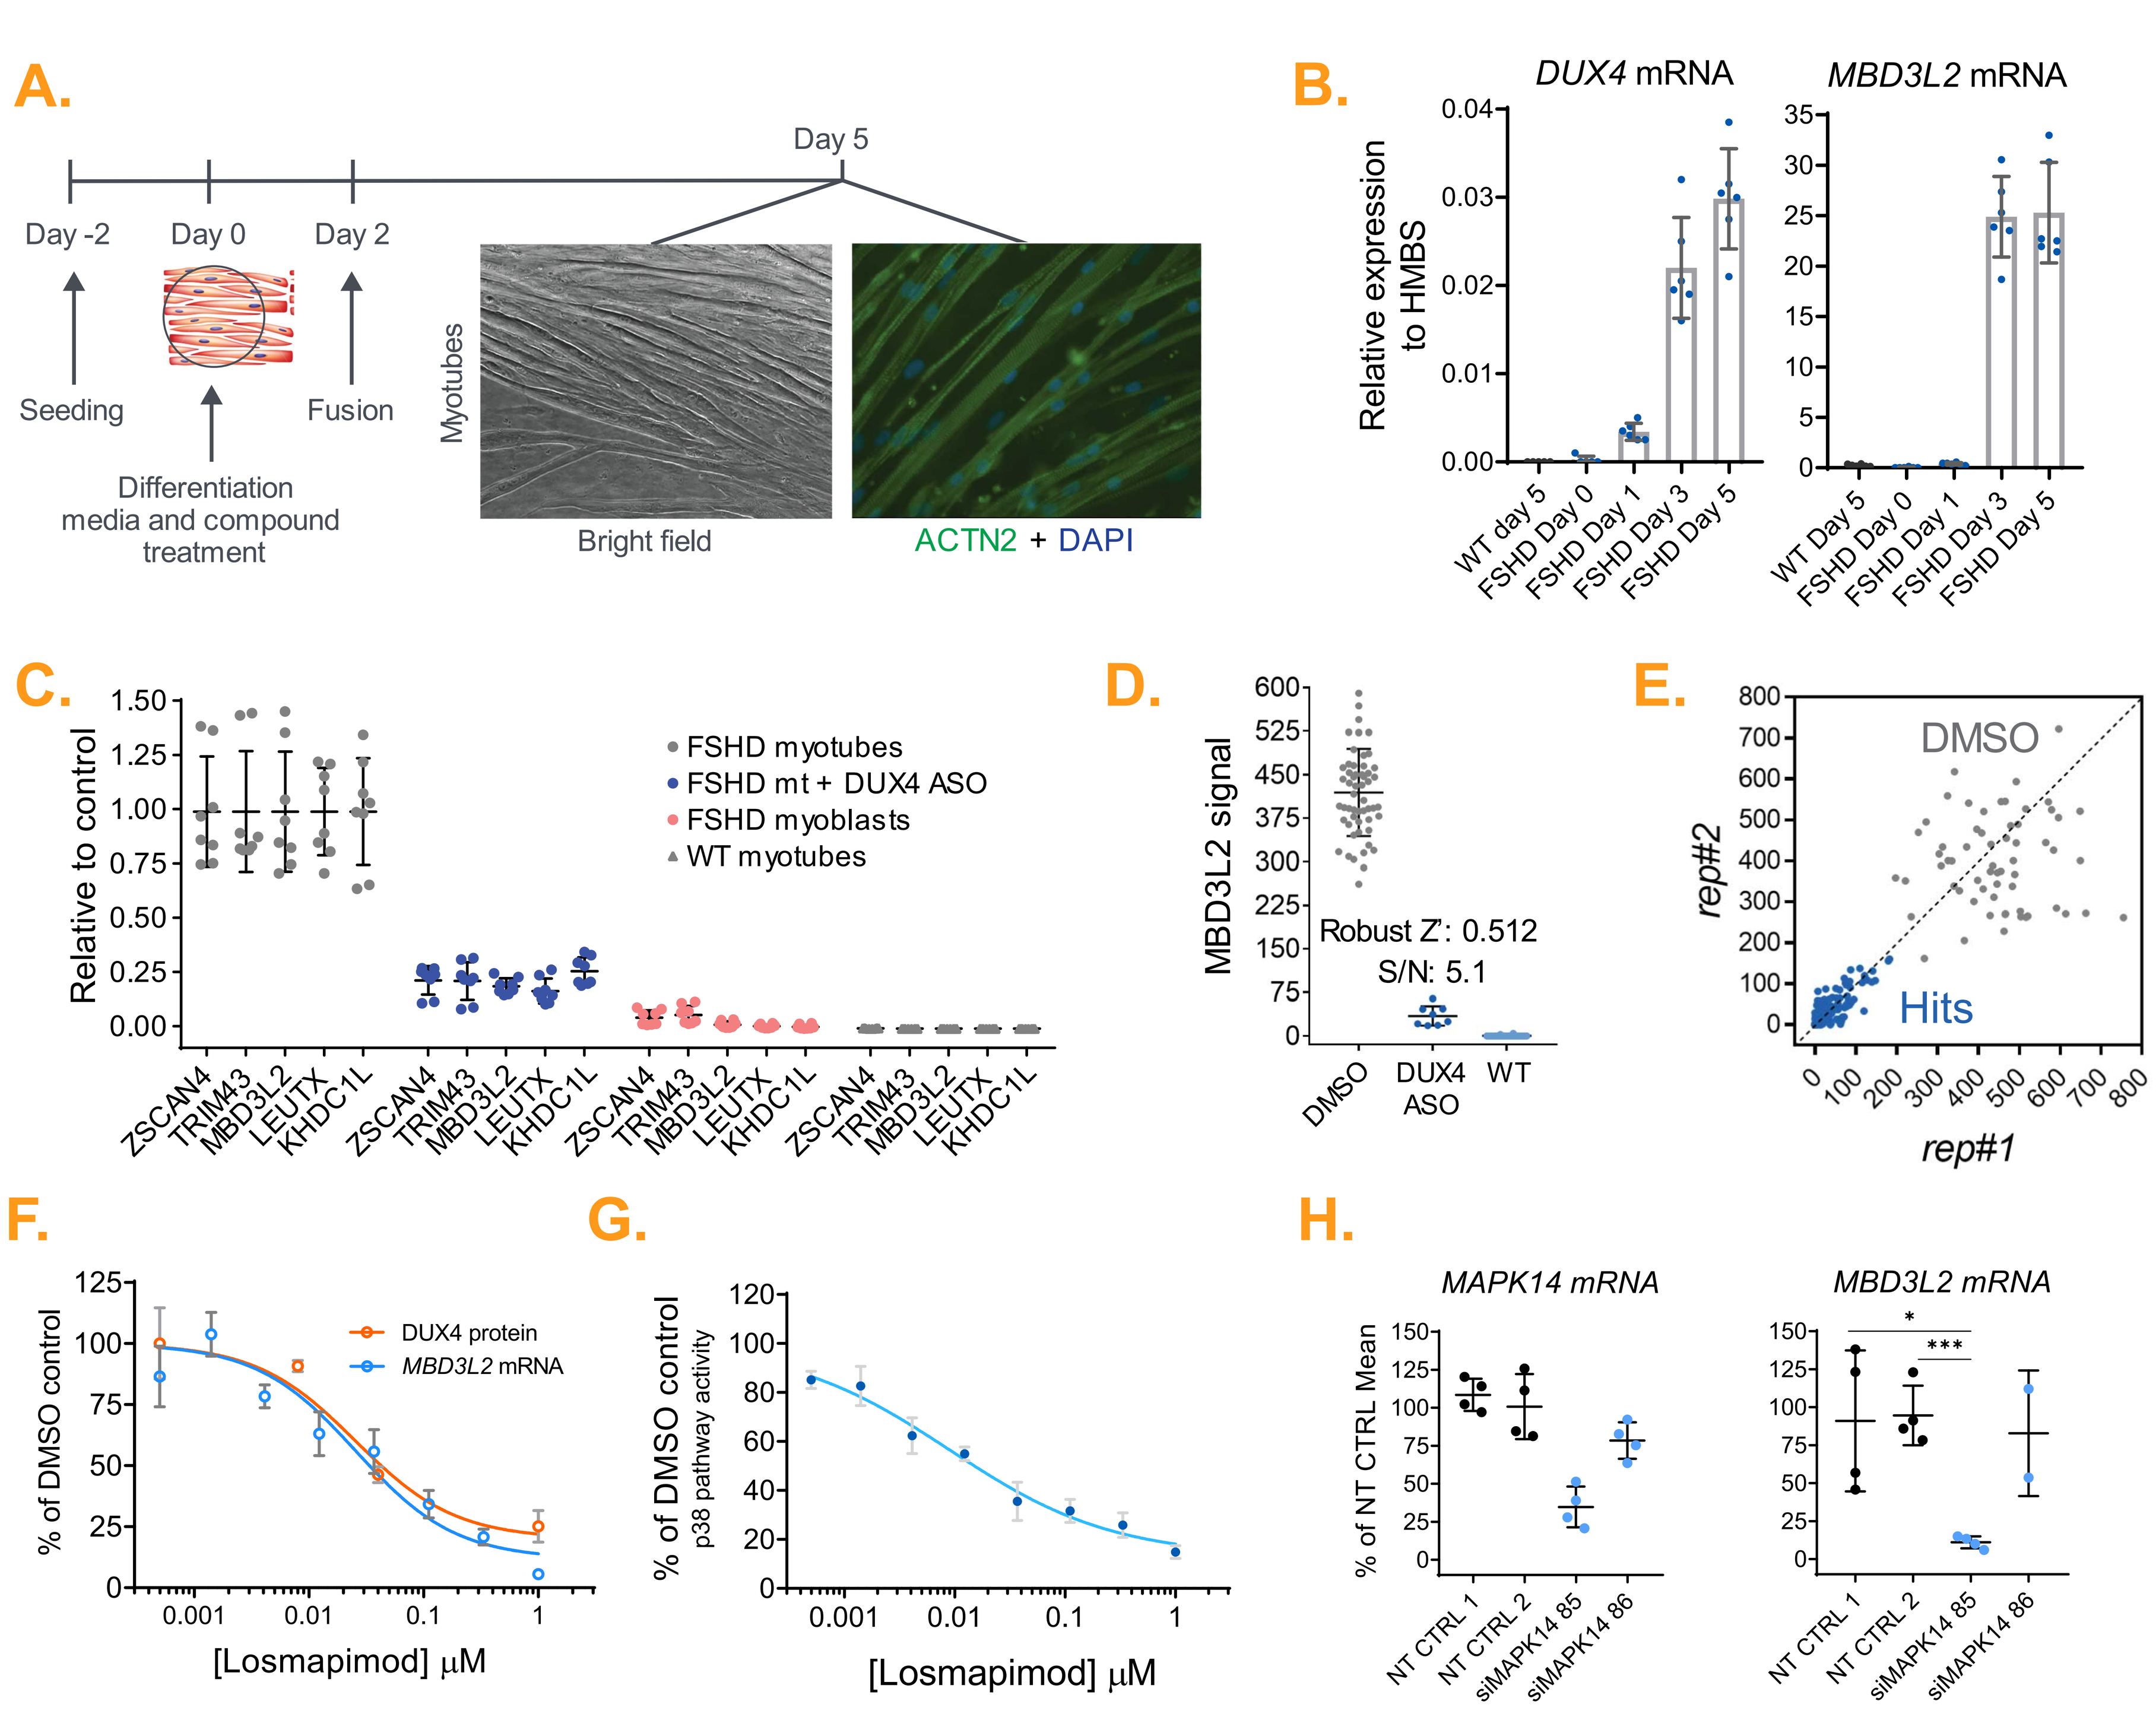
A.
Day -2
Day 0
Day 2
Day 5
Seeding
Fusion
Differentiation media and compound treatment
Myotubes
Bright field
ACTN2 + DAPI
B.
DUX4 mRNA
MBD3L2 mRNA
Relative expression to HMBS
WT day 5
FSHD Day 0
FSHD Day 1
FSHD Day 3
FSHD Day 5
C.
Relative to control
FSHD myotubes
FSHD mt + DUX4 ASO
FSHD myoblasts
WT myotubes
ZSCAN4
TRIM43
MBD3L2
LEUTX
KHDC1L
D.
MBD3L2 signal
Robust Z': 0.512
S/N: 5.1
DMSO
DUX4 ASO
WT
E.
DMSO
Hits
rep#2
rep#1
F.
DUX4 protein
MBD3L2 mRNA
% of DMSO control
[Losmapimod] µM
G.
% of DMSO control
p38 pathway activity
[Losmapimod] µM
H.
MAPK14 mRNA
MBD3L2 mRNA
% of NT CTRL Mean
NT CTRL 1
NT CTRL 2
siMAPK14 85
siMAPK14 86

(A) Cell-based assay schematic. **(B)** Expression of DUX4 and DUX4 target gene *MBD3L2* during FSHD myotube differentiation in vitro. **(C)** DUX4 target gene selection for HTS. **(D)** 96-well format assay allows for identification of targets modulating the expression of DUX4. **(E)** Hits identified showed high correlation in between biological replicates. **(F)** Losmapimod reduces expression of DUX4 in a concentration dependent manner in FSHD myotubes. **(G)** Losmapimod reduces p38 α/β activity in FSHD myotubes. **(H)** P38α knockdown reduces activity of DUX4 in FSHD myotubes.

4. Losmapimod reduces DUX4 activation and its downstream consequences

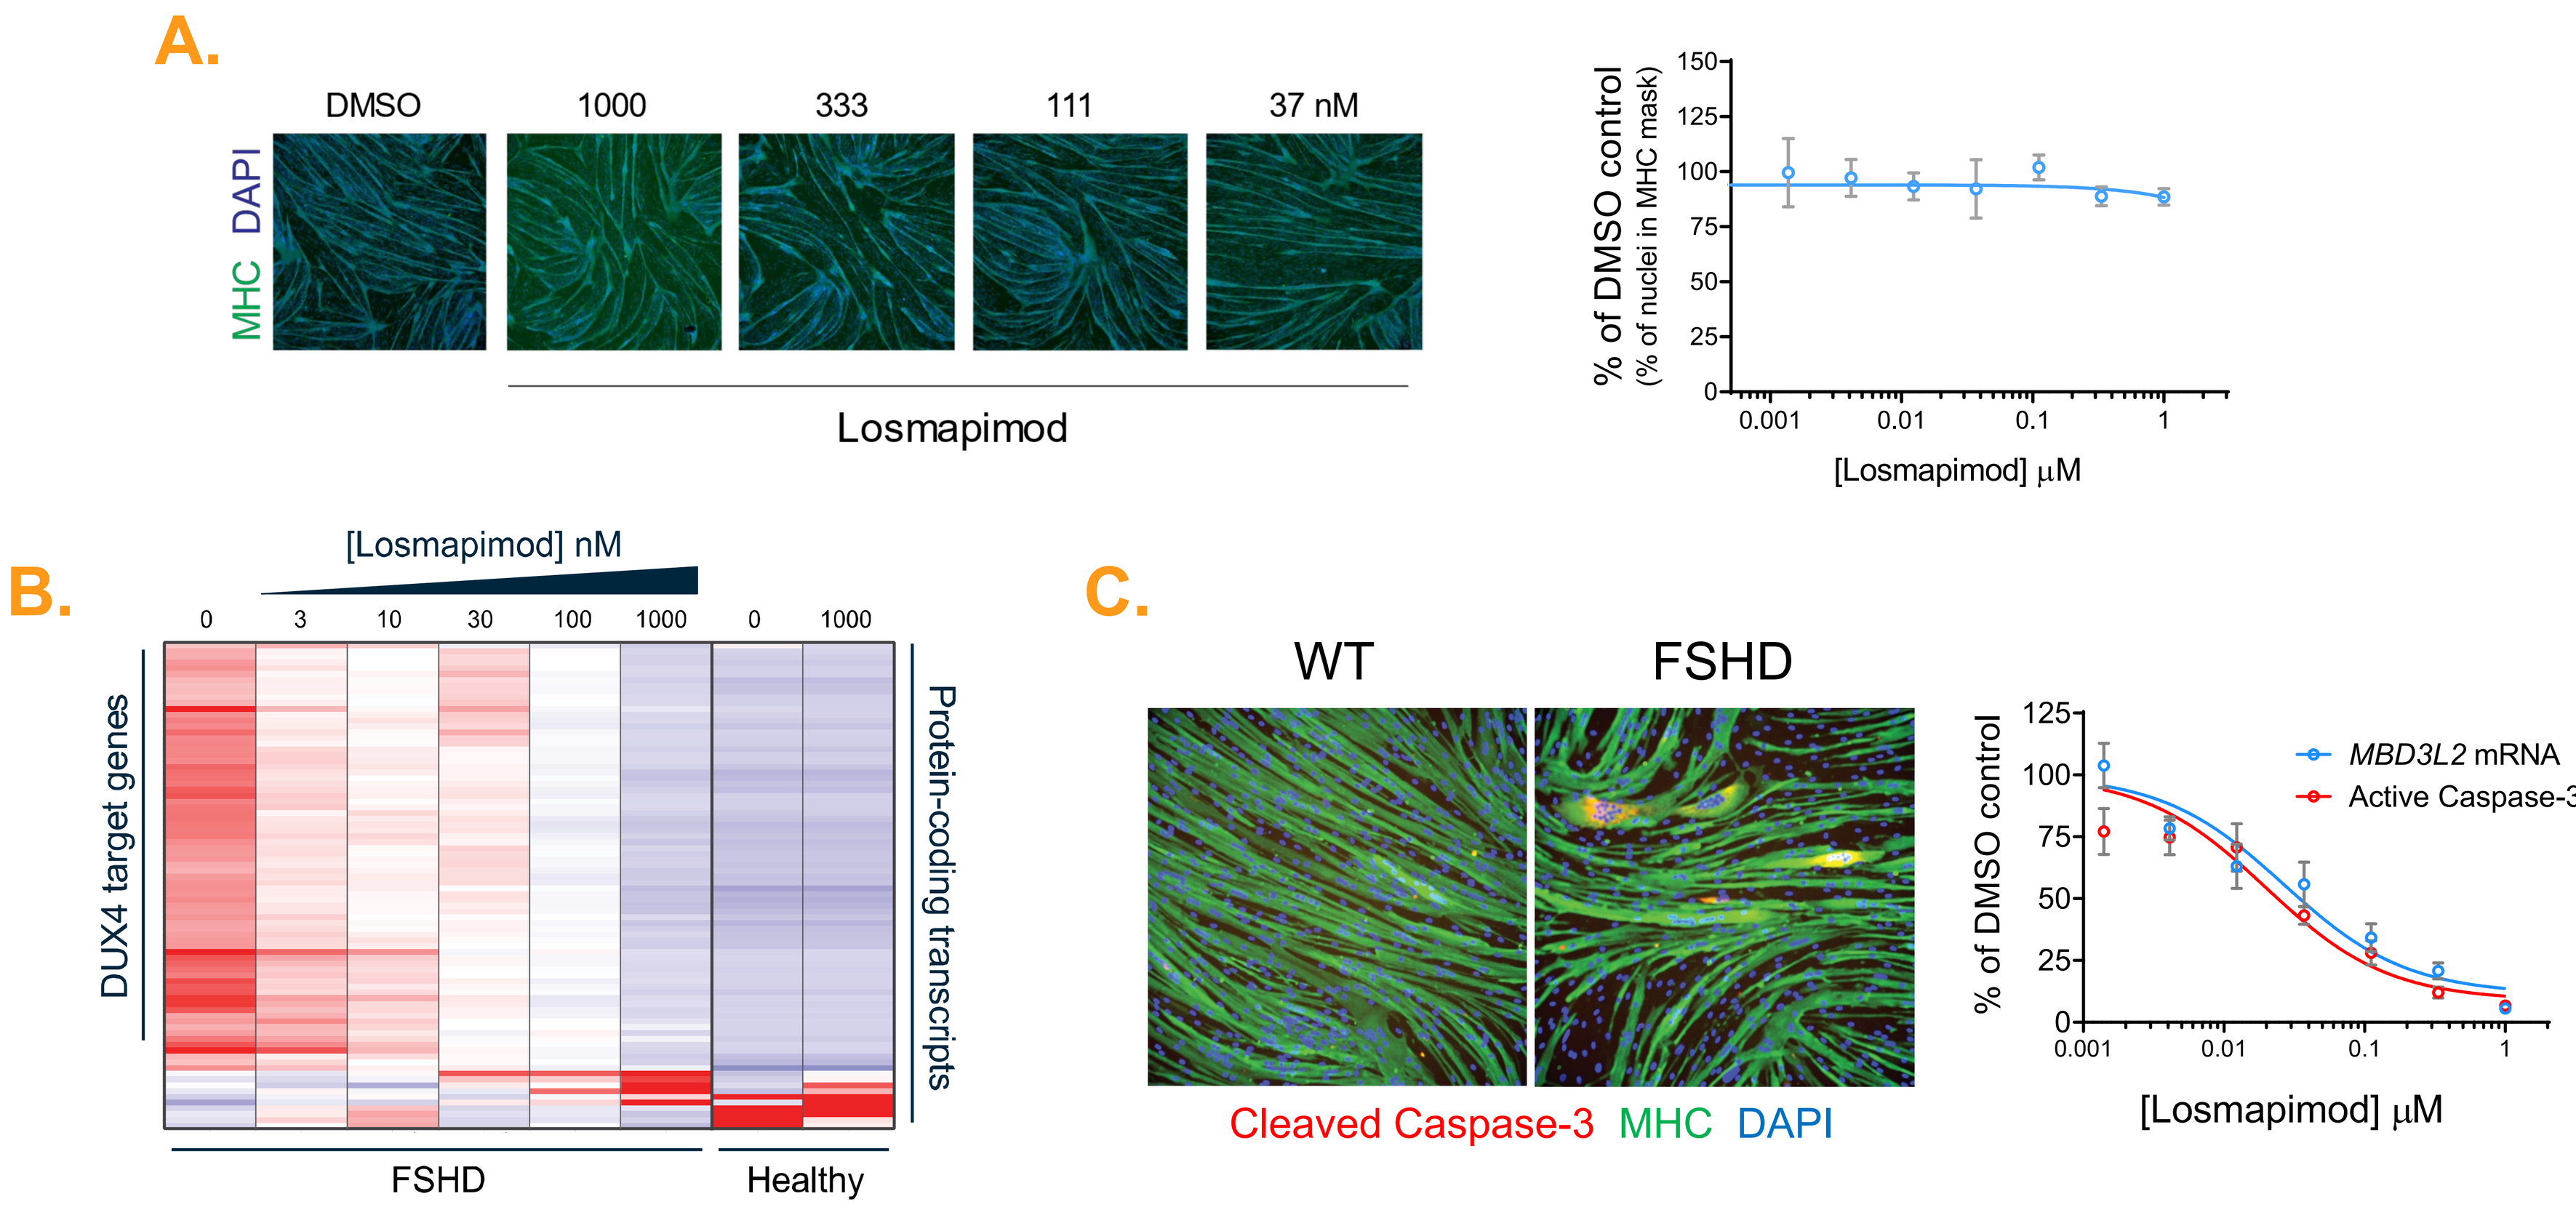
A.
DMSO
1000
333
111
37 nM
MHC DAPI
Losmapimod
% of DMSO control
(% of nuclei in MHC mask)
[Losmapimod] µM
B.
[Losmapimod] nM
DUX4 target genes
Protein-coding transcripts
FSHD
Healthy
C.
WT
FSHD
Cleaved Caspase-3 MHC DAPI
MBD3L2 mRNA
Active Caspase-3
% of DMSO control
[Losmapimod] µM

(A) Losmapimod treatment does not affect in vitro differentiation of myotubes. **(B)** RNA-seq analysis of myotubes indicates that losmapimod selectively inhibits DUX4 and its downstream program expression in a concentration dependent manner with minimal impact across the transcriptome of FHSD myotubes. <100 differentially expressed genes (abs(FC)>4; FDR<0.001) **(C)** Losmapimod reduces cleaved caspase-3 signal detected in FSHD myotubes indicating reduction of cell death.

5. DUX4 activity and apoptosis in primary myotubes

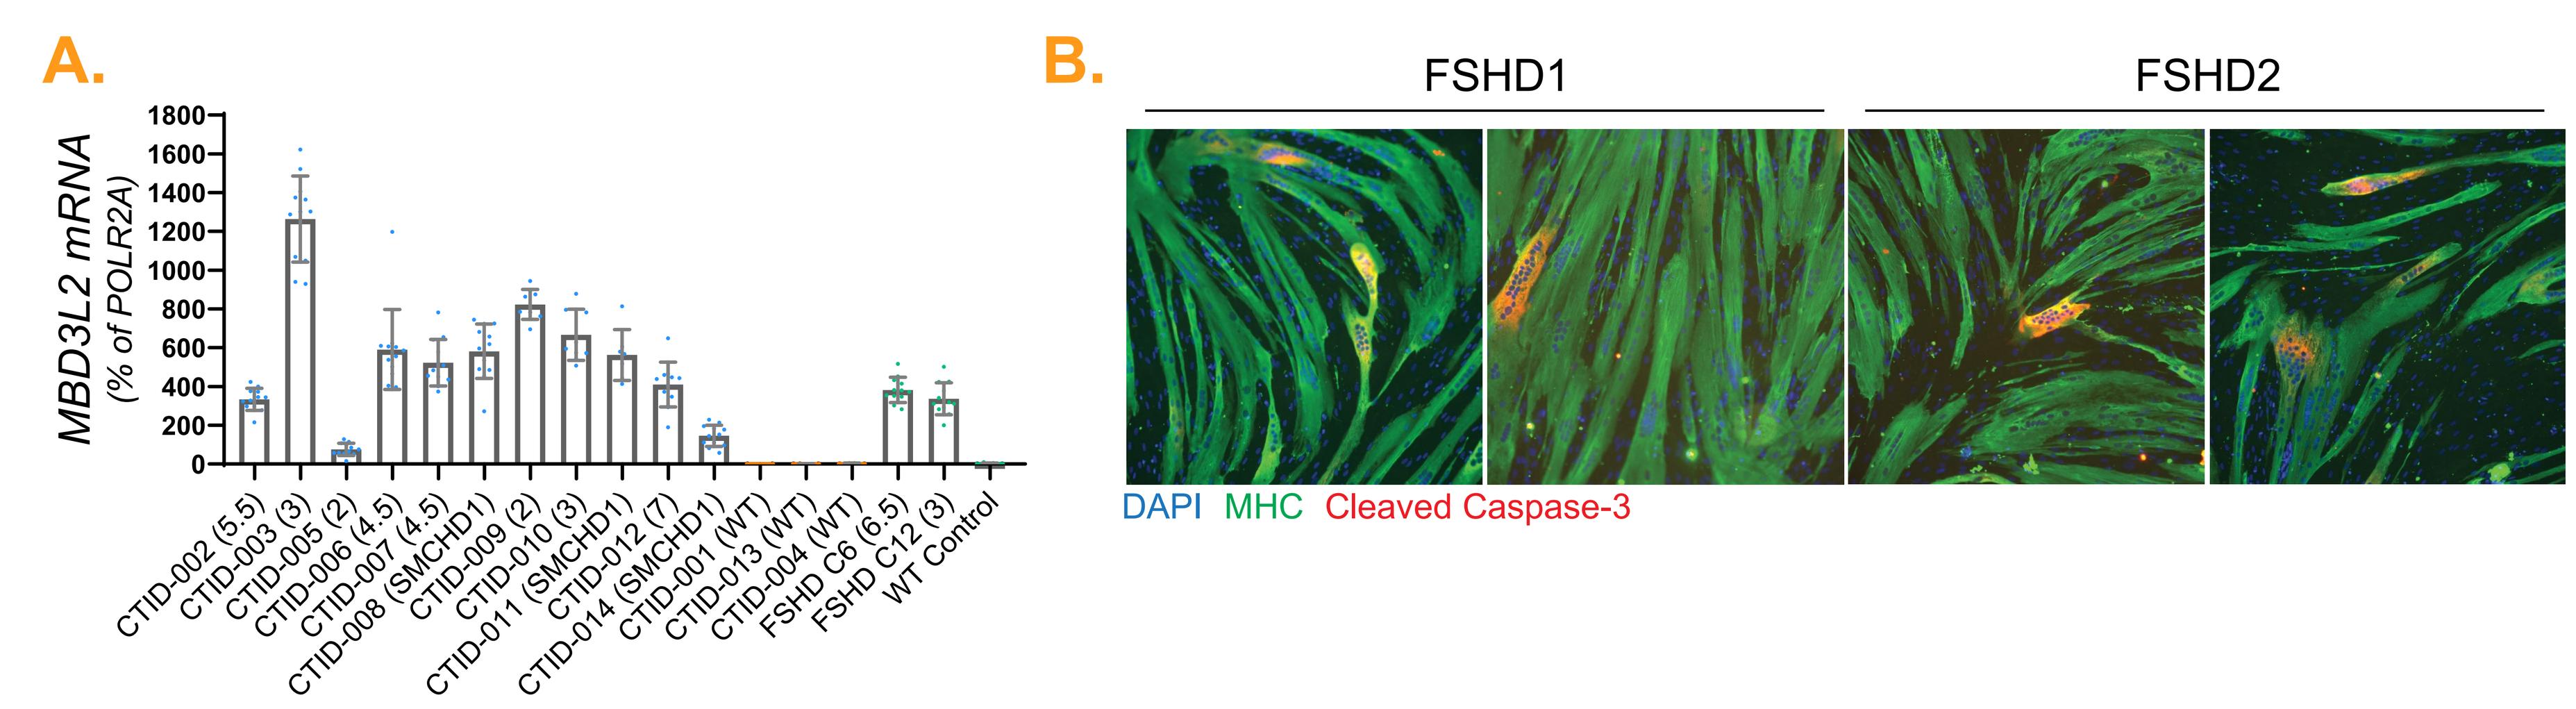
A.
MBD3L2 mRNA
(% of POLR2A)
CTID-002 (5.5)
CTID-003 (3)
CTID-005 (2)
CTID-006 (4.5)
CTID-007 (4.5)
CTID-008 (SMCHD1)
CTID-009 (2)
CTID-010 (3)
CTID-011 (SMCHD1)
CTID-012 (7)
CTID-014 (SMCHD1)
CTID-001 (WT)
CTID-013 (WT)
CTID-004 (WT)
FSHD C6 (6.5)
FSHD C12 (3)
WT Control
B.
FSHD1
FSHD2
DAPI MHC Cleaved Caspase-3

(A) *MBD3L2* expression across a variety of primary FSHD patient- and non-FSHD (WT) derived myotubes. **(B)** Detection of cleaved caspase-3 in primary FSHD1 and FSHD2 patient-derived myotubes.

6. Studying p38 inhibition across FSHD genotypes

"Clinical trial in a dish"

- 11 primary FSHD cell lines: 8 FSHD1 and 3 FSHD2.
- 3 non-FSHD cells as negative control and one immortalized FSHD cell as positive control.
- Primary Efficacy Analysis: Wilcoxon matched-pairs signed rank test was perform between different treatment groups and vehicle group samples to determine significant differences.

A.

CTID #	p/i	Cell type	Repeat #	Control/FSHD
CTID-004	primary	Myoblast	NA	Healthy Control
CTID-013	primary	Myoblast	12	Healthy Control
CTID-002	primary	Myoblast	5.5	FSHD1
CTID-003	primary	Myoblast	3	FSHD1
CTID-005	primary	Myoblast	2	FSHD1
CTID-006	primary	Myoblast	4.5	FSHD1
CTID-007	primary	Myoblast	4.5	FSHD1
CTID-009	primary	Myoblast	2	FSHD1
CTID-010	primary	Myoblast	3	FSHD1
CTID-012	primary	Myoblast	7	FSHD1
CTID-008	primary	Myoblast	SMCHD1	FSHD2
CTID-011	primary	Myoblast	SMCHD1	FSHD2
CTID-014	primary	Myoblast	SMCHD1	FSHD2
CTID-001	primary	Fibroblast	39	Negative control
CTID-015	immortalized	Myoblast	6.5	Positive FSHD control

B.

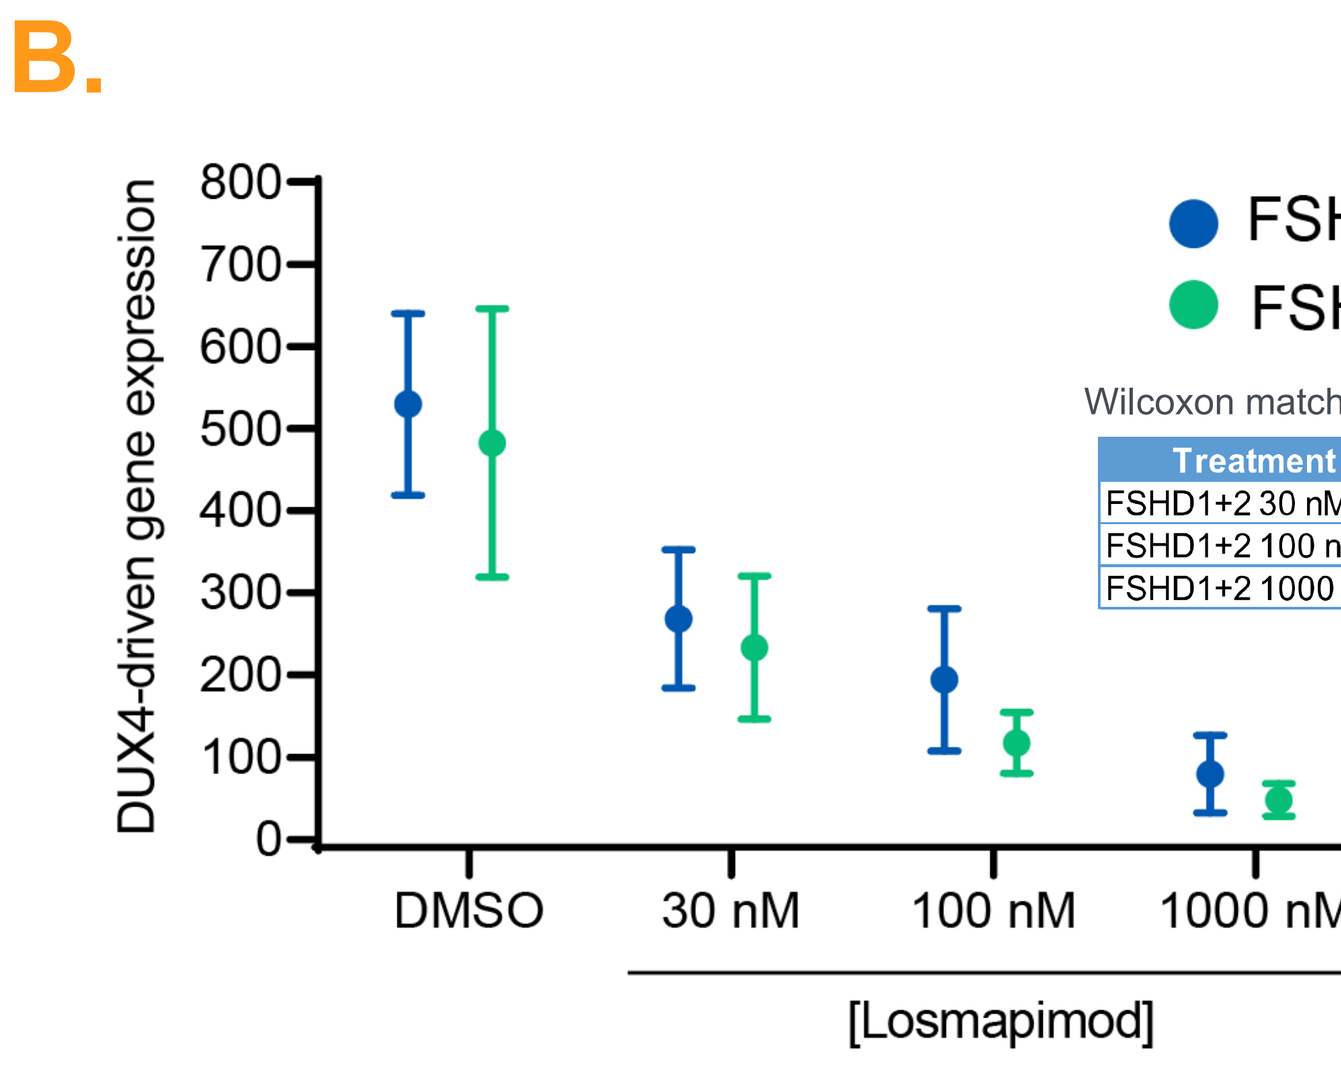
FSHD1
FSHD2
DUX4-driven gene expression
DMSO
30 nM
100 nM
1000 nM
[Losmapimod]

Wilcoxon matched-pairs signed rank test

Treatment	Difference	p-value
FSHD1+2 30 nM	-258.6	0.001
FSHD1+2 100 nM	-350.5	0.001
FSHD1+2 1000 nM	-469.9	0.001

C.

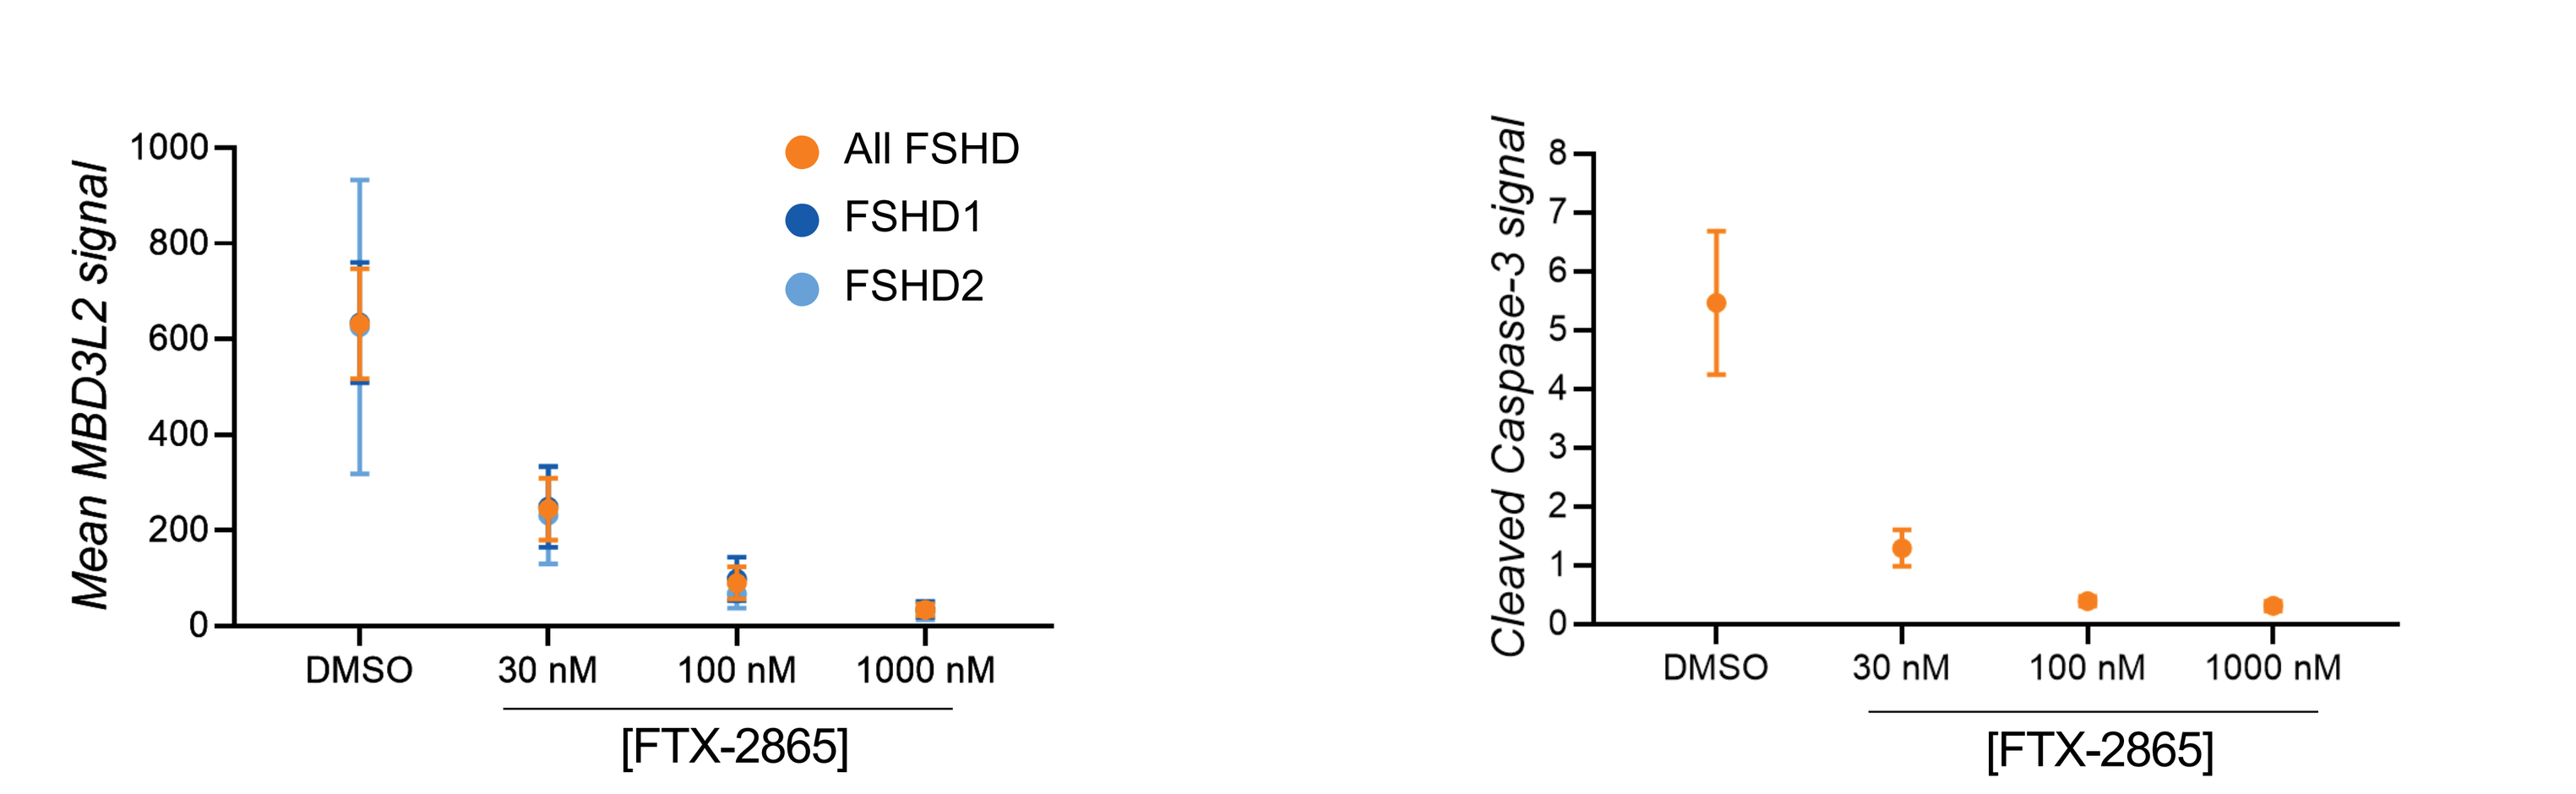
Mean MBD3L2 signal
All FSHD
FSHD1
FSHD2
Cleaved Caspase-3 signal
DMSO
30 nM
100 nM
1000 nM
[FTX-2865]

(A) Table describing the genotypes of cells used in this study. **(B)** *MBD3L2* expression was reduced across all FSHD1 and FSHD2 patient-derived myotubes after losmapimod treatment.
(C) Inhibition of p38α/β MAPK pathway using a structurally distinct inhibitor (FTX-2865) results in reduction of DUX4 expression and inhibition of cell death in vitro across FSHD1 and FSHD2 genotypes.

6. Conclusions

- Using an in vitro model of FSHD, we identify novel regulators of aberrant DUX4 expression.
- Losmapimod is a selective p38α/β inhibitor that reduces DUX4 expression in FSHD myotubes.
- Further in vitro characterization demonstrates that the DUX4-driven gene expression and cell death are inhibited in FSHD myotubes exposed to losmapimod.
- Losmapimod reduced expression of DUX4 in all FSHD1 and FSHD2 cell lines tested.
- Reduction of DUX4 expression resulted in inhibition of cell death across all genotypes tested.