An Open-Label Study of Losmapimod to Evaluate the Safety, Tolerability, and Biomarker and Clinical Outcome Assessment Changes in Subjects with FSHD1

Joost Kools[1], Nicol Voermans[1], Karlien Mul[1], Lucienne Ronco[2], John Jiang[2], Jennifer Shoskes[2], Kelly Marshall[2], Diego Cadavid[2], Michelle L. Mellion[2], Baziel van Engelen[1]
[1]Radboud University Medical Centre (Radboudumc), Nijmegen, NL, [2]Fulcrum Therapeutics, Inc., Cambridge, MA, USA

Introduction

- FSHD is a serious, rare, progressive and disabling disease, caused by the aberrant expression of DUX4 in skeletal muscle
- Stochastic DUX4 expression contributes to disease heterogeneity, asymmetry, and variability in disease phenotype
- Pathological activity and maladaptive remodeling lead to muscle fiber death and immune and fat infiltration
- Progressive muscle loss and fatty replacement causes heterogeneous, progressive descending weakness and loss of function
- Muscle pathology leads to accumulation of disability

Currently, there are with no approved disease modifying therapies for people living with FSHD that prevent and/or slow muscle wasting and weakness

Rationale

A treatment that reduces or prevents aberrant DUX4 activity in skeletal muscles may stop or prevent functional impairment and accumulation of disability and decrease/arrest replacement of muscle by fat.

- **Losmapimod** is an investigational small molecule inhibitor of p38α/β Mitogen Activated Protein Kinase (MAPK).
- Pre-clinical studies demonstrated treatment with losmapimod and other p38 inhibitors resulted in dose-dependent reduction of DUX4 protein, DUX4 transcriptional program and skeletal muscle cell death in FSHD myotubes across all genotypes tested.
- Clinical studies in over 3,600 subjects across a diversity of diseases evidenced acceptable safety and tolerability for up to one year of treatment at relevant doses.

Objective

- Evaluate the long-term safety and tolerability of losmapimod in participants with FSHD
- Evaluate on-treatment change from baseline in selected clinical outcome assessments and imaging modalities

Open-Label Study Design

- This is an open-label, single-site study of losmapimod to evaluate the safety, tolerability, and changes in biomarker and clinical outcome assessments in subjects with facioscapulohumeral muscular dystrophy 1 (FSHD1)

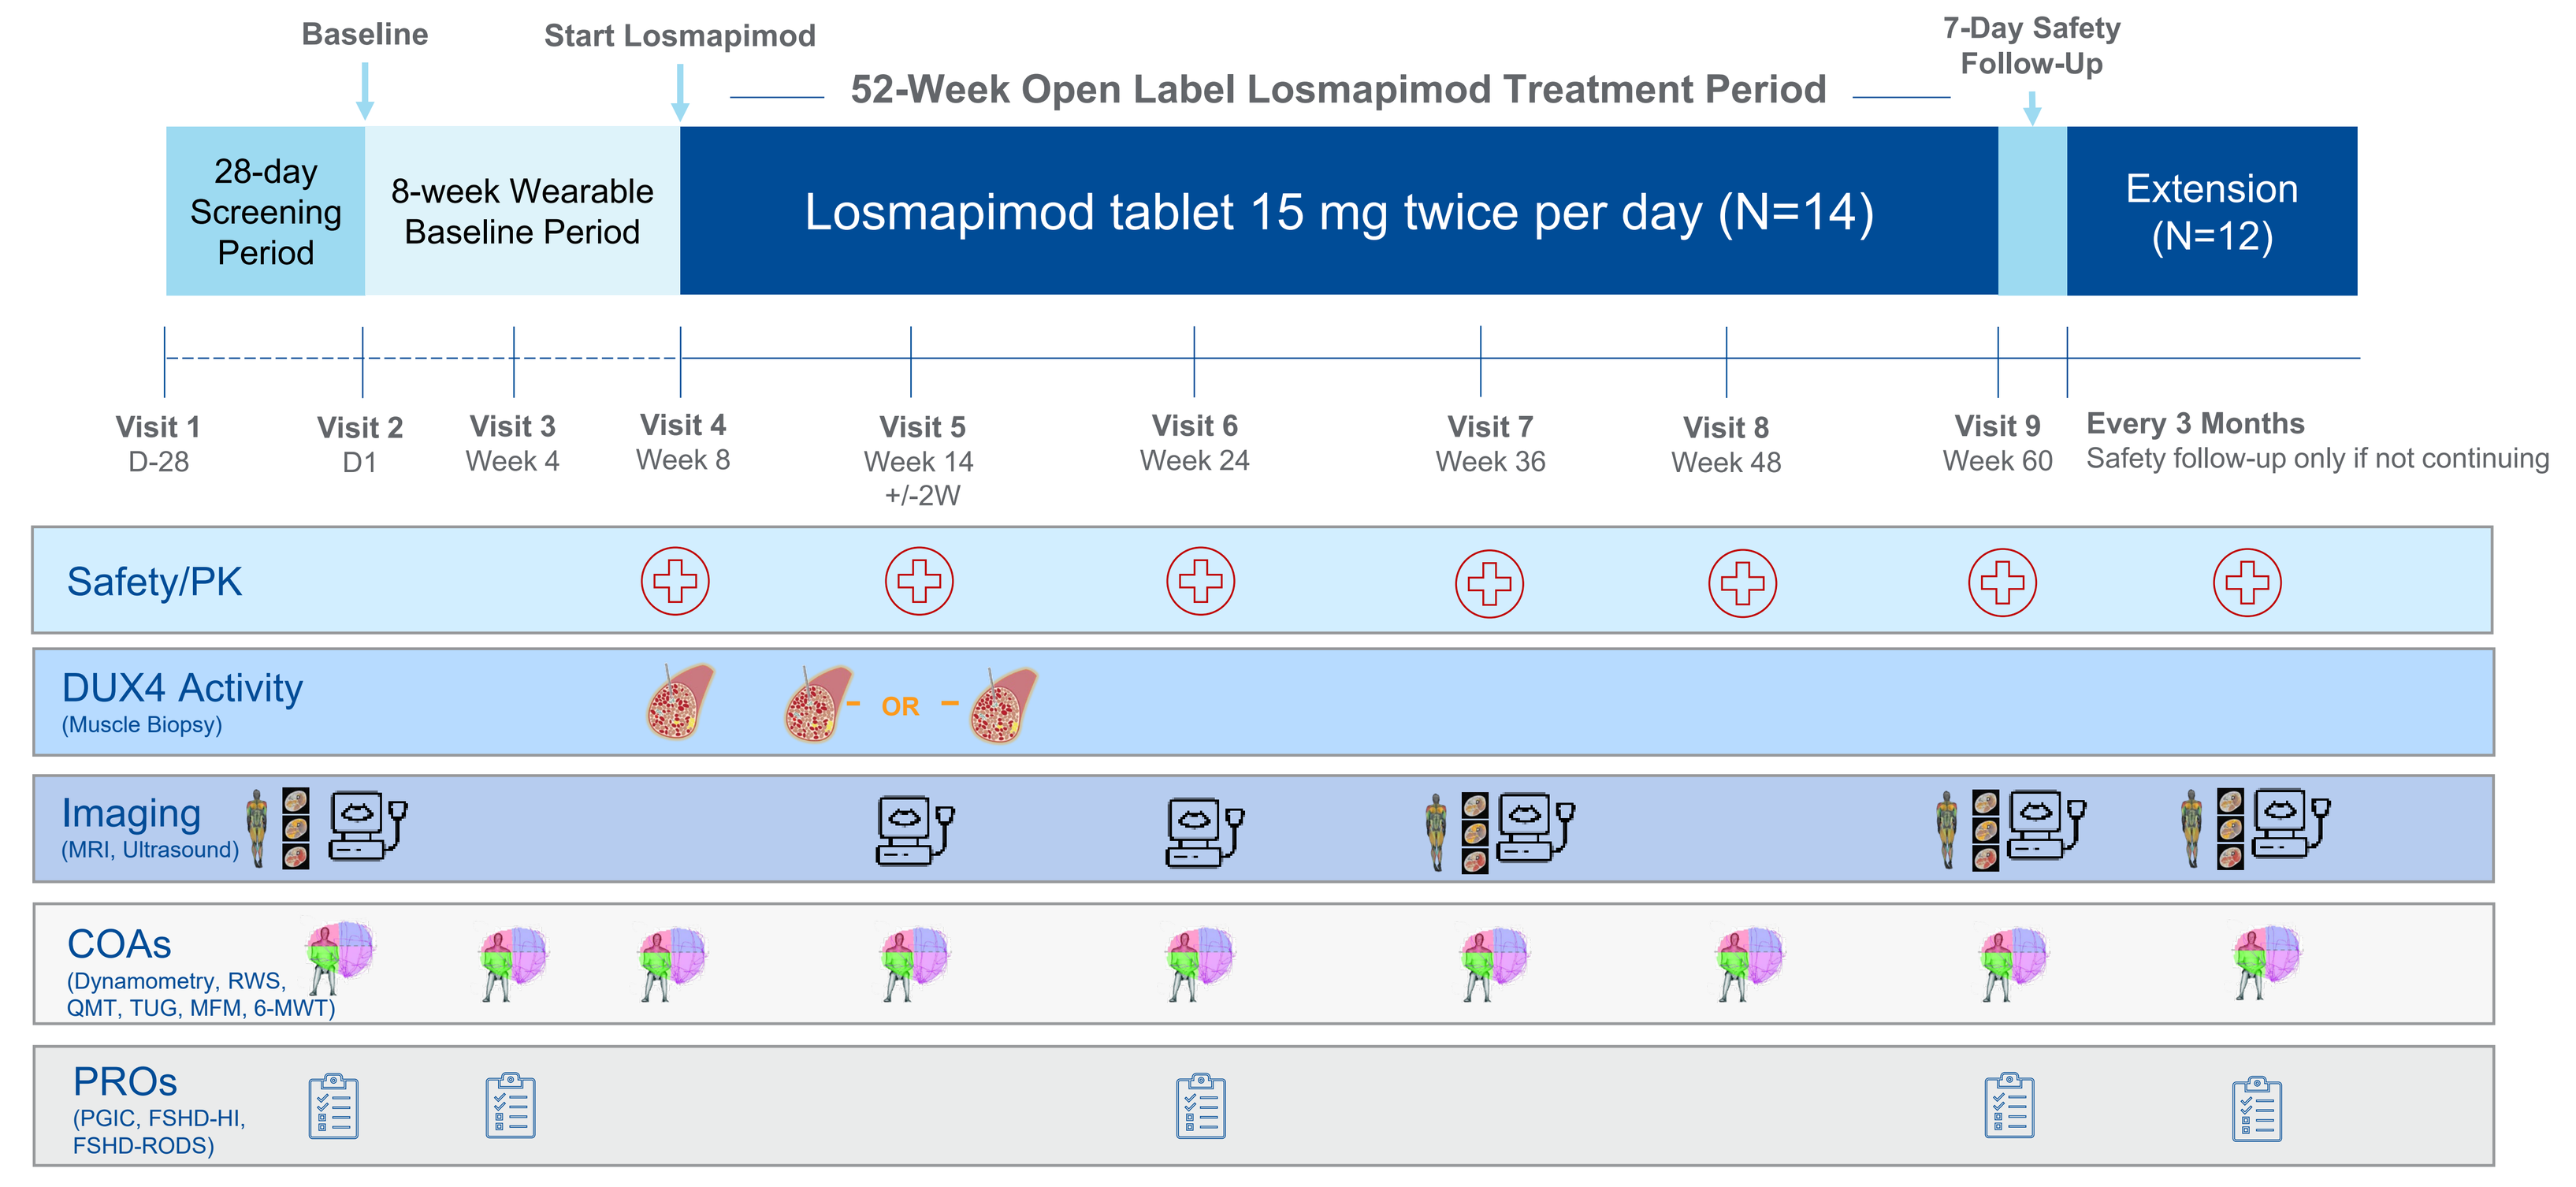
Baseline
Start Losmapimod
52-Week Open Label Losmapimod Treatment Period
7-Day Safety Follow-Up
28-day Screening Period
8-week Wearable Baseline Period
Losmapimod tablet 15 mg twice per day (N=14)
Extension (N=12)
Visit 1 D-28
Visit 2 D1
Visit 3 Week 4
Visit 4 Week 8
Visit 5 Week 14 +/-2W
Visit 6 Week 24
Visit 7 Week 36
Visit 8 Week 48
Visit 9 Week 60
Every 3 Months Safety follow-up only if not continuing
Safety/PK
DUX4 Activity (Muscle Biopsy)
OR
Imaging (MRI, Ultrasound)
COAs (Dynamometry, RWS, QMT, TUG, MFM, 6-MWT)
PROs (PGIC, FSHD-HI, FSHD-RODS)

Muscle biopsies were to be preformed at baseline, pre-treatment (Visit 4, Week 8), and on-treatment 4 or 8 weeks later (Visit 5, Week 14 ± 2 weeks) Due to COVID-19, visit windows were extended

Main Inclusion Criteria:	Main Exclusion Criteria:
▪ Age 18-65 years ▪ Genetically confirmed diagnosis of FSHD1 ▪ Ricci score 2-4 ▪ STIR+ muscle, as determined by a central reader, safely accessible by needle biopsy	▪ Medical conditions that can confound results of the study ▪ Contraindication to MRI ▪ Contraindication to muscle biopsy

Study Endpoints

1. Safety and Tolerability
2. Pharmacodynamics in muscle and blood and Pharmacokinetics in plasma and muscle
3. Exploratory Endpoints – see right

Exploratory Endpoints

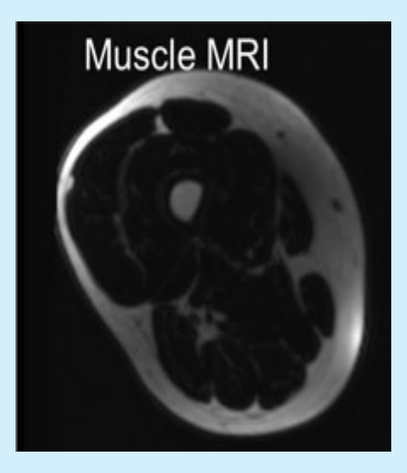
Muscle MRI

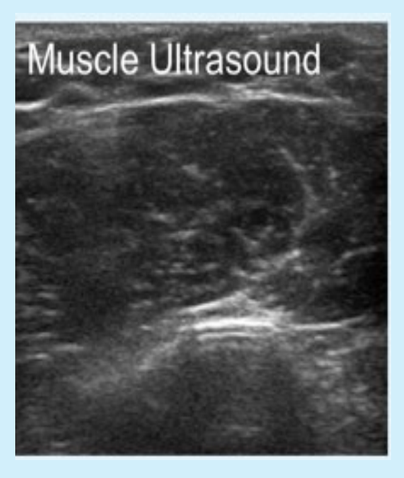
Muscle Ultrasound

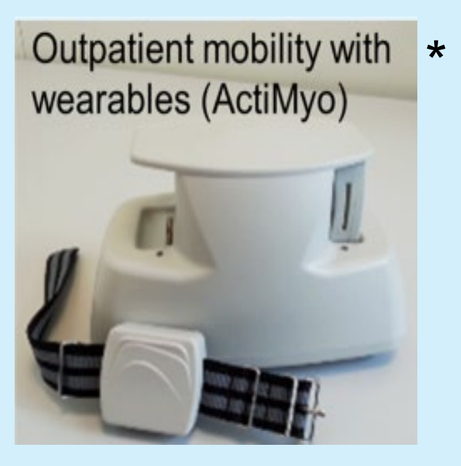
Outpatient mobility with wearables (ActiMyo)

*

Reachable Workspace

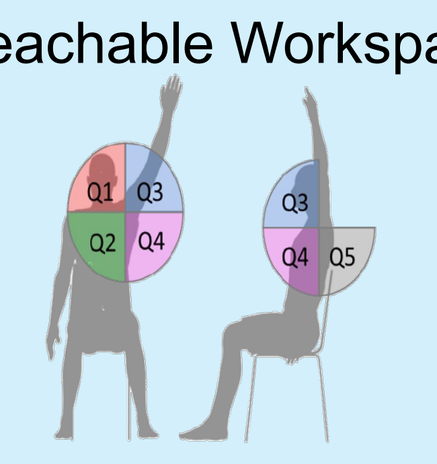
Q1
Q2
Q3
Q4
Q5

Timed Up and Go (TUG) FSHD-TUG

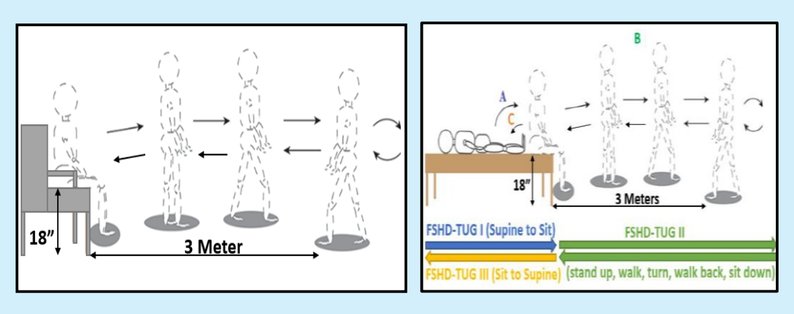
3 Meter
3 Meters

Other:
- Spirometry
- 6-Minute Walk Test
- MFM Domain 1

Quantitative Myometry

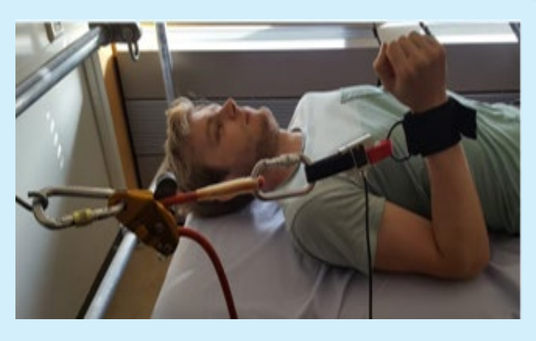

Handheld Dynamometry

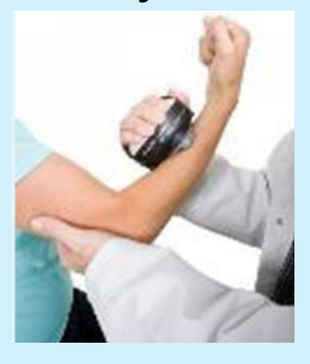

PROs:
- FSHD-HI
- FSHD-RODS
- PGIC

NCT04004000 *outpatient wearables to be presented separately

Results

Baseline and Demographic Information

		Losmapimod 15 mg BID (N=14)
Age (years)	Mean (SD)	45.7 (11.12)
Race, White	n (%)	13 (92.9)
Body Mass Index (BMI) (kg/m²)	Mean (SD)	24.0 (2.94)
D4Z4 Repeat Category, n (%)	1-3 Repeats	3 (21.4)
	4-9 Repeats	11 (78.6)
Ricci Score, n (%)	2	0
	2.5	1 (7.1)
	3	5 (35.7)
	3.5	2 (14.3)
	4	6 (42.9)
Enrolled	n (%)	14
Completed the study	n (%)	14 (100)
Discontinued from study	n (%)	0
Entered extension	n (%)	12 (85.7)

- All subjects completed the study
- 2 subjects declined participation in the extension study for reasons unrelated to study drug/adverse events

Safety and Tolerability

	Overall (N=14) n (%)
Any TEAE	14 (100)
Any treatment-related TEAE	14 (100)
Any serious adverse event (SAE)	0
Any TEAE leading to treatment discontinuation	0
Any TEAE leading to death	0
AE by Maximum Severity	
Mild	4 (28.6)
Moderate	5 (35.7)
Severe	5 (35.7)
Most Common AEs (>25%)	
Alanine aminotransferase (ALT) increased*	5 (35.7)
Dry skin	4 (28.6)
Myalgia	4 (28.6)

- *ALT increases were mild and transient – All cases resolved with continued dosing, and none led to treatment discontinuation
- No significant changes in vital signs, laboratory studies or EKG were observed
- Nine subjects (64.3%) reported events in the SOC of Skin and Subcutaneous tissue disorders

Results (cont'd)

Pharmacodynamics and Pharmacokinetics

- Blood and muscle concentrations were within the expected range based on pre-clinical data
- Target engagement in blood was within the expected range (~40% to 55% change from baseline at Cmax)

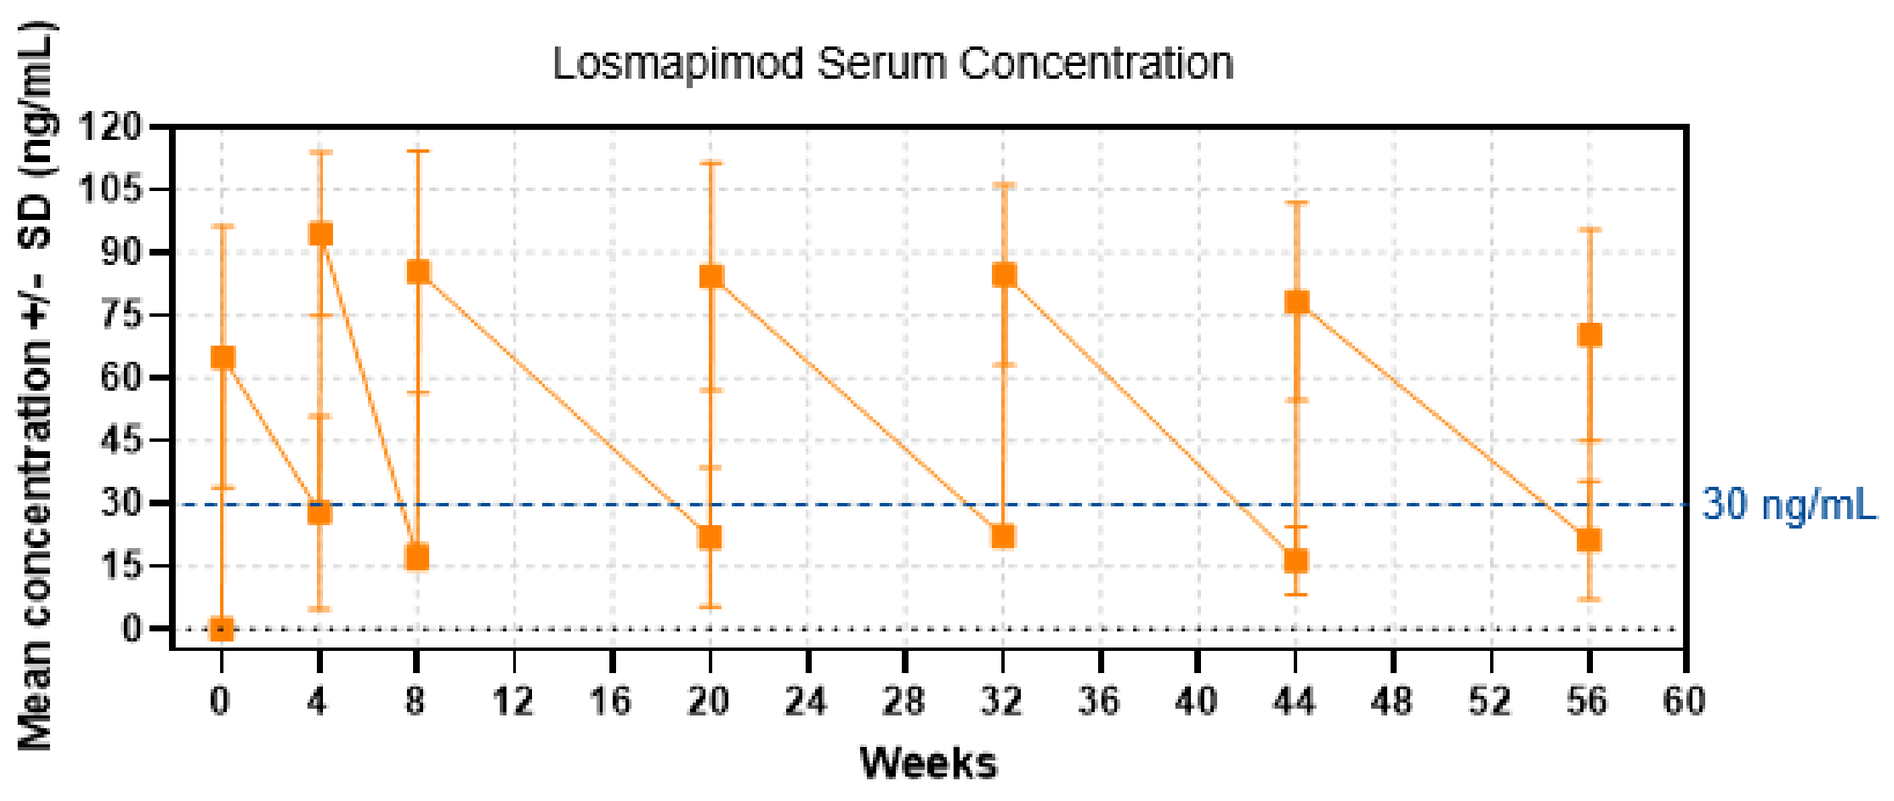
Losmapimod Serum Concentration
Mean concentration +/- SD (ng/mL)
Weeks
30 ng/mL

DUX4 Activity

- Changes from baseline were not observed
- DUX4-driven gene expression was highly variable

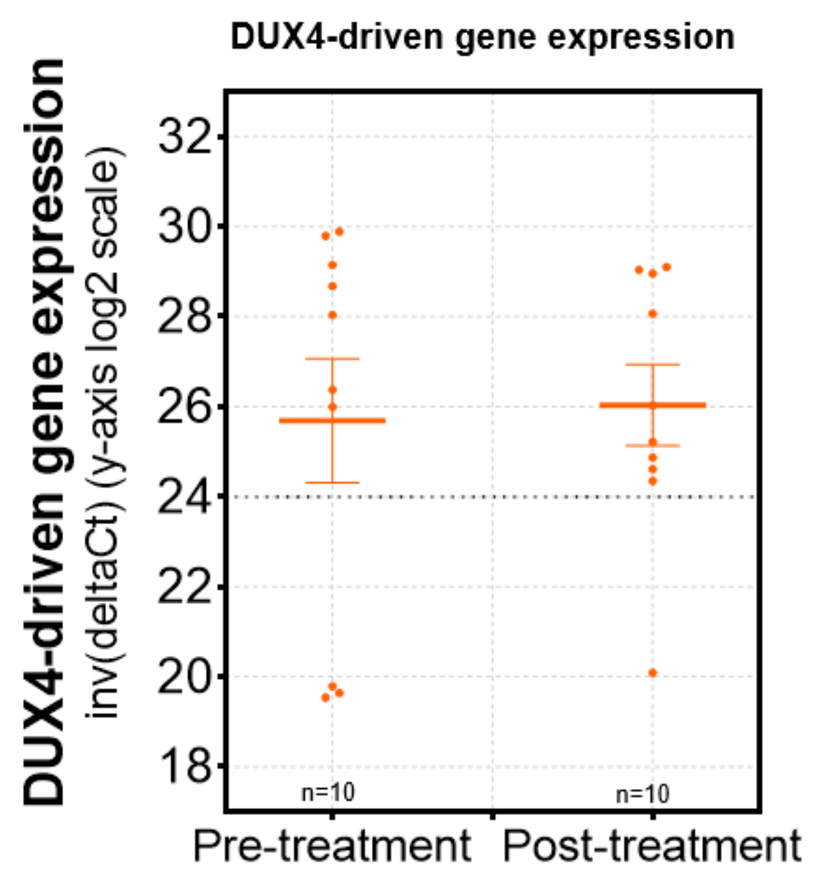
DUX4-driven gene expression
DUX4-driven gene expression
inv(deltaCt) (y-axis log2 scale)
n=10
n=10
Pre-treatment
Post-treatment

Whole Body MSK MRI

- Essentially no change was observed in this population. This result is likely related to the small sample size of this study and broad range of disease severity with more severe disease (Ricci 4) in nearly half of the population.

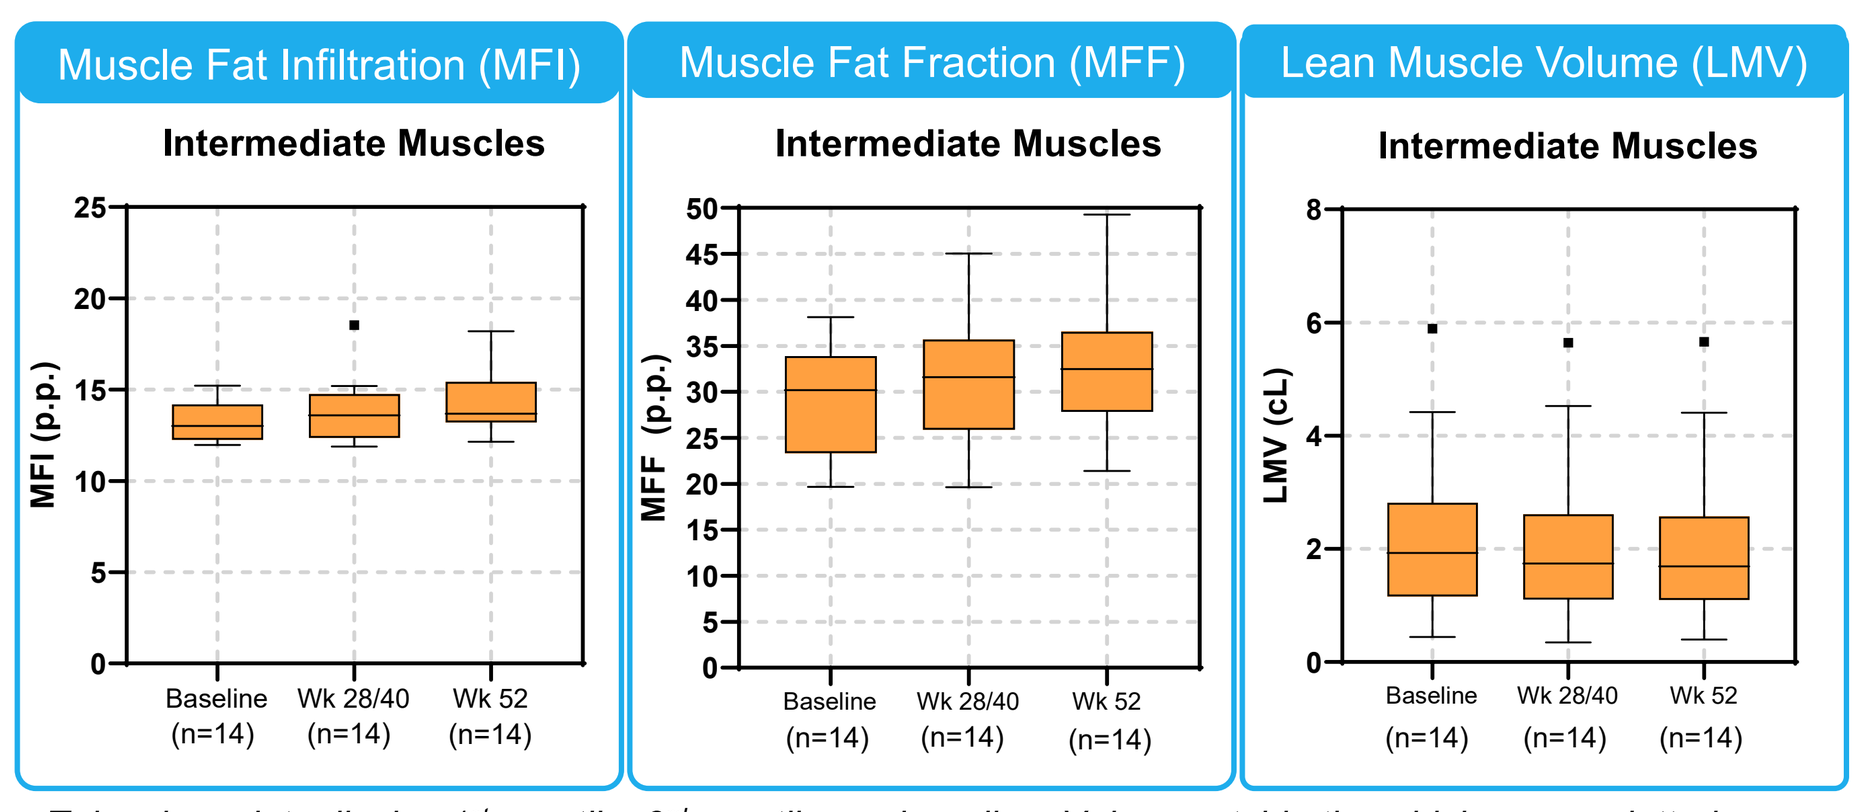
Muscle Fat Infiltration (MFI)
Muscle Fat Fraction (MFF)
Lean Muscle Volume (LMV)
Intermediate Muscles
MFI (p.p.)
MFF (p.p.)
LMV (cL)
Baseline (n=14)
Wk 28/40 (n=14)
Wk 52 (n=14)

Tukey box plots display 1st quartile, 3rd quartile, and median. Values outside the whiskers are plotted as individual points. MSK: musculoskeletal

Cross-sectional Composite Correlations

- Moderate and strong correlations between MRI and TUG and FSHD-TUG at Week 52 (n=14)
- Moderate correlations to Total RWS for LMV and MFF at Week 52 (n=14)

MRI Composite	Statistic	TUG Cross-sectional	FSHD-TUG Cross-sectional
LMV (L)	r (p-value)	-0.89 (<0.0001)	-0.83 (0.0002)
MFF (%)	r (p-value)	0.86 (<0.0001)	0.77 (0.0003)
MFI (%)	r (p-value)	0.77 (0.0014)	0.77 (0.0014)
MRI Composite	**Statistic**	**Dom Total RSA Weighted**	**Non-Dom Total RSA weighted**
LMV (L)	r (p-value)	0.55 (0.0666)	0.43 (0.1591)
MFF (%)	r (p-value)	-0.66 (0.0199)	-0.52 (0.0800)
MFI (%)	r (p-value)	-0.24 (0.4568)	-0.09 (0.7787)

Dom = dominant; Non-Dom = non-dominant; RSA = relative surface area

Muscle Ultrasound Echogenicity

- Various structural changes in the muscle, such as fatty infiltration, fibrosis, or edema, produce an increase in echogenicity
 - Echogenicity is expressed as z-score compared to reference healthy control populations
- Most muscles demonstrated stability or improvement over 52 weeks

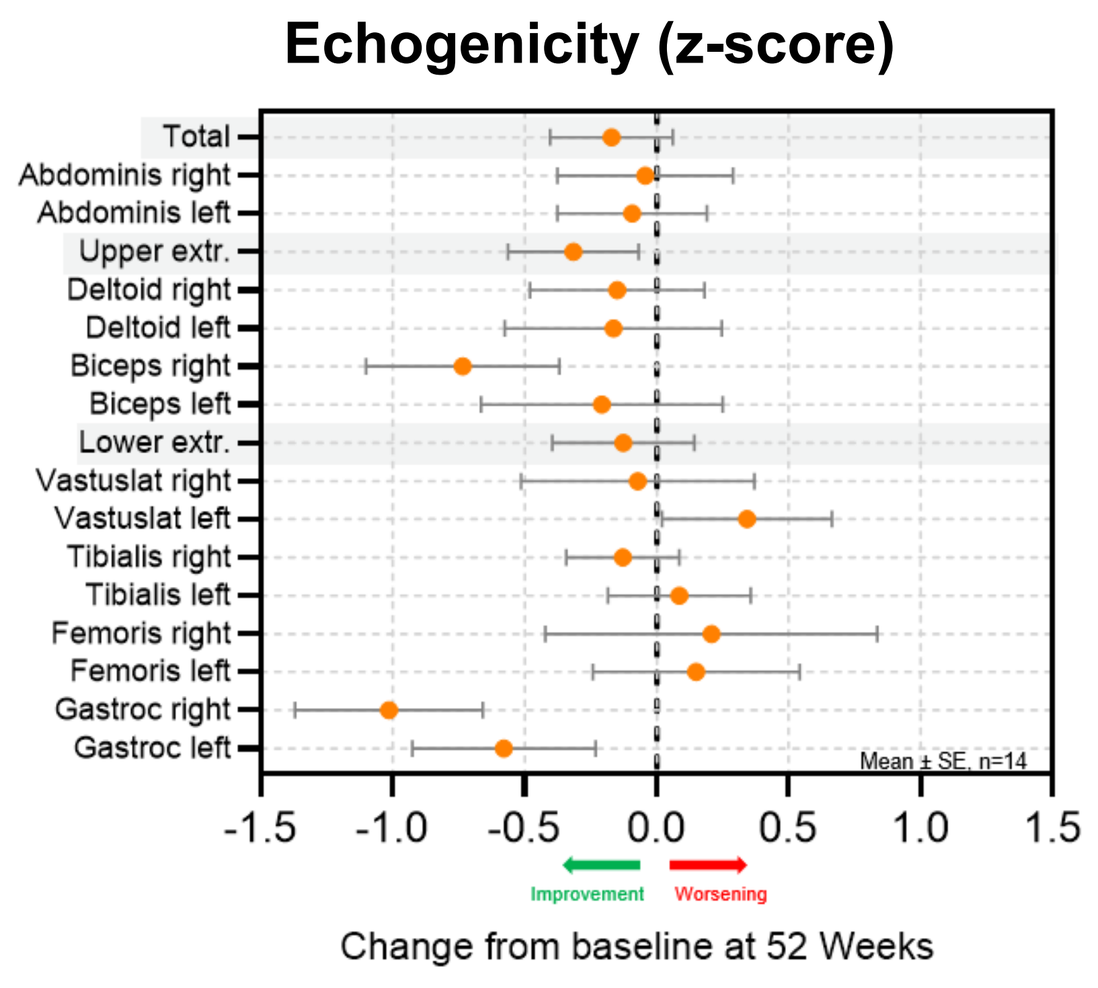
Echogenicity (z-score)
Total
Abdominis right
Abdominis left
Upper extr.
Deltoid right
Deltoid left
Biceps right
Biceps left
Lower extr.
Vastuslat right
Vastuslat left
Tibialis right
Tibialis left
Femoris right
Femoris left
Gastroc right
Gastroc left
Mean ± SE, n=14
Improvement
Worsening
Change from baseline at 52 Weeks

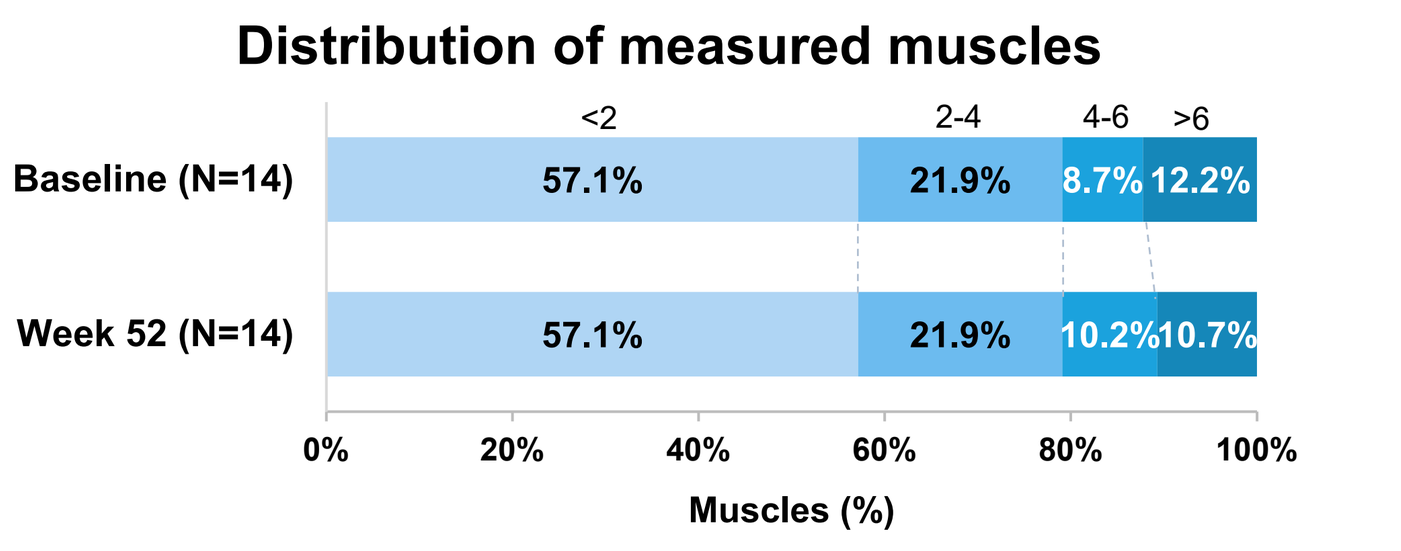
Distribution of measured muscles
<2
2-4
4-6
>6
Baseline (N=14)
57.1%
21.9%
8.7%
12.2%
Week 52 (N=14)
57.1%
21.9%
10.2%
10.7%
Muscles (%)

- Z-scores <2 are considered normal, z-scores >2 are considered abnormal
- The distribution of the z-scores of muscles at baseline vs the last visit stayed the same or decreased

Reachable Workspace (RWS)

- Losmapimod treated patients in OLS showed improvements in total RSA (Q1-Q5) in both weighted and non-weighted measurements in both dominant and non-dominant arms, with the largest improvements in the upper quadrants (Q1 and Q3)
- Slowing of disease progression and/or improvement was observed on multiple RWS metrics, including annualized RWS

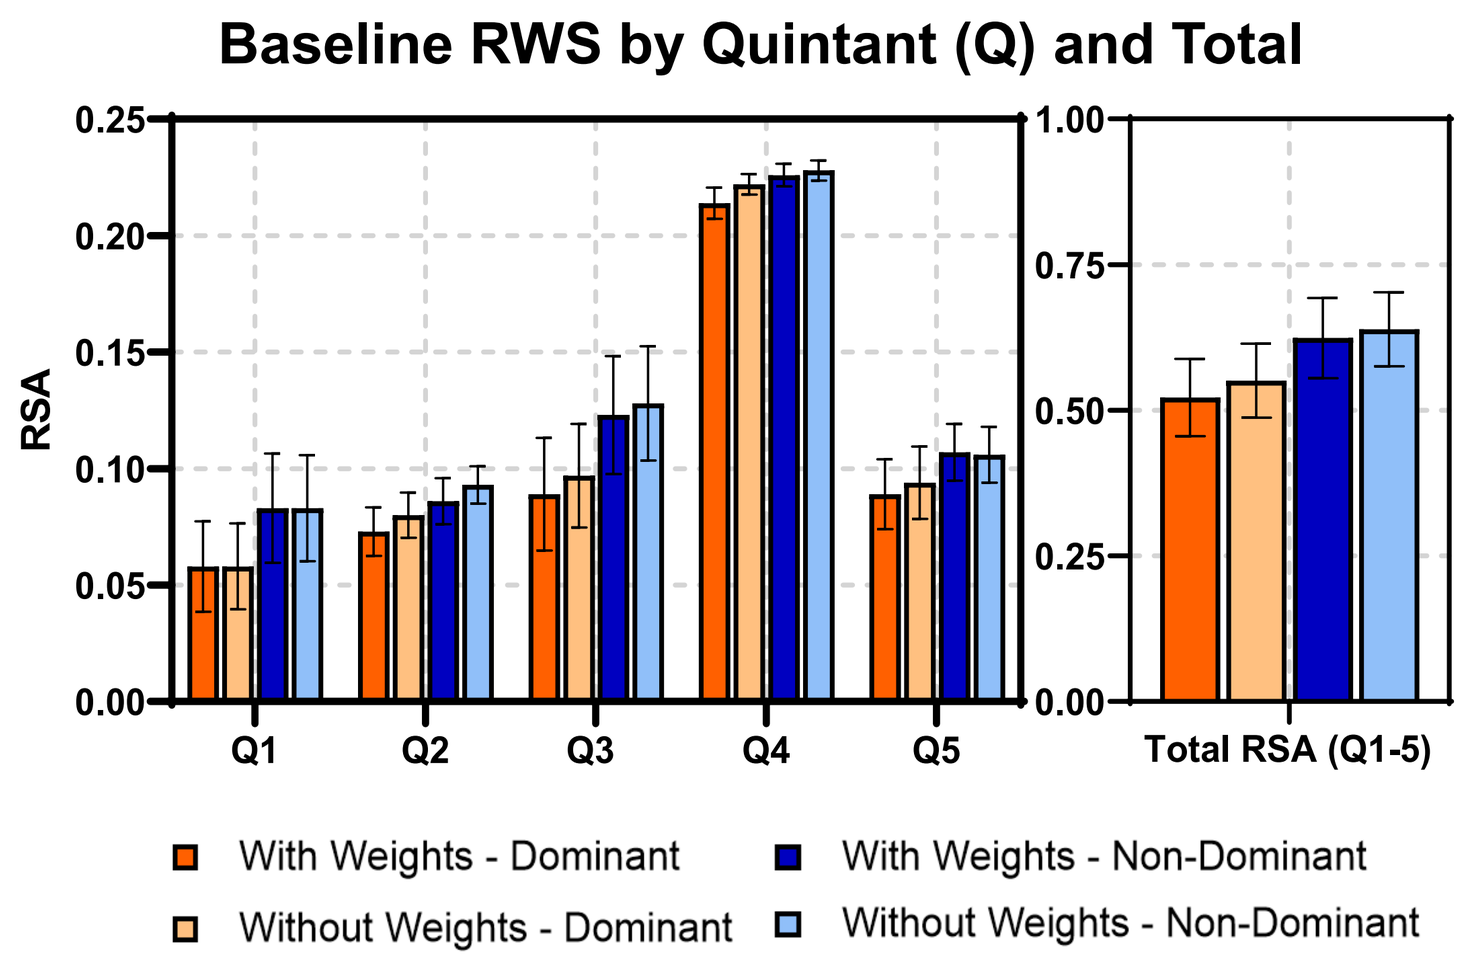
Baseline RWS by Quintant (Q) and Total
RSA
Q1
Q2
Q3
Q4
Q5
Total RSA (Q1-5)
With Weights - Dominant
With Weights - Non-Dominant
Without Weights - Dominant
Without Weights - Non-Dominant

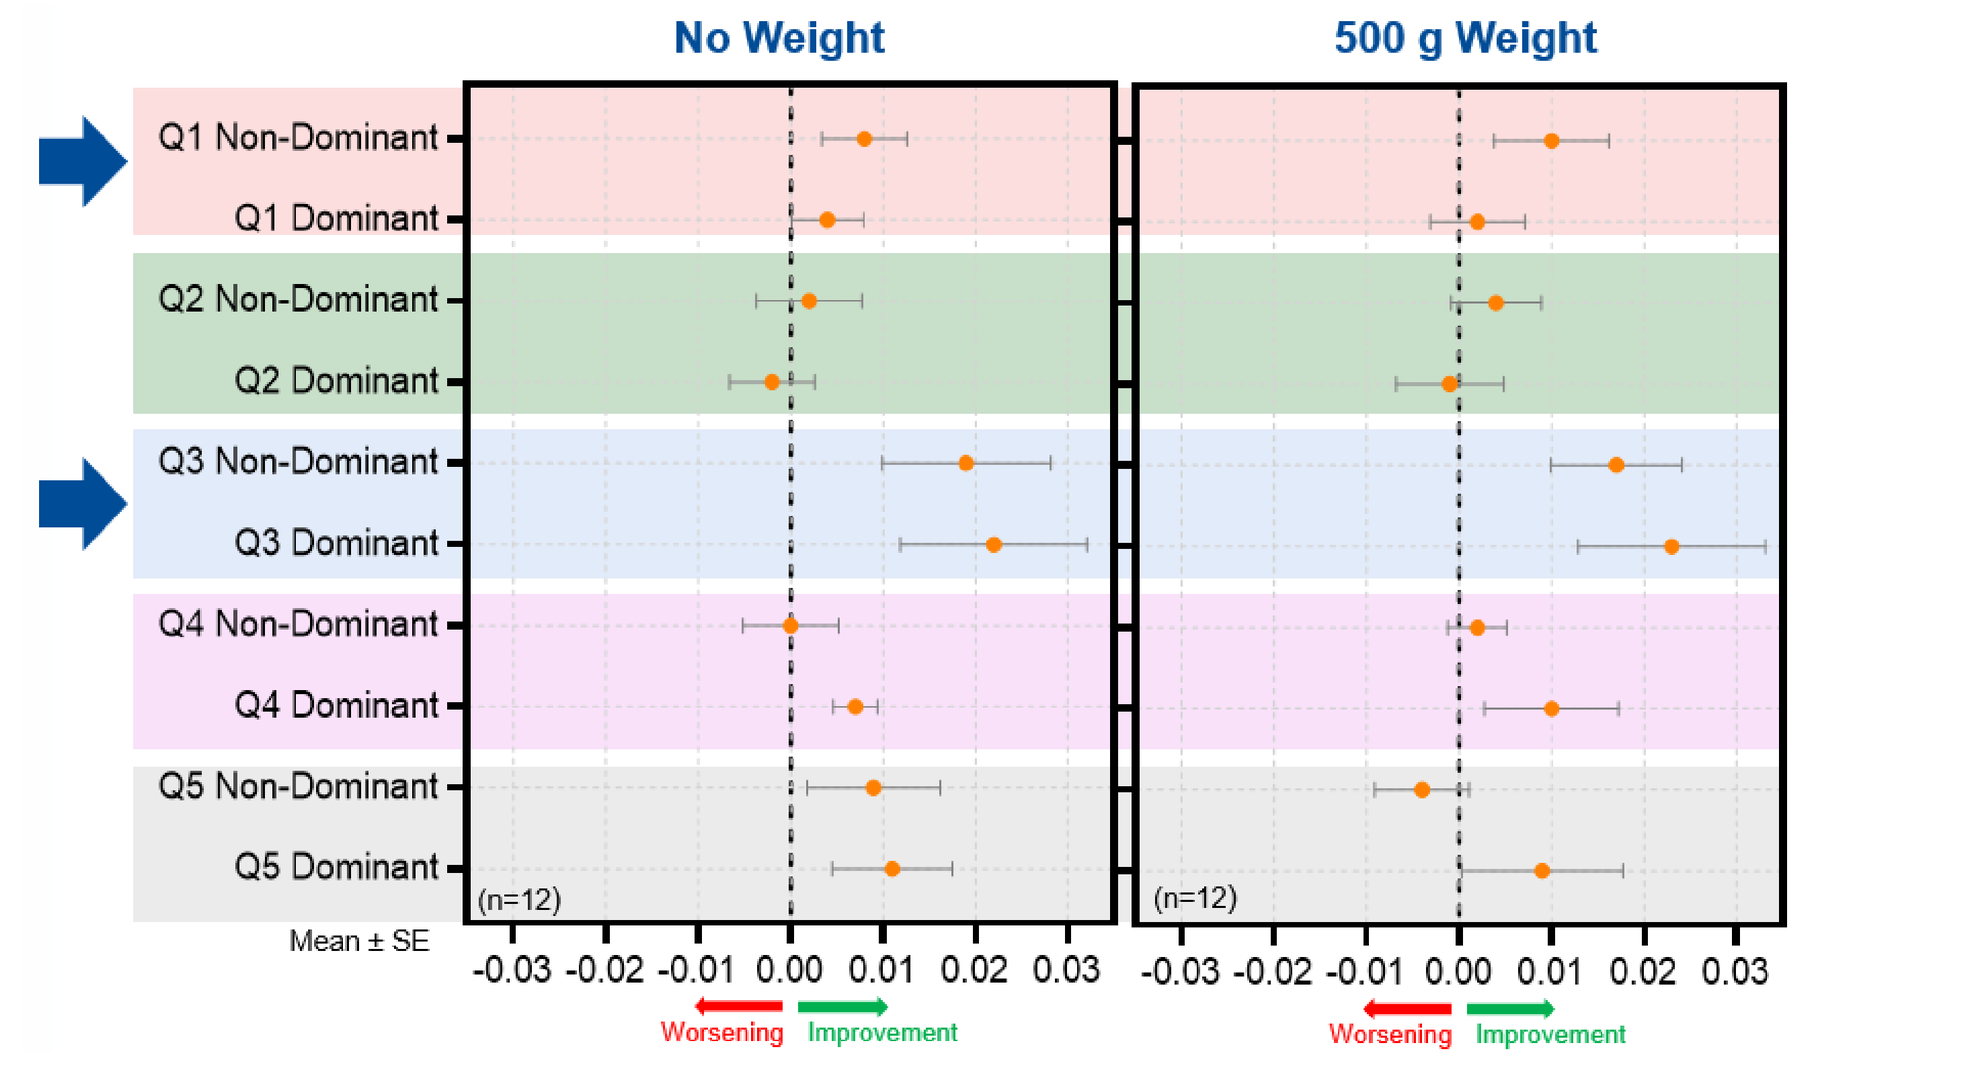
No Weight
500 g Weight
Q1 Non-Dominant
Q1 Dominant
Q2 Non-Dominant
Q2 Dominant
Q3 Non-Dominant
Q3 Dominant
Q4 Non-Dominant
Q4 Dominant
Q5 Non-Dominant
Q5 Dominant
(n=12)
Mean ± SE
Worsening
Improvement

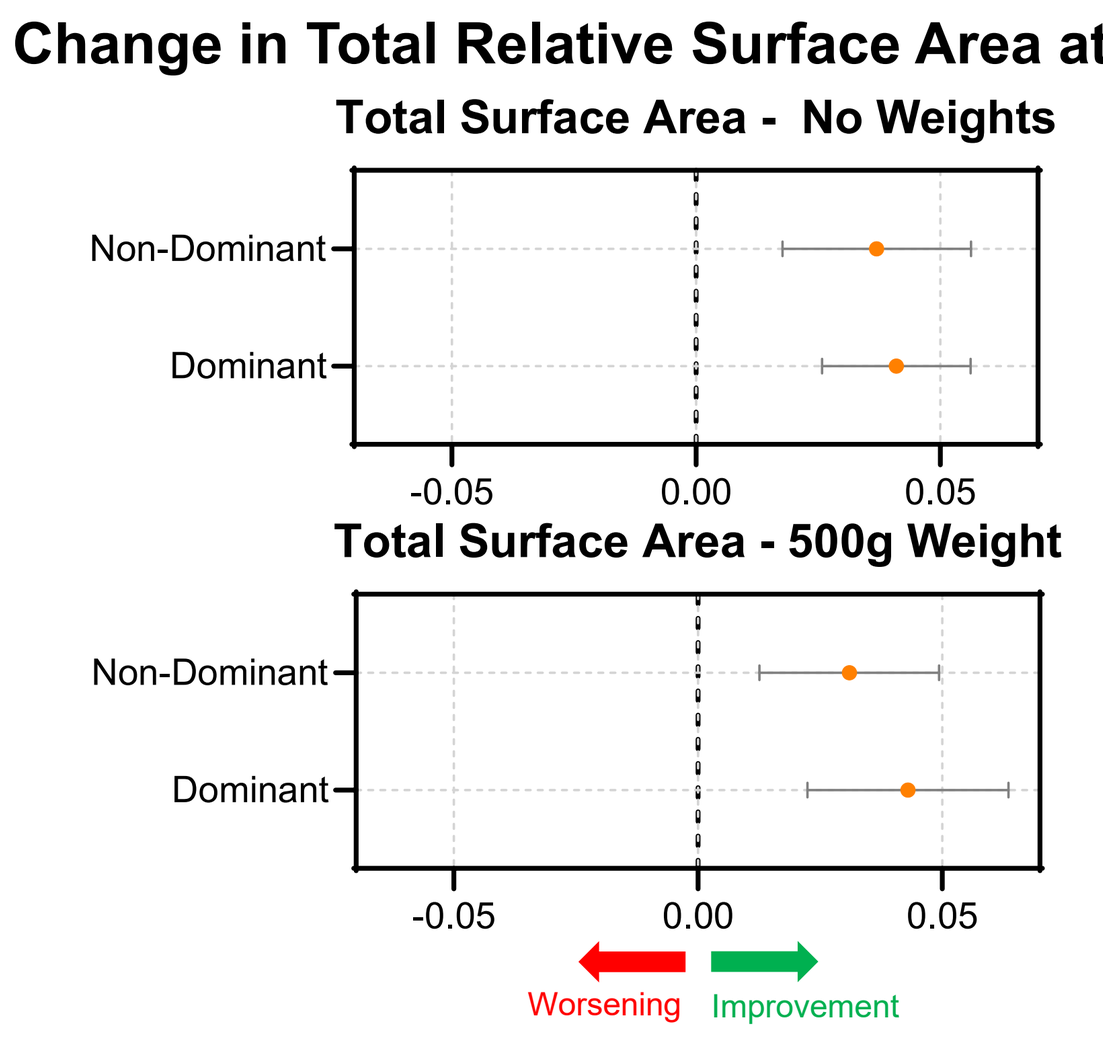
Change in Total Relative Surface Area at 52 Weeks
Total Surface Area - No Weights
Non-Dominant
Dominant
Total Surface Area - 500g Weight
Worsening
Improvement

Annualized Reachable Workspace (percent change from baseline per year)

	Without Weights		With Weights	
Quintant (Q)	**Dom**	**Non-Dom**	**Dom**	**Non-Dom**
Q1	11.95	7.73	9.16	8.60
Q2	-3.39	1.36	-0.28	1.34
Q3	16.05	9.37	16.66	10.72
Q4	1.10	0.18	2.23	0.58
Q5	4.44	6.25	4.49	-2.11
Q1+Q3	14.53	8.58	13.92	9.79
Total RSA	4.90	4.28	5.68	3.28

Dom = dominant; Non-Dom = non-dominant; RSA = relative surface area

Patient Global Impression of Change (PGIC)

- Over 80% of subjects reported improvement or no change after 52 weeks of treatment; no patients reported feeling much worse over 52 weeks

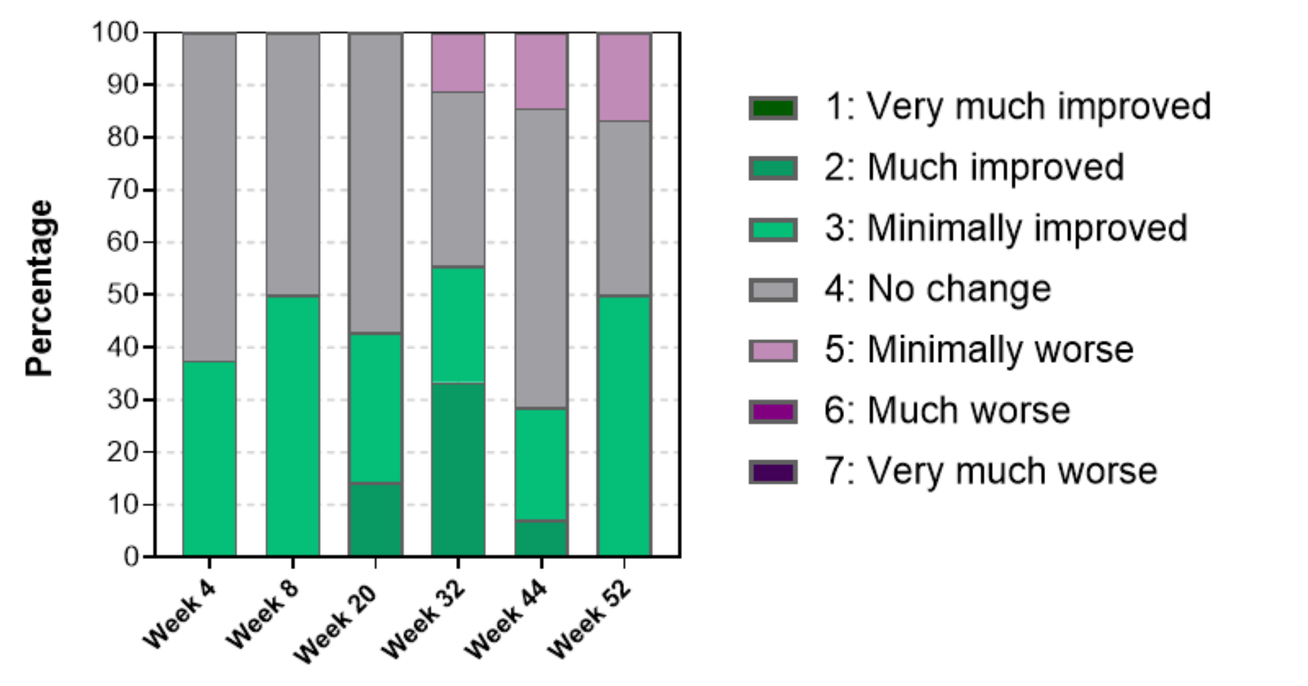
Percentage
Week 4
Week 8
Week 20
Week 32
Week 44
Week 52
1: Very much improved
2: Much improved
3: Minimally improved
4: No change
5: Minimally worse
6: Much worse
7: Very much worse

Hand-held Dynamometry and Quantitative Myometry

- Stability or improved muscle strength from baseline in maximum hand-held dynamometry and percent predicted Quantitative Myometry was observed

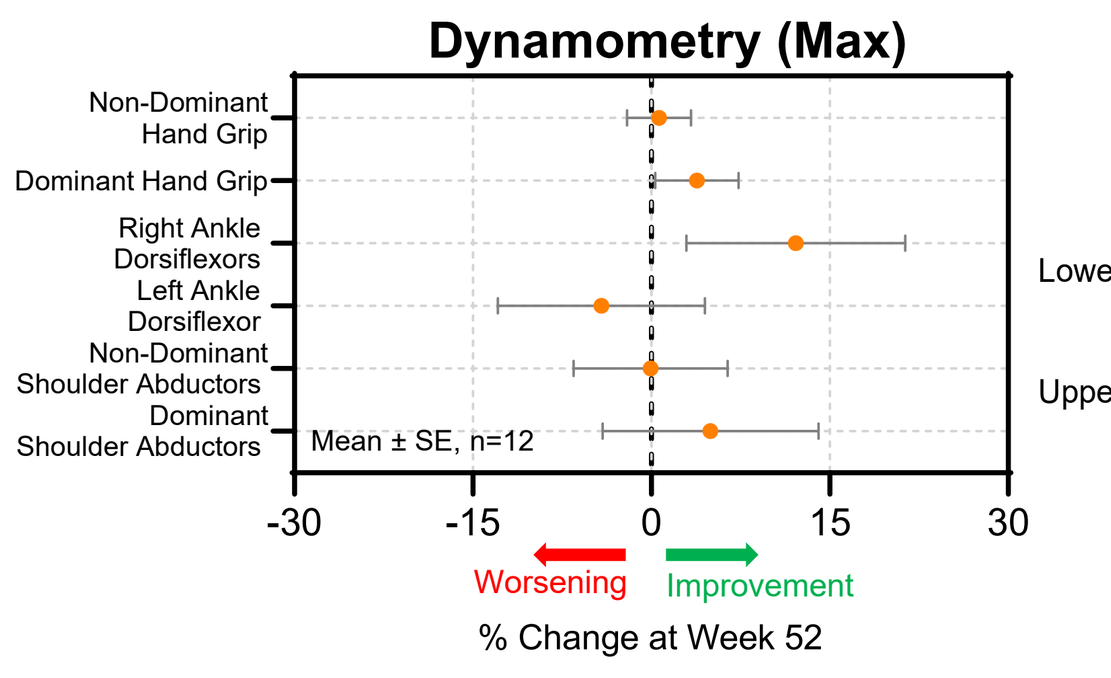
Dynamometry (Max)
Non-Dominant Hand Grip
Dominant Hand Grip
Right Ankle Dorsiflexors
Left Ankle Dorsiflexor
Non-Dominant Shoulder Abductors
Dominant Shoulder Abductors
Mean ± SE, n=12
Worsening
Improvement
% Change at Week 52

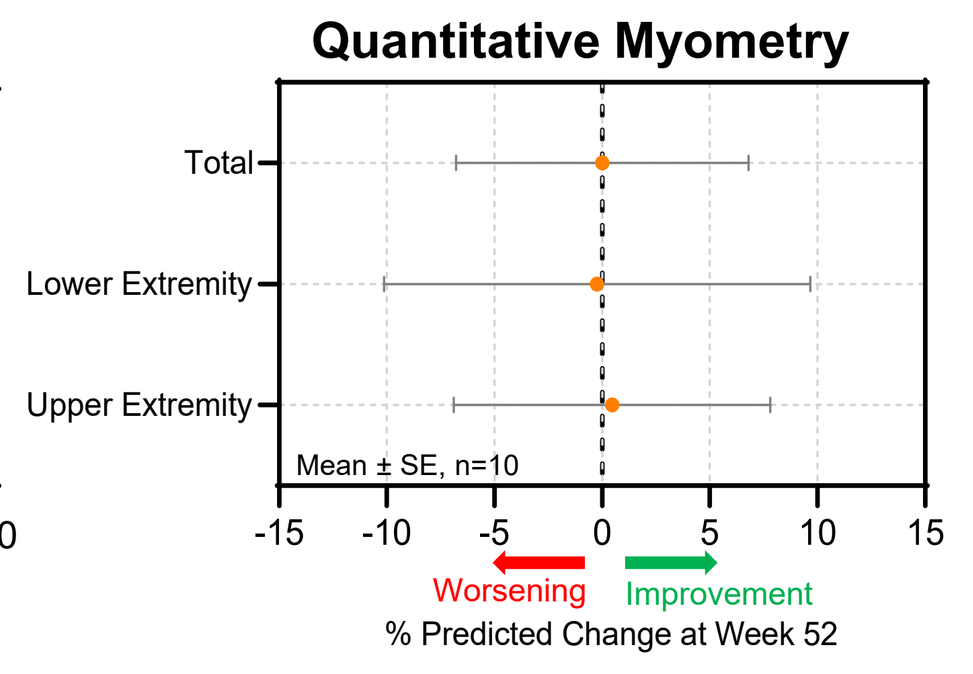
Quantitative Myometry
Total
Lower Extremity
Upper Extremity
Mean ± SE, n=10
Worsening
Improvement
% Predicted Change at Week 52

TUG, FHSD-TUG, FSHD-RODS, Motor Function Measurement, FSHD-HI, 6-MWT, and Spirometry showed minimal changes over 52 weeks of treatment

Conclusion

- Losmapimod was generally well-tolerated in FSHD study participants for approximately 52 weeks of treatment in an open-label, single-site study
- Losmapimod demonstrated expected PK and PD
- Improvement or minimal changes were observed in structural (MRI and ultrasound) and FSHD-relevant functional measures (RWS, HHD, and QMT)
 - Improvements in Reachable Workspace were demonstrated across quintants, with the largest improvements in the upper quadrants (Q1 and Q3)
- Patients recognized benefit of treatment based on reported improvement on PGIC
- The results of this open-label, single site study seem to confirm and extend in an independent cohort the results of the ReDUX4 study, supporting the benefit/risk of losmapimod as a disease-modifying therapy and continued development of losmapimod in FSHD

Acknowledgements

We thank the patients and their families for their participation in this study; the staff at our study site for their contributions to the care of the patients. We also thank our collaborators and supporting patient organizations for their support.