Feasibility of Measuring Functional Performance of FSHD Patients Using Wearable Sensors to Quantify Physical Activity

Joost Kools[1], Nicol Voermans[1], Karlien Mul[1], Baziel van Engelen[1], Michelle L. Mellion[2], John Jiang[2], Jennifer Shoskes[2], Kelly Marshall[2], David Jackson[2], Yuxi Zhao[2], Anil Tarachandani[3], Joanita Figueredo[3] Damien Eggenspieler[4]

[1]Radboud University Medical Centre (Radboudumc), Nijmegen, NL, [2]Fulcrum Therapeutics, Cambridge, MA, USA, [3]UsinLife LLC, Whitehouse Station, NJ, USA ,[4]Sysnav SAS, Vernon, France

Introduction

- FSHD is a serious, rare, progressive and heterogeneous disease, caused by the aberrant expression of DUX4 in skeletal muscle leading to progressive muscle loss and accumulation of disability.
- Wearable technology allows the collection of quantitative real-world data over long periods of time in a clinical study, providing deep insights into how patients feel and function
- Use of wearable devices in FSHD can provide critical data about how people living with FSHD function and manage their activities of daily living providing a sensitive assessment of disease progression and potentially response to treatment
- ActiMyo (Sysnav, Vernon, France) was developed to quantify the movements of ambulatory or non-ambulatory patients. This device provides reliable, quantifiable, and integrated estimate of patient activity through a patented magneto-inertial navigation measuring 3-dimensional (3D) movement of a patient's limbs with magnetic and inertial sensors.

Rationale

Measuring real-life activities provides critical assessments of how participants function in the real world, unique insight into disease progression and potentially efficacy of treatments.

Objective

- Evaluate feasibility to monitor daily activity and assess functional outcomes using wearable sensor devices in an open label study (OLS) of losmapimod in facioscapulohumeral muscular dystrophy (FSHD).
- Feasibility was measured by the reliability of the data over time and amount of background noise.
- The secondary objective was patients' compliance with using the wearable device.

Methods

Upper limb and lower limb mobility were evaluated with "Actimyo" made by Sysnav

- Actimyo, quantifies movements of patients with neuromuscular disorders such as Duchenne muscular dystrophy (DMD), Spinal amyotrophy (SMA), Parkinson's and records activity based on magneto-inertial navigation
- Raw data from the sensors are loaded on to the internal memory of the docking station/charging unit, and transmitted DAILY via an internet connection to a remote storage platform

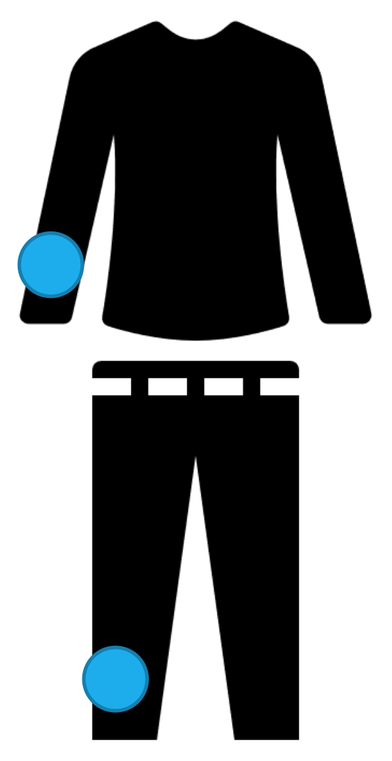

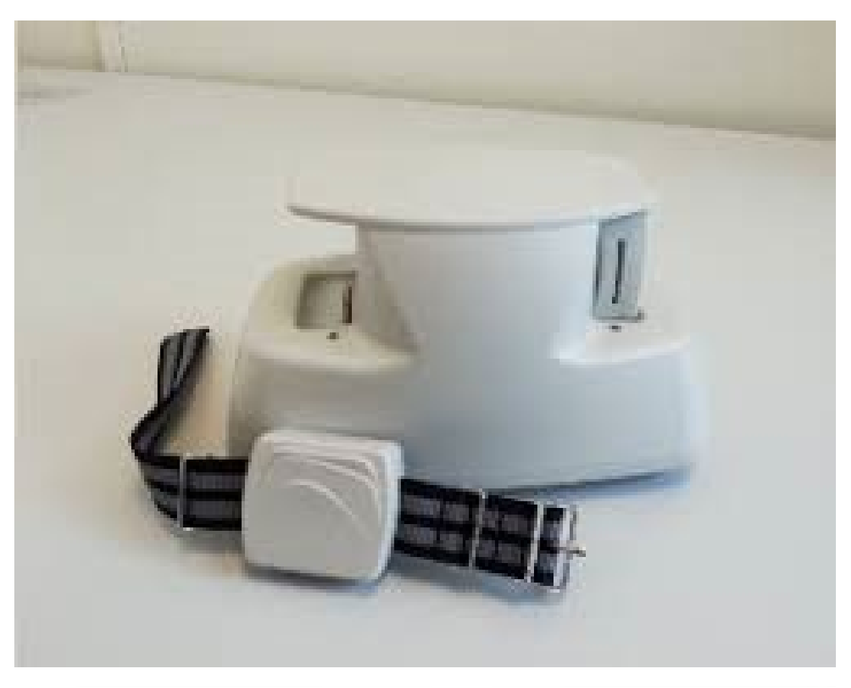

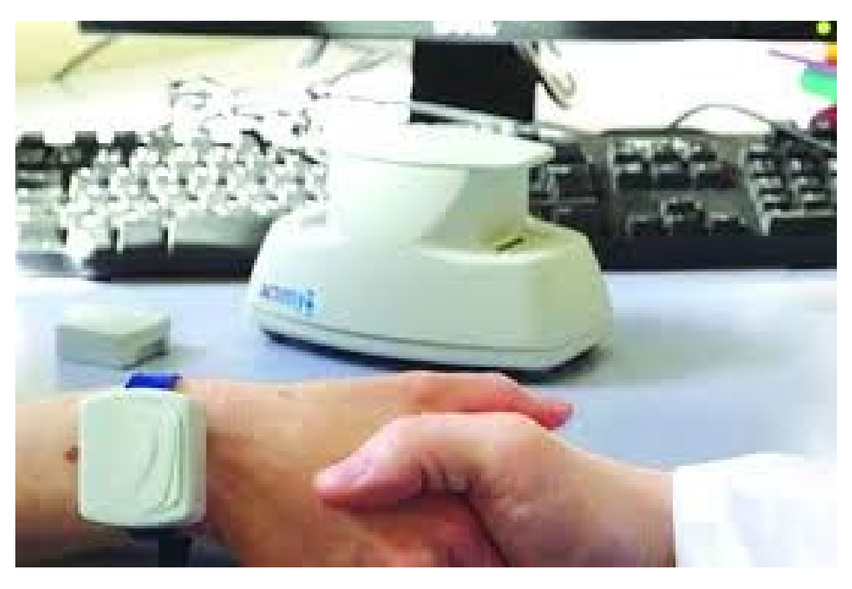

Participants are provided with two watch like sensors to measure movements:

1. On the ankle for lower extremity movements like gait
2. On the wrist for upper extremity movements like mobility

Two kinds of movements were evaluated:

- **Free living movement** – recorded by wearing two devices all day (12-16 hours)
- **Imposed movements (2 times per day)** set of movements twice daily

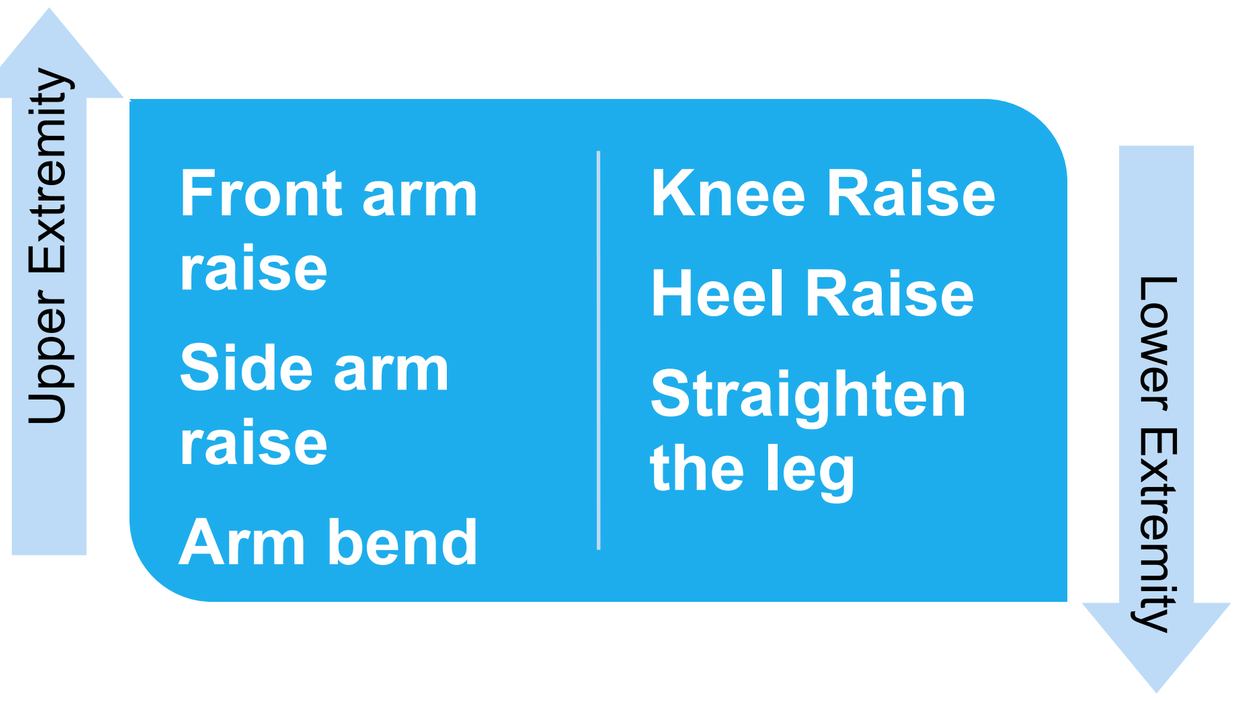
Upper Extremity
Front arm raise
Side arm raise
Arm bend
Knee Raise
Heel Raise
Straighten the leg
Lower Extremity

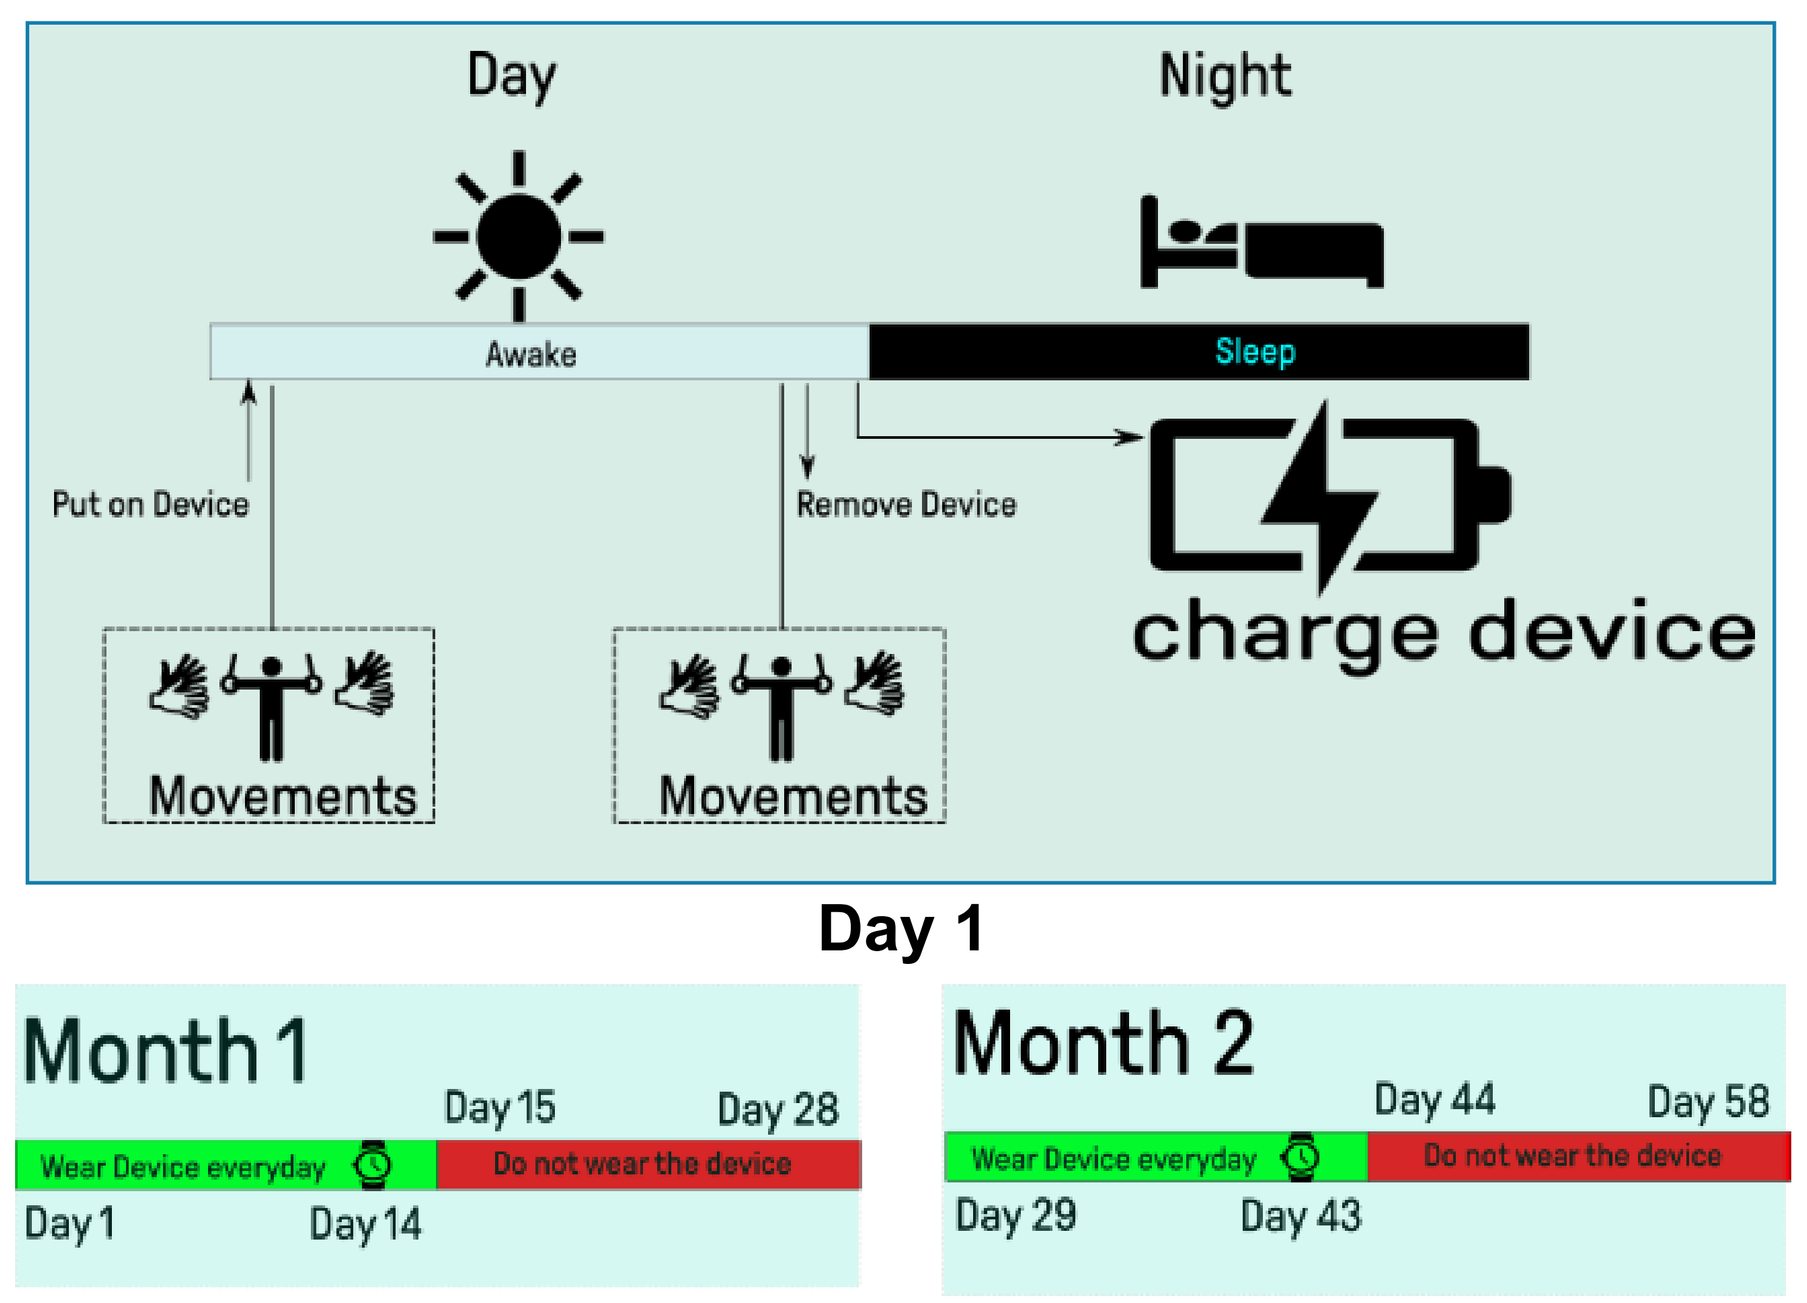
Day
Night
Awake
Sleep
Put on Device
Remove Device
Movements
Movements
charge device
Day 1
Month 1
Day 15
Day 28
Wear Device everyday
Do not wear the device
Day 1
Day 14
Month 2
Day 44
Day 58
Wear Device everyday
Do not wear the device
Day 29
Day 43

Study Design

- Single center open label study (OLS) at Radboud University, Netherlands
- Study Population: Enrolled 14 participants with genetically confirmed FSHD1

Open-Label Study (OLS): Phase 2 Open-Label Single-Center, 52-Week Study

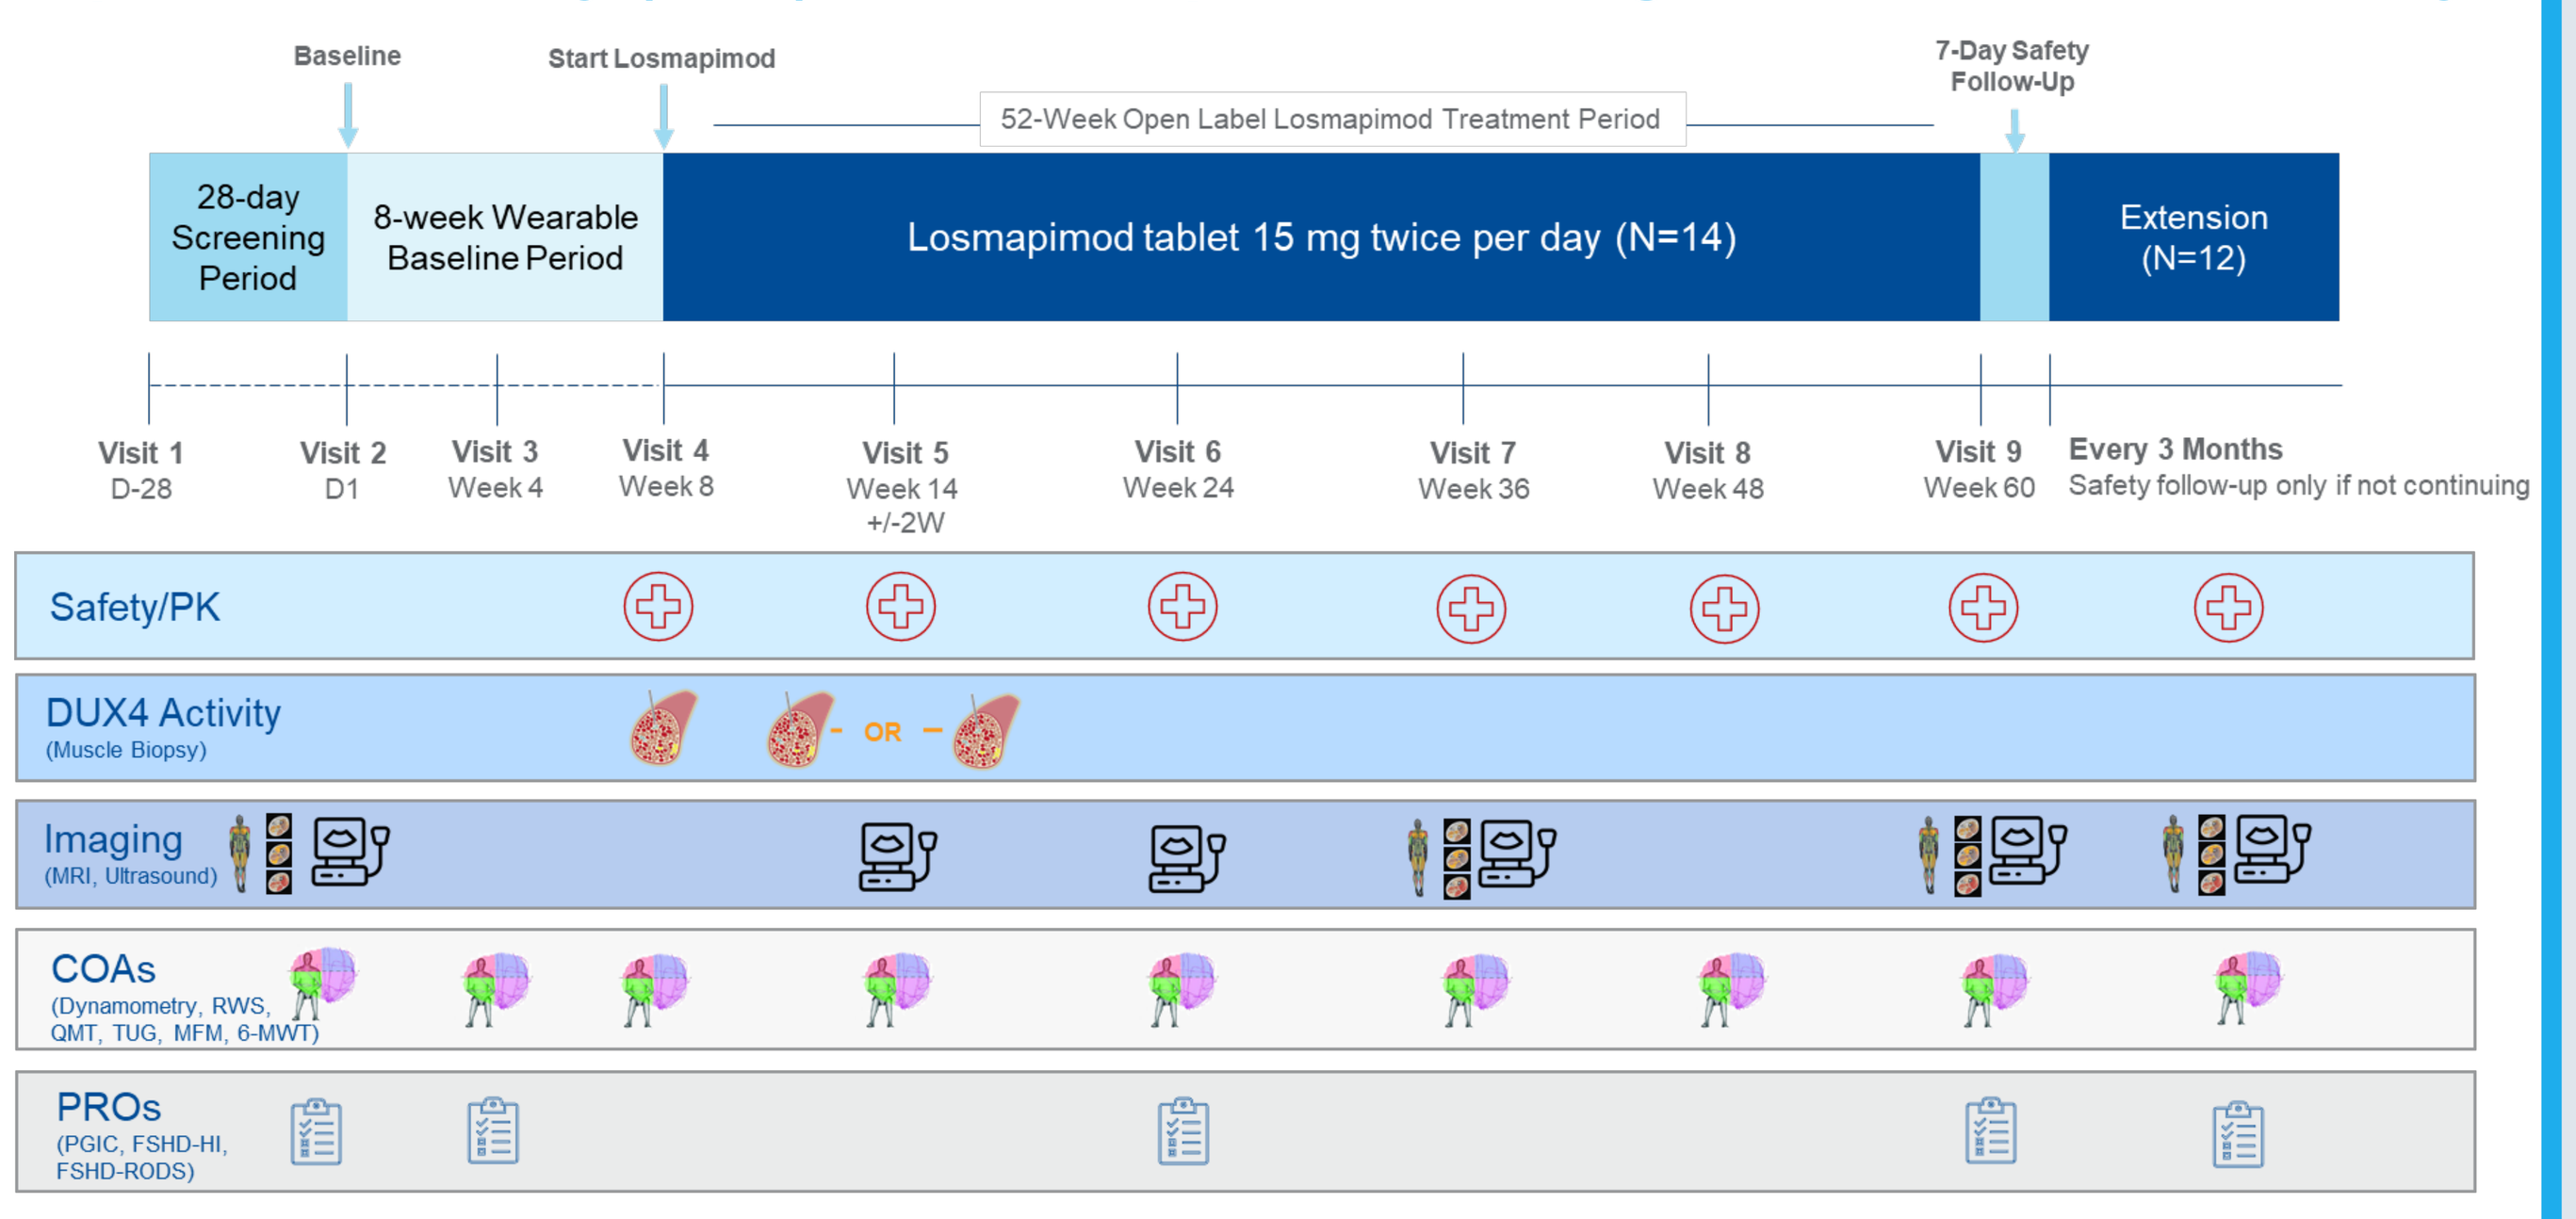
Baseline
Start Losmapimod
52-Week Open Label Losmapimod Treatment Period
7-Day Safety Follow-Up
28-day Screening Period
8-week Wearable Baseline Period
Losmapimod tablet 15 mg twice per day (N=14)
Extension (N=12)
Visit 1 D-28
Visit 2 D1
Visit 3 Week 4
Visit 4 Week 8
Visit 5 Week 14 +/-2W
Visit 6 Week 24
Visit 7 Week 36
Visit 8 Week 48
Visit 9 Week 60
Every 3 Months Safety follow-up only if not continuing
Safety/PK
DUX4 Activity (Muscle Biopsy)
OR
Imaging (MRI, Ultrasound)
COAs (Dynamometry, RWS, QMT, TUG, MFM, 6-MWT)
PROs (PGIC, FSHD-HI, FSHD-RODS)

Main Inclusion Criteria:	Main Exclusion Criteria:
▪ Age 18-65 years ▪ Genetically confirmed diagnosis of FSHD1 ▪ Ricci score 2-4 ▪ STIR+ muscle, as determined by a central reader, safely accessible by needle biopsy	▪ Medical conditions that can confound results of the study ▪ Contraindication to MRI ▪ Contraindication to muscle biopsy

Demographics Characteristics

		Losmapimod 15 mg BID (N=14)
Age (years)	Mean (SD)	45.7 (11.12)
Race, White	n (%)	13 (92.9)
Body Mass Index (BMI) (kg/m²)	Mean (SD)	24.0 (2.94)
D4Z4 Repeat Category, n (%)	1-3 Repeats	3 (21.4)
	4-9 Repeats	11 (78.6)
Ricci Score, n (%)	2	0
	2.5	1 (7.1)
	3	5 (35.7)
	3.5	2 (14.3)
	4	6 (42.9)
Enrolled	n (%)	14
Completed the study	n (%)	14 (100)
Discontinued from study	n (%)	0
Entered extension	n (%)	12 (85.7)

- All subjects completed the study
- 2 subjects declined participation in the extension study for reasons unrelated to study drug/adverse events

Results

Feasibility and Compliance

Participant compliance for wearing the devices was extremely high at 99%. The total number of days that all 14 participants were monitored was 2,941 days or 36,758 hours (an average of 2,626 hours per participant).

Period –> Patient	P-1 hrs	P-2 hrs	P-3 hrs	P-4 hrs	P-5 hrs	P-6 hrs	P-7 hrs	P-8 hrs	P-9 hrs	P-10 hrs	P-11 hrs	P-12 hrs	P-13 hrs	P-14 hrs	P-15 hrs	P-16 hrs	P-17 hrs
101	175	170	150	112	178	138	132	125	118	109	111	0	123	113	147	171	
103	176	179	149	163	174	162	142	166	182	182	173	188	179	178	167	183	168
104	171	182	184	184	163	175	150	162	189	174	190	174	184	189	141	137	
105	184	183	164	248	183	157	169	142	140	228	88	129	132	142	197	203	109
108	187	190	174	179	167	132	118	126	154	128	134	124	161	145	115	130	
112	153	163	121	169	177	180	185	151	128	163	127	195	165	155	193		
113	203	190	153	179	173	181	180	195	189	168	111	147	141	150	156	148	
114	134	190	125	144	141	174	179	188	132	133	129	124	187	173	171	174	
115	145	149	153	136	125	128	126	140	155	126	138	145	135	154	132	end	
116	113	167	174	159	138	147	208	187	117	148	196	178	186	177	152	163	
119	152	174	162	169	139	175	181	171	126	176	169	167	168	170	163	136	
120	113	102	110	108	109	108	112	110	99	115	103	108	111	108	115	98	
123*	186	194	175	114	166	209	136	146	196	150	211	187	169	208	199		
124*	143	147	137	157	153	165	118	171	153	141	177	165	153	170	130		

Data Periods	
< 50	Wear time in hours per period
50 -100	
100- 180	
>180	
	Baseline periods
	Periods post treatment (except for 123 and 124)
	First treatment period for 123 and 124

Analysis, Processing and Reliability

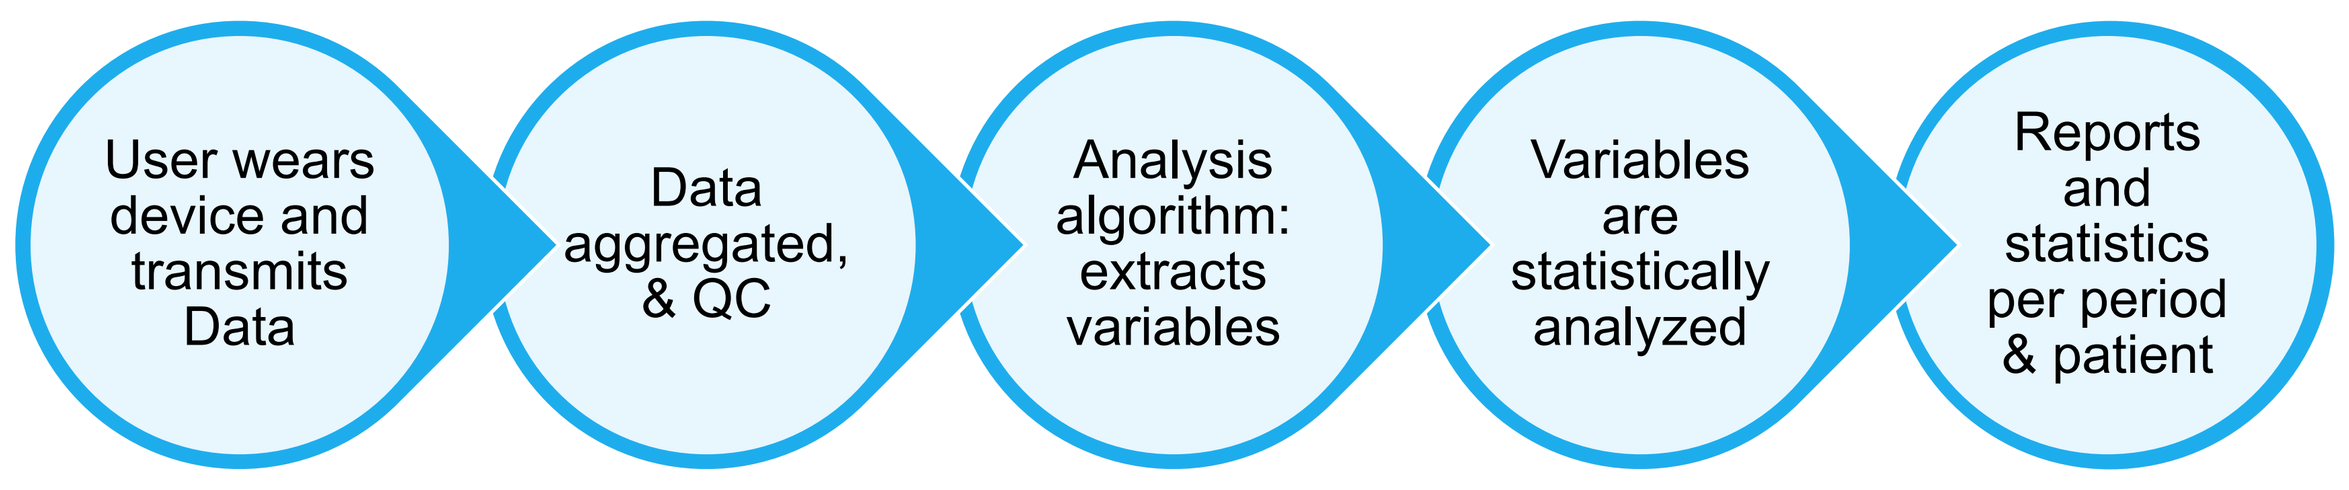
User wears device and transmits Data
Data aggregated, & QC
Analysis algorithm: extracts variables
Variables are statistically analyzed
Reports and statistics per period & patient

Variable	SEM	[Min ; Max] Period 1	Standard Deviation (P1)	ICC (P1,P2)
95th percentile stride velocity	0.02 m/s	[0.62 ; 1.57] m/s	0.24 m/s	99.1%
90th percentile walking perimeter	1.6 m	[10 ; 60] m	11.6 m	98.1%
Upper limb power	0.05 W	[0.63 ; 1.69] W	0.25 W	96.4%
Imposed movements	4.1°	[75 ; 175] °	29.8°	97.1%
Arm swing during walk	5.0°	[25 ; 80] °	13.9°	91.3%

P = Period

Variables derived from raw data

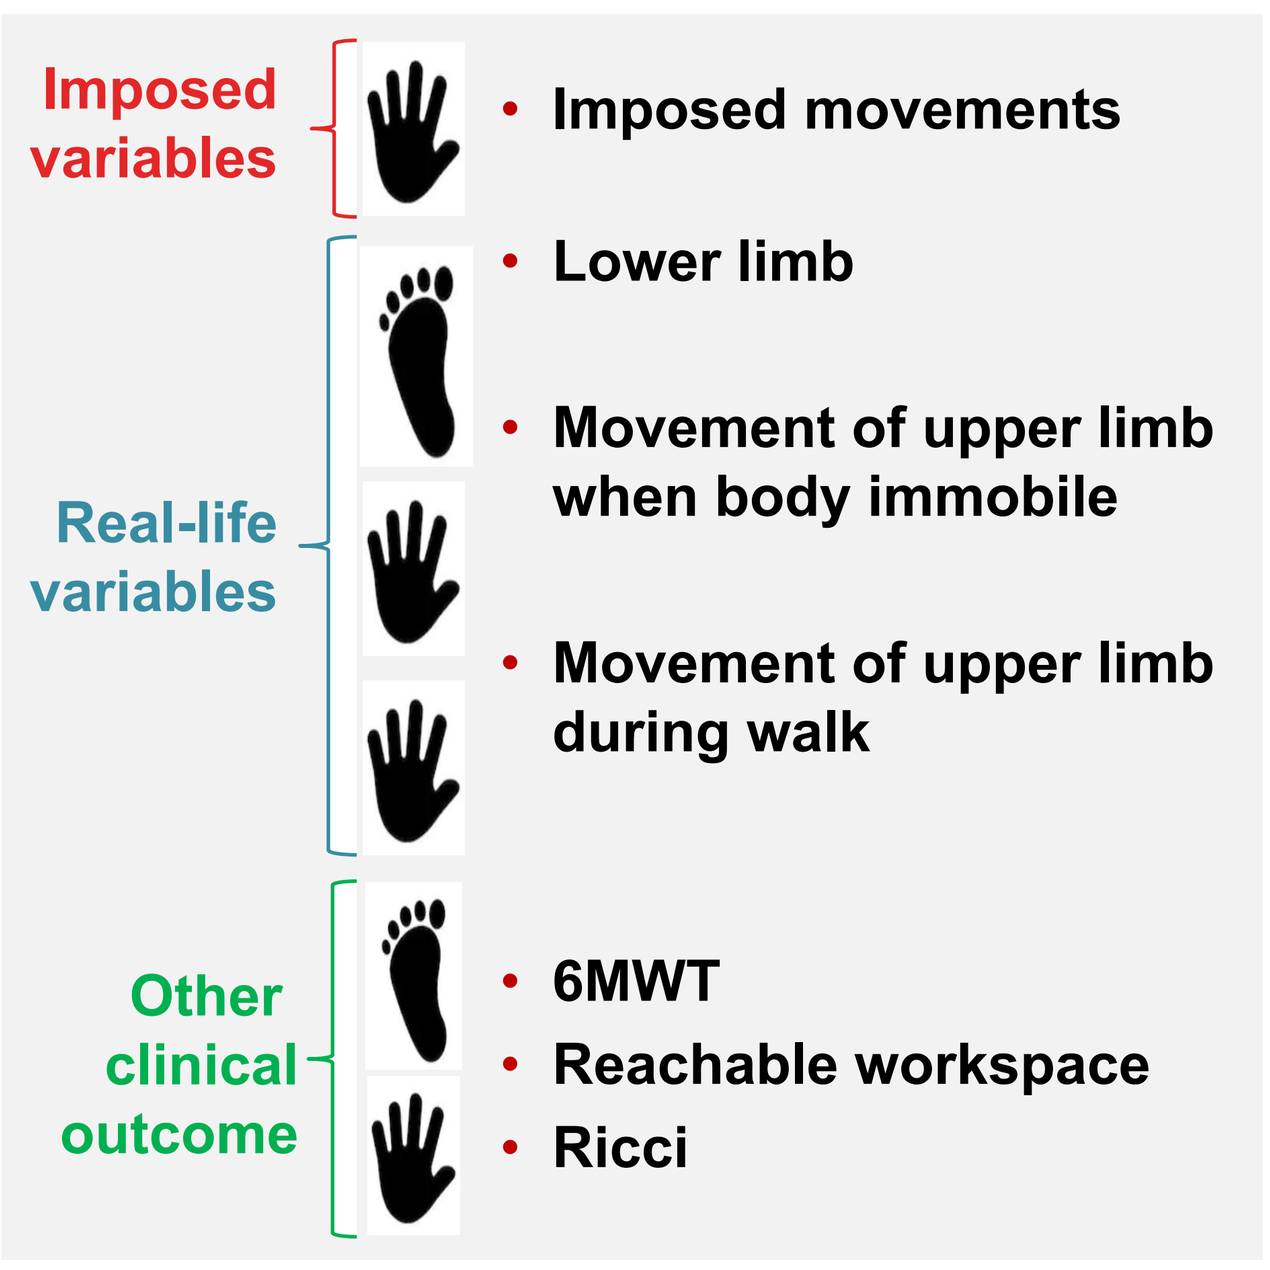
Imposed variables
Imposed movements
Lower limb
Real-life variables
Movement of upper limb when body immobile
Movement of upper limb during walk
Other clinical outcome
6MWT
Reachable workspace
Ricci

Moderate to Strong Correlations between Clinic and Wearable Variables at Baseline

Upper Extremity Variables	50th Percentile Pitch Angle	50th Percentile Height of Imposed Movements	95th Percentile of pitch angle
Right Arm RWS Total RSA	0.25	0.79*	-0.10
Max HHD R Elbow Flexor	0.56*	-0.05	0.66*
Lower Extremity Variables	**Mean Distanced Walked per Hour (Stride Distance)**	**95th percentile Stride Length**	**95th percentile Stride Velocity**
6-MWT	0.54*	0.85*	0.92*
Classic TUG	-0.46*	-0.52*	-0.61*
Max HHD R Ankle	0.47*	0.61*	0.64*

* = p<0.05; values represented are Pearson correlations; RWS = reachable workspace; RSA = reachable surface area; R = right; HHD= hand-held dynamometry; 6-MWT=6-minute walk test; TUG=Timed up and go
Color Representation: High correlation (r >0.7) = green, Moderate correlation (0.5-0.7) = blue, Weak correlation (0.3-0.5) = yellow

FSHD participants are distinct in the location of disability. When divided into 4 groups[1], in-clinic and wearable variables are coordinated with severity location

Baseline (Mean, SD)	Upper (n=3)	Lower (n=2)	Both (n=6)	Mild (n=3)	All Patients (n=14)
Classic TUG (s)	7.2 (0.5)	11.3 (2.7)	13.9 (5.7)	8.4 (0.8)	10.9 (4.7)
95 Percentile Stride Velocity (m/s)	1.4 (0.1)	1.2 (0.1)	1.2 (0.4)	1.5 (0.1)	1.3 (0.3)
Stride Distance (m/hr)	95.4 (19.6)	74.0 (8.3)	55.4 (29.1)	182.1 (91.8)	93.8 (65.1)
50th Percentile Pitch Angle (degrees)	16.4 (2.8)	26.9 (5.6)	16.7 (5.3)	19.8 (4.5)	18.7 (5.6)

(1) Division into groups: The coordinator assessments of severity location and the comments in the EDC were used to divide the participants into 4 subjective groups. N represents participants in each group.

- Upper – greater disability in the upper extremity n=3
- Lower – greater disability in the lower extremity n=2
- Both – High disability in both upper and lower extremity n=6
- Mild – Mild disability in both upper and lower extremity n=3

Physical function as measured by gait velocity tends to increase over 1 year treatment period in "Mild" or "Upper" group as compared to "Lower" or "Both" group.

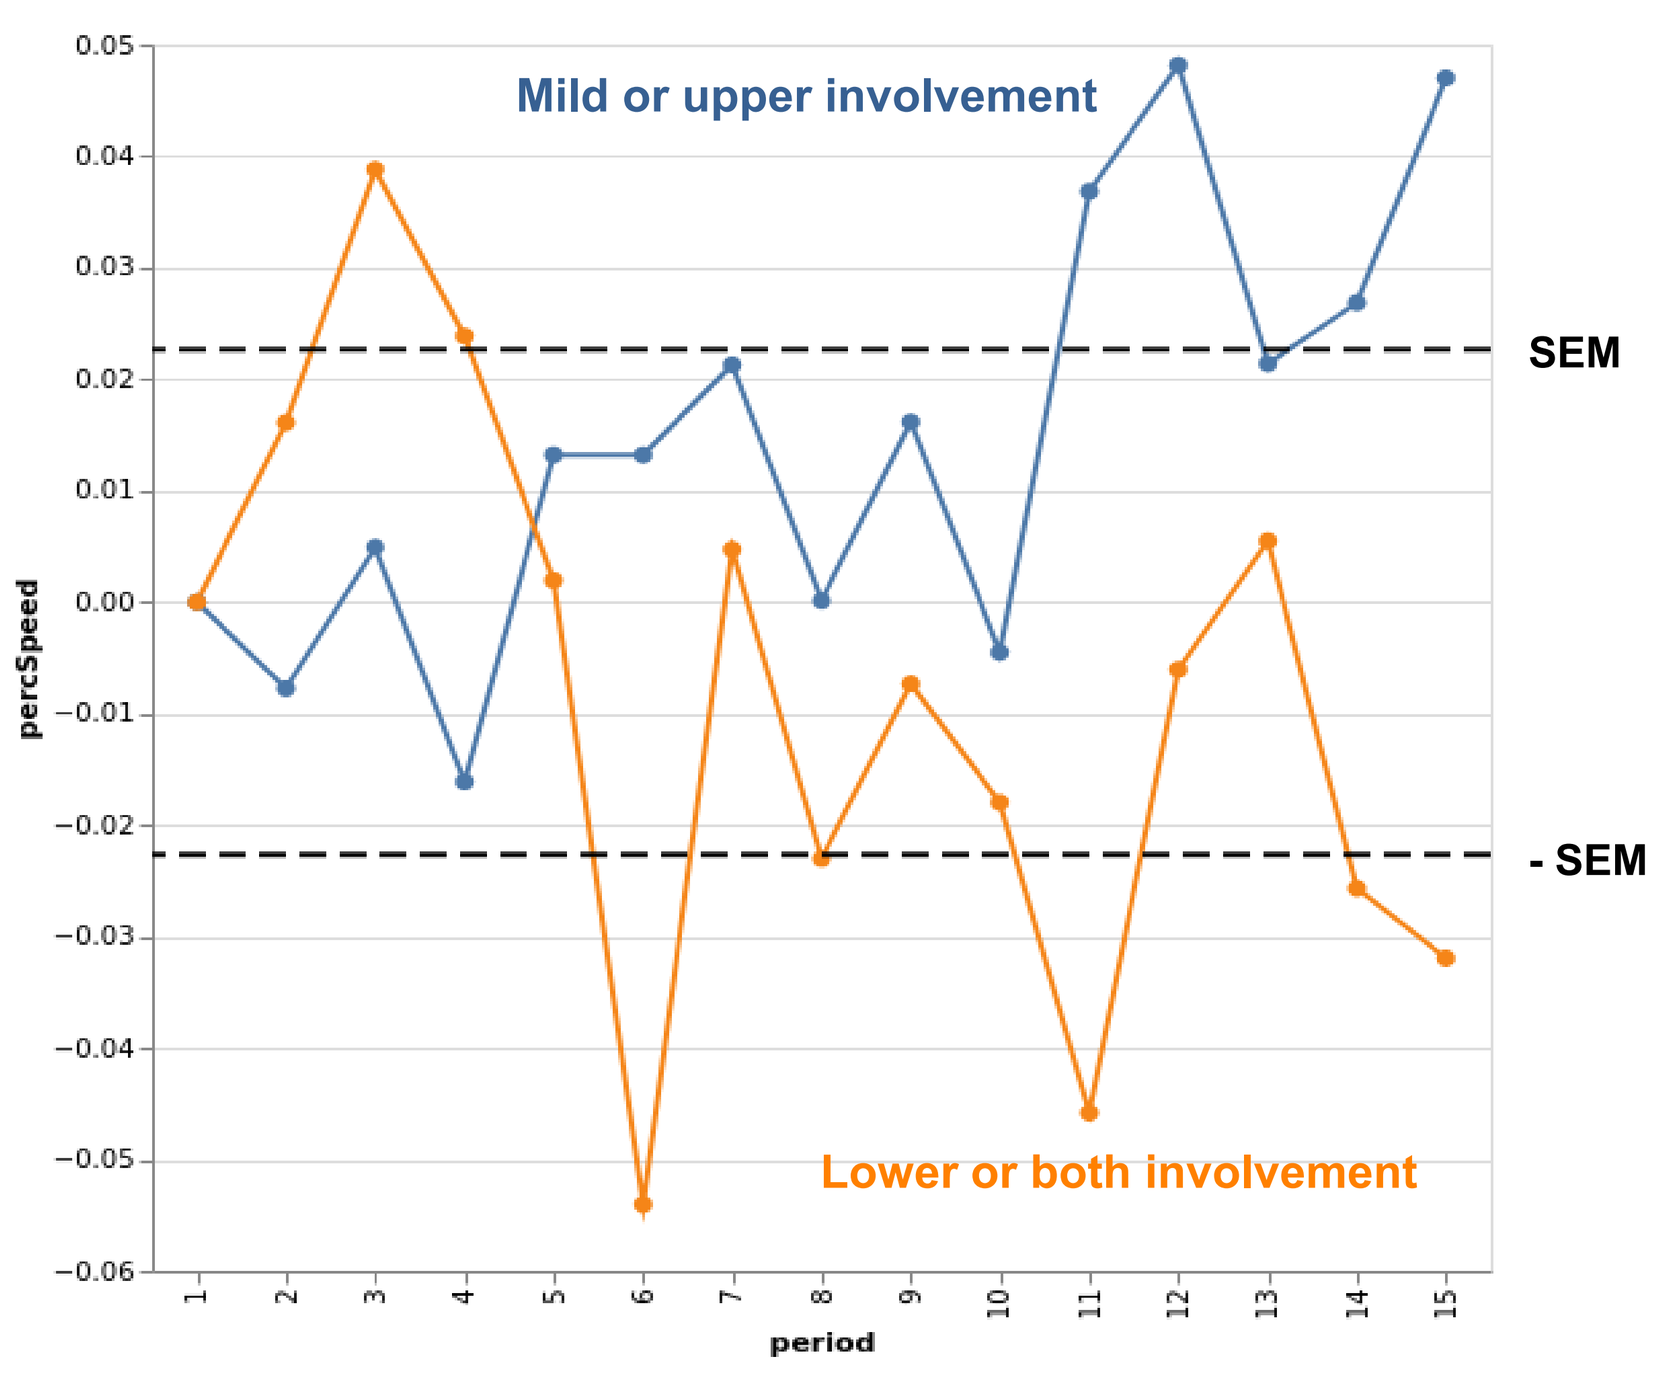
Mild or upper involvement
Lower or both involvement
SEM
- SEM
percSpeed
period
0.05
0.04
0.03
0.02
0.01
0.00
−0.01
−0.02
−0.03
−0.04
−0.05
−0.06
1
2
3
4
5
6
7
8
9
10
11
12
13
14
15

Note: The observed change was greater than the measurement error (SEM)

Conclusion

- Measurement of functional performance in FSHD patients using wearable sensors in FSHD is feasible, reliable, and correlates with multiple clinical outcome assessments of upper and lower extremity function, activities of daily living and mobility.
- Patients were 99% compliant over the year long assessment.
- This assessment could provide critical data about disease progression and treatment efficacy in future clinical trials.