EP.330

Whole Body MRI Quantitative muscle analysis to evaluate Efficacy of Losmapimod in a Phase 2 Placebo-Controlled Study in Subjects with FSHD (ReDUX4): ReDUX4 (NCT04003974)

Shared First Authorship: Rabi Tawil, MD[1], Kathryn Wagner, MD, PhD[2*], Jeffrey Statland, MD[3], Leo Wang, MD, PhD[4], Angela Genge, MD[5], Sabrina Sacconi, MD, PhD[6], Hanns Lochmüller, MD, PhD[7], David Reyes Leiva, MD[8], Jordi Diaz-Manera, MD, PhD[8,9], Jorge Alonso-Perez, MD,[8], Nuria Muelas, MD, PhD[10], Juan J. Vilchez, MD, PhD[11], Alan Pestronk, MD[12], Summer Gibson, MD[13], Namita A. Goyal, MD[14], Johanna Hamel, MD[1], Lawrence Hayward, MD[15], Nicholas Johnson, MD[16], Samantha LoRusso, MD[17], Miriam Freimer, MD[17], Perry B. Shieh, MD, PhD[18], Sankarasubramoney H. Subramony, MD[19], Doris Leung MD, PhD[2], Baziel van Engelen MD, PhD[20], Joost Kools, MD [20], Olof Dahlqvist[21], Per Widholm[21], John Jiang, PhD[22], Adefowope Odueyungbo, PhD [22], William Tracewell, PhD[22], Alisa Rahilly, BSc[22], L. Alejandro Rojas, PhD[22], Anthony Accorsi, PhD[22], , Steve Mennen, PhD[22], Lucienne Ronco, PhD[22], Diego Cadavid, MD[22], Christopher M. Moxham, PhD[22], Christopher Morabito, MD[22], Robert Gould, PhD[22], Michelle L. Mellion, MD[22].

[1]University of Rochester, [2]Kennedy Krieger Institute, Johns Hopkins University, [3]University of Kansas, [4]University of Washington, [5]Montreal Neurological Institute and Hospital, [6]Nice University Hospital, Peripheral Nervous System and Muscle Department, University Cote Azur, Nice, France, [7]Children's Hospital of Eastern Ontario Research Institute; Division of Neurology, Department of Medicine, The Ottawa Hospital; and Brain and Mind Research Institute, University of Ottawa, Canada, [8]Hospital Universitari Santa Creu i Sant Pau. Institut de Recerca IIB Sant Pau, Barcelona, Spain, [9]John Walton Muscular Dystrophy Research Center, Newcastle University, Newcastle, United Kingdom, [10]Hospital Universitari i Politecnic La Fe de Valencia. Instituto de Investigación Sanitaria IIS La Fe. CIBERER, [11]Neuromuscular and Ataxias Research Group, Instituto de Investigación Sanitaria La Fe, Valencia, Spain [12]Washington University, [13]University of Utah, [14]University of California Irvine, [15]University of Massachusetts, [16]University of Virginia[17] Ohio State University, Wexner Medical Center, [18]University of California Los Angeles, [19]University of Florida, [20]Radboud University Medical Center, Nijmegen [21]AMRA Medical AB, [22]Fulcrum Therapeutics

Introduction

- FSHD is a serious, rare, progressive and disabling disease, with no approved disease modifying therapies.
- DUX4 activates a downstream transcriptional program that causes myofiber death and replacement of muscle with fat resulting in progressive motor disability.
- Fatty and fibrotic tissue infiltration contribute to loss of function by altering muscle biomechanical properties.
- Assessment of treatment efficacy is challenging because of the heterogeneity of muscles involvement within and between patients.
- Functional assessments show limited sensitivity in detecting slowing or arrest of disease progression in FSHD.

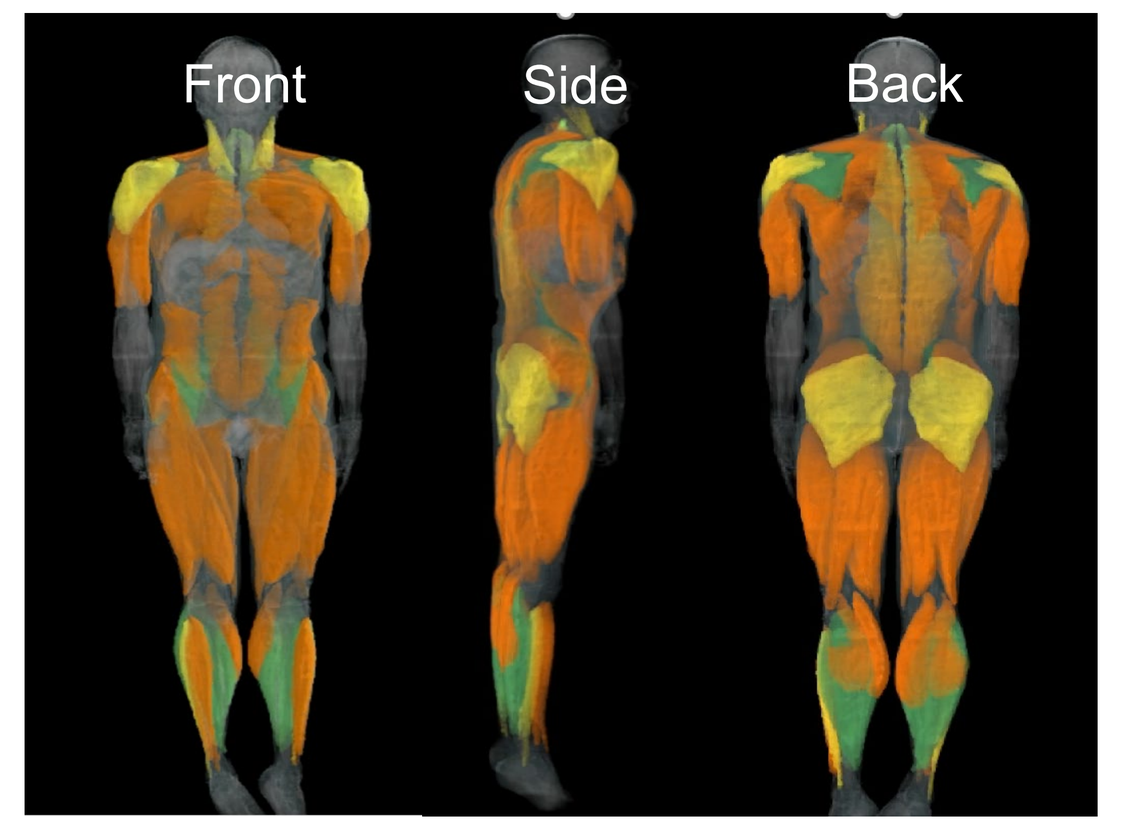
Front
Side
Back

Skeletal Muscle Replacement by Fat in FSHD as Reported in the Literature
Green= not affected
Yellow= small increases reported in some studies
Orange=high fat replacement in almost all studies

MRI can potentially detect pathological changes in muscle prior to changes in clinical outcome assessments (COA) and correlates with COAs.

Rationale

A treatment that reduces or prevents aberrant DUX4 activity in skeletal muscles has the potential to stop or prevent functional impairment and accumulation of disability and decrease/arrest replacement of muscle by fat.

- **Losmapimod** is a small molecule inhibitor of p38α/β Mitogen Activated Protein Kinase (MAPK).
- Pre-clinical studies demonstrated treatment with losmapimod and other p38 inhibitors resulted in dose-dependent reduction of DUX4 protein, DUX4 transcriptional program and skeletal muscle cell death in FSHD myotubes across all genotypes tested.
- Clinical studies with losmapimod in over 3,600 subjects across a diversity of diseases evidenced acceptable safety and tolerability for up to one year of treatment at relevant doses.

Objective

- Assess the efficacy of losmapimod with whole-body MRI using quantitative muscle and composite analyses.
- Quantitative muscle analyses include muscle fat infiltration (MFI), muscle fat fraction (MFF), and lean muscle volume (LMV).

ReDUX4 Study Design

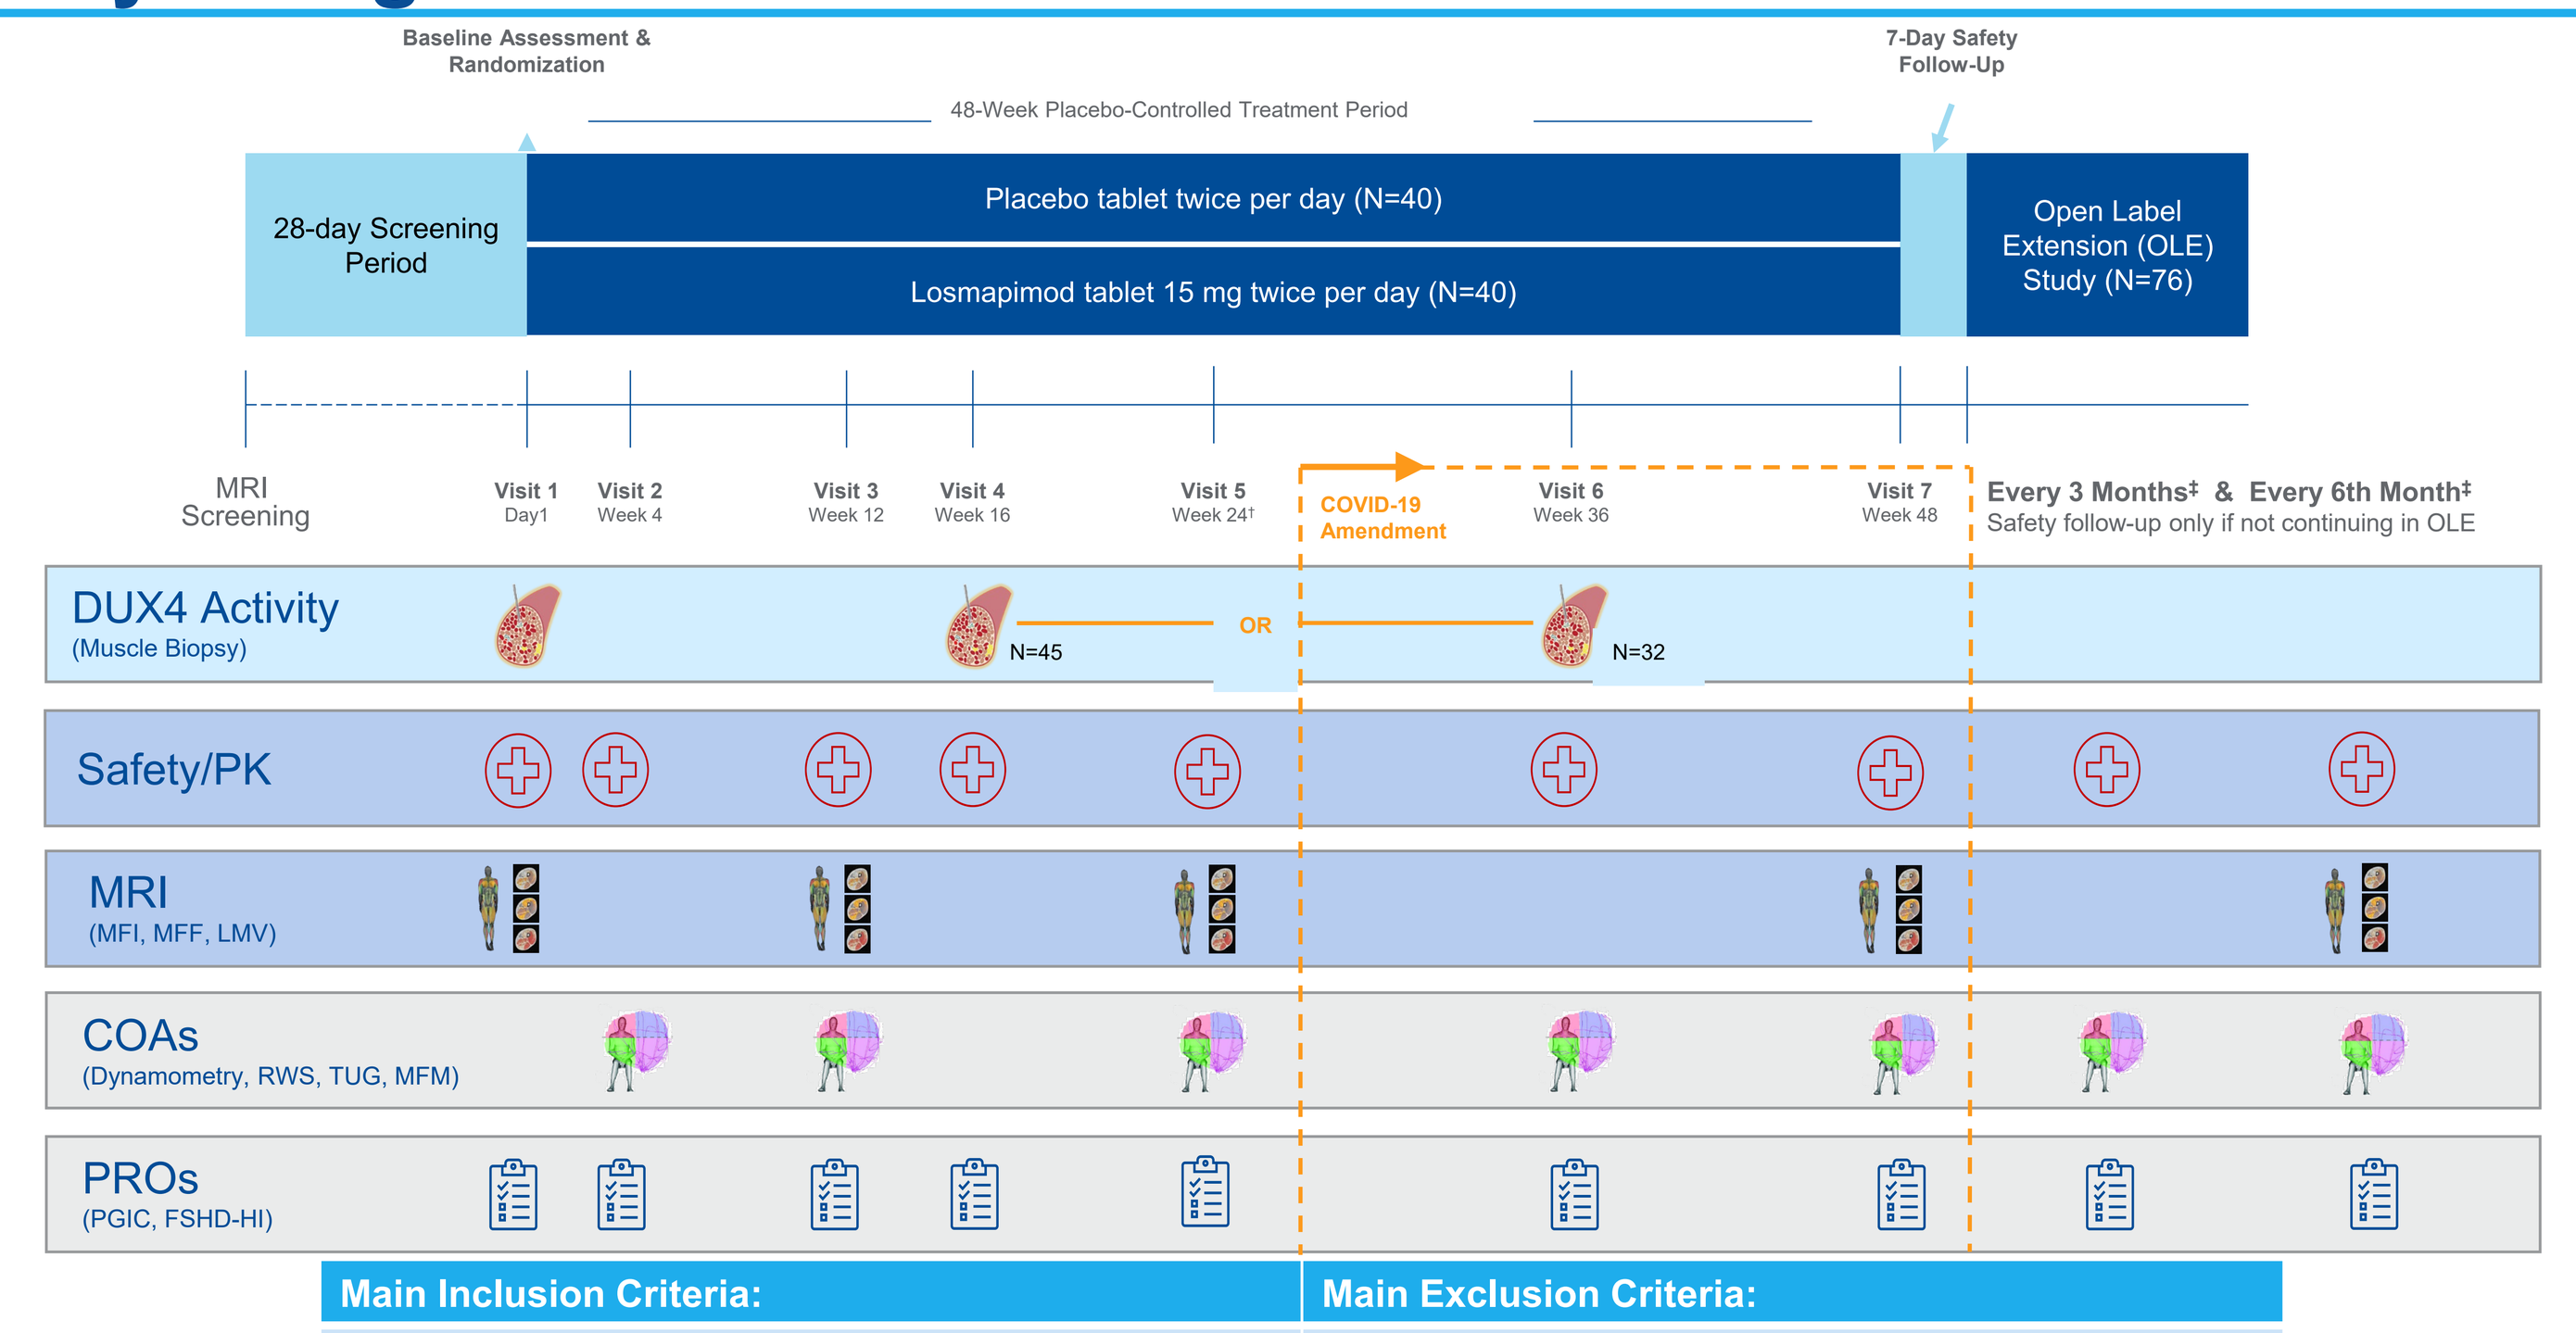
Baseline Assessment & Randomization
48-Week Placebo-Controlled Treatment Period
7-Day Safety Follow-Up
28-day Screening Period
Placebo tablet twice per day (N=40)
Losmapimod tablet 15 mg twice per day (N=40)
Open Label Extension (OLE) Study (N=76)
MRI Screening
Visit 1 Day1
Visit 2 Week 4
Visit 3 Week 12
Visit 4 Week 16
Visit 5 Week 24†
COVID-19 Amendment
Visit 6 Week 36
Visit 7 Week 48
Every 3 Months‡ & Every 6th Month‡
Safety follow-up only if not continuing in OLE
DUX4 Activity (Muscle Biopsy)
N=45
OR
N=32
Safety/PK
MRI (MFI, MFF, LMV)
COAs (Dynamometry, RWS, TUG, MFM)
PROs (PGIC, FSHD-HI)

Main Inclusion Criteria:	Main Exclusion Criteria:
▪ Age 18-65 years ▪ Genetically confirmed diagnosis of FSHD1 ▪ Ricci score 2-4 ▪ STIR+ muscle, as determined by a central reader, safely accessible by needle biopsy	▪ Medical conditions that can confound results of the study ▪ Contraindication to MRI ▪ Contraindication to muscle biopsy

Patient Demographics		Placebo BID (N=40)	Losmapimod 15 mg BID (N=40)
Age (years)	N	40	40
	Mean (SD)	45.7 (+/- 12.69)	45.7 (+/- 12.44)
Race n (%)	White	39 (97.5)	31 (77.5)
Body Mass Index (BMI) (kg/m²)	N	39	40
	Mean (SD)	26.19 (+/- 4.914)	25.71 (+/- 5.434)
D4Z4 Repeat Unit n (%)	1-3	6 (15.0)	7 (17.5)
	4-6	26 (65.0)	29 (72.5)
	7-9	8 (20.0)	4 (10.0)
D4Z4 Repeat Category n (%)	1-3 Repeats	6 (15.0)	7 (17.5)
	4-9 Repeats	34 (85.0)	33 (83.50)
Ricci Score n (%)	2	0	0
	2.5	7 (17.5)	5 (12.5)
	3	18 (45.0)	19 (47.5)
	3.5	7 (17.5)	11 (27.5)
	4	8 (20.0)	5 (12.5)

Methods

For full details see poster Widholm P, et al. Quantitative muscle analysis in FSHD using Whole-Body MRI: Composite Muscle Measurements for Cross-Sectional Analysis

Muscles Studied- 18 muscles Bilaterally; 36 Total

Neck
- Supraspinatus
- Infraspinatus
- Subscapularis
- Teres Minor

Legs
- Quadriceps
- Hamstrings
- Adductors
- Tibialis Anterior
- Gastrocnemius Medialis

Torso
- Pectoralis Major
- Rhomboids
- Latissimus Dorsi & Teres Major
- Trapezius
- Serratus Anterior
- Paraspinal (C3-Sacrum)

Arm
- Deltoid
- Biceps Brachii
- Triceps Brachii

Muscles Classified as Normal-Appearing "A", Intermediate "B", or End-Stage "C"

Intermediate "B" muscles are at high risk of progression

Normal-Appearing "A"

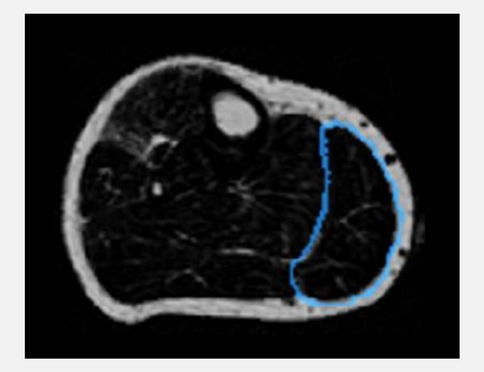

- Muscles do not appear to be affected by disease
- MFI < 10%; MFF < 50%

Intermediate "B"

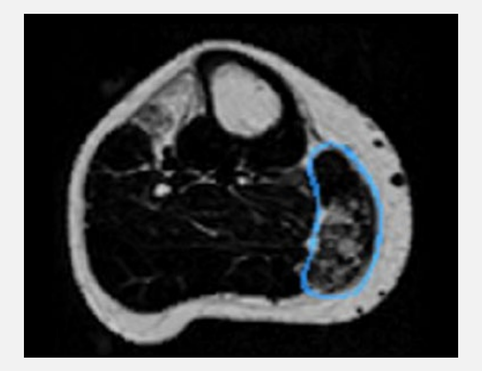

- Muscles clearly affected by disease, but not so severely fat replaced to have lost all function
- Included in the longitudinal composite score because they are most likely to progress
- MFI ≥ 10%; MFF < 50%

End-Stage "C"

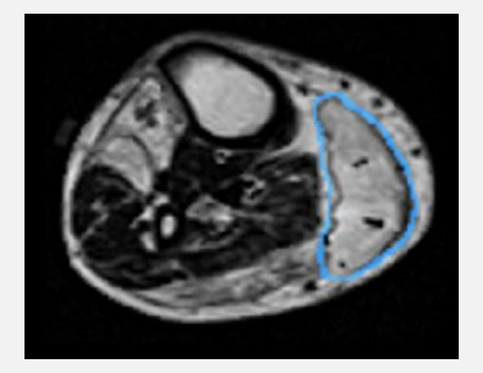

- Muscles severely fat replaced and have likely lost most if not all function
- MFF ≥ 50%

Quantitative Assessment of Skeletal Muscle Health

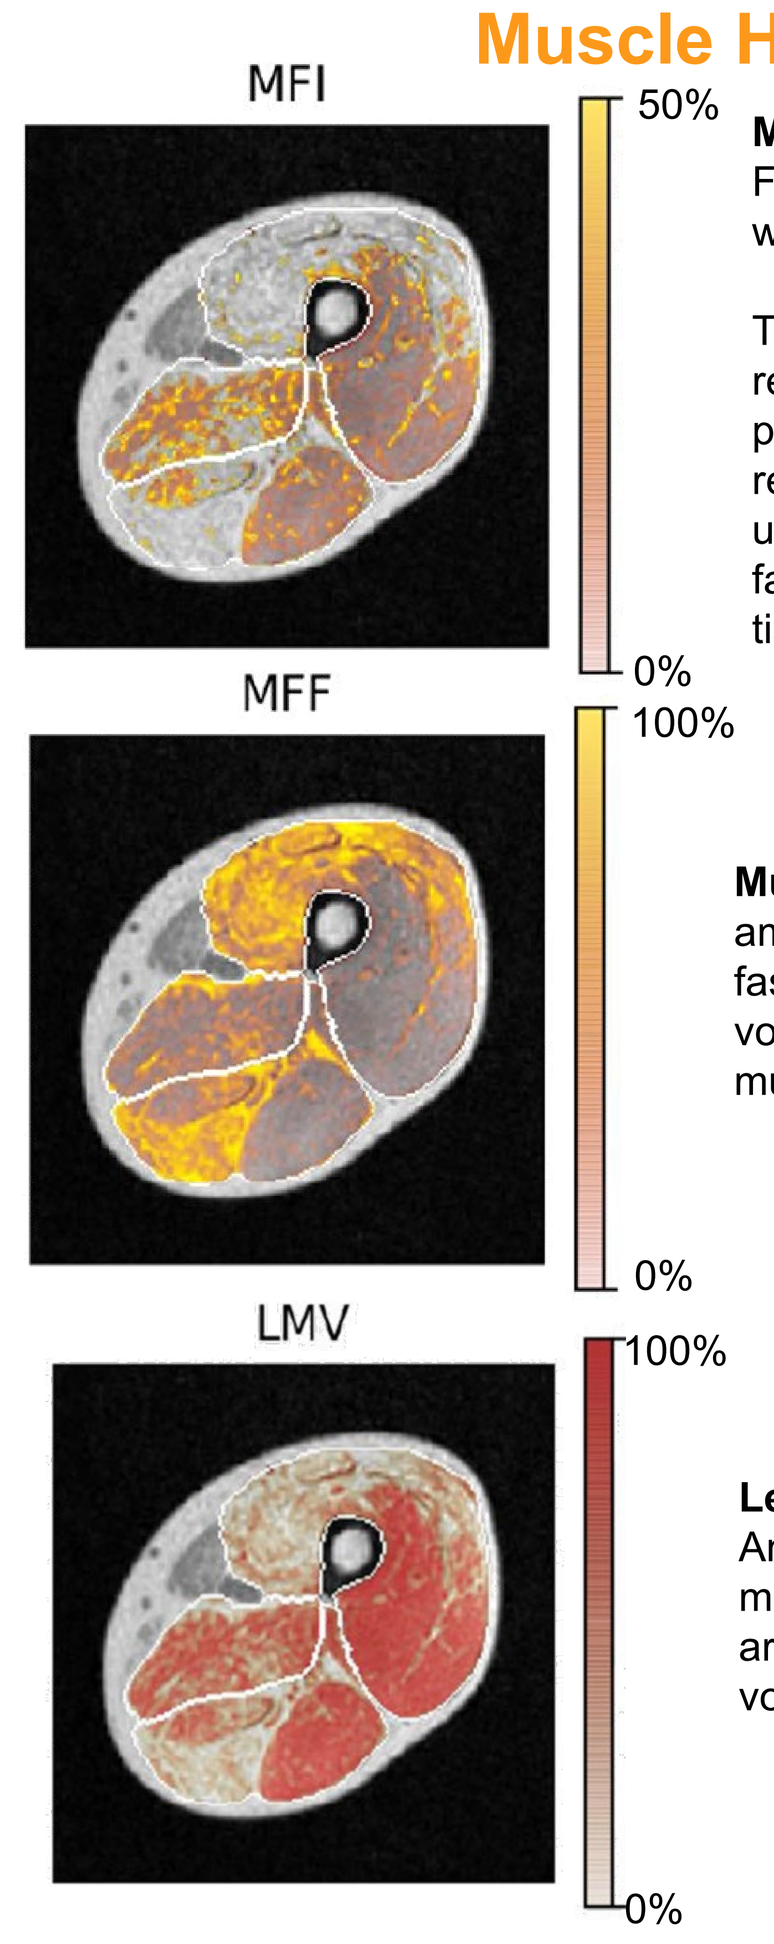
MFI
50%
0%
MFF
100%
0%
LMV
100%
0%

Muscle fat infiltration (MFI): Fat fraction calculated within voxels with less than 50% fat.

This measurement is a representation of the fat content in parts of the muscle that are not fat-replaced and is analogous to the use of MR spectroscopy to measure fat fraction in homogenous muscle tissue.

Muscle fat fraction (MFF): Relative amount of fat within the muscle fascia, calculated as the total volume of fat divided by the total muscle volume (TMV).

Lean Muscle Volume (LMV): Amount of muscle tissue within the muscle; similar to cross-sectional area. Calculated by subtracting volume of fat from TMV.

Composite Measurement to Evaluate Treatment Efficacy and Correlations with FSHD relevant COAs*

Whole Body MRI

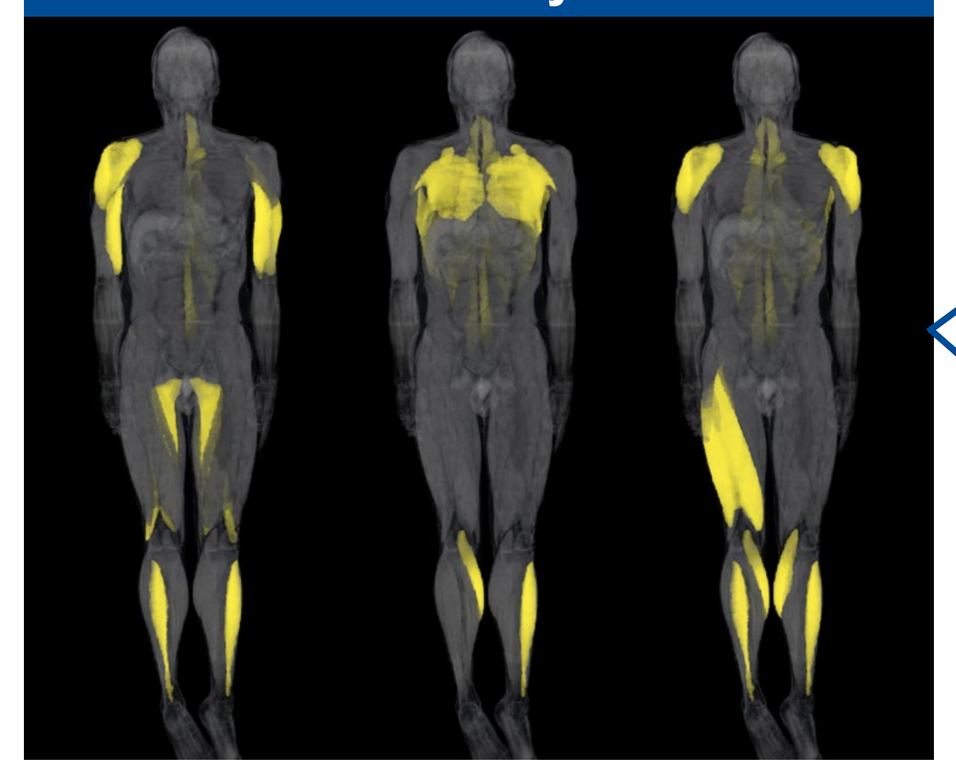

Treatment Efficacy Composites
- Measure treatment effect
- Only include B muscles
- No SQI are allowed at either time point

→ MFI_{tot}: Average MFI, weighted by lean volume of each muscle
→ MFF_{tot}: Average MFF, weighted by total volume of each muscle
→ LMV_{tot}: Sum of all LMV

Functional Correlation Composites
- Cross-sectional analysis
- Functional correlation
- All muscle categories
- Minor SQI are allowed

→ MFI_{tot}
→ MFF_{tot}

*Analyses at 48 weeks. LMV_{tot}=lean muscle volume total; MFF_{tot}=muscle fat fraction total; MFI_{tot}=muscle fat infiltration total; SQI=Signal Quality Issue.

Muscles Included in Functional Correlations and Associated COAs

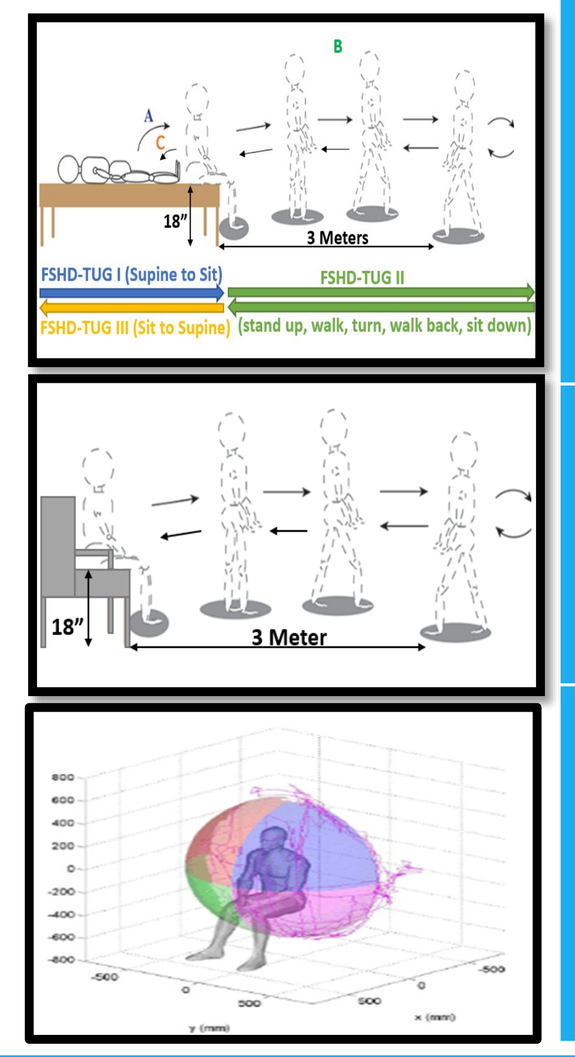
18"
3 Meters
FSHD-TUG I (Supine to Sit)
FSHD-TUG II
FSHD-TUG III (Sit to Supine)
(stand up, walk, turn, walk back, sit down)
3 Meter

Test	Muscles: Upper Extremity	Muscles: Trunk	Muscles: Lower Extremity
FSHD TUG	Supraspinatus Infraspinatus Subscapularis Teres Minor Deltoid Biceps Brachii Triceps Brachii	Pectoralis Major Rhomboids Latissimus Dorsi & Teres Major Trapezius Serratus Anterior Paraspinal (C3-Sacrum)	Quadriceps Hamstrings Adductors Tibialis Anterior Gastrocnemius Medialis
Classic TUG	N/A	N/A	Quadriceps Hamstrings Adductors Tibialis Anterior Gastrocnemius Medialis
RWS	N/A	Pectoralis Major Rhomboids Latissimus Dorsi & Teres Major Trapezius Serratus Anterior	N/A

Results

	Losmapimod N=40	Placebo N=40	Total N=80
Baseline			
Total Number of Evaluable Muscles	1325	1358	2,683
Week 48			
Average Number of 'A' Muscle per subject	15	17	N/A
Average Number of 'B' Muscles per subject	6	7	N/A
Subjects with non-missing values	28	30	58
Muscles with non-missing Values	899	1002	1901
Category A n (%)	423 (47.1)	515 (51.4)	938 (49.3)
Category B n (%)	173 (19.2)	208 (20.8)	381 (20.0)
Category C n (%)	303 (33.7)	279 (27.8)	582 (30.6)

Results – WB-MSK MRI Analysis

Muscle Categorization Captures FSHD Disease Heterogeneity and enables identification of a unique population of muscles at high risk of progression and normal appearing muscles to be followed and assessed for treatment efficacy

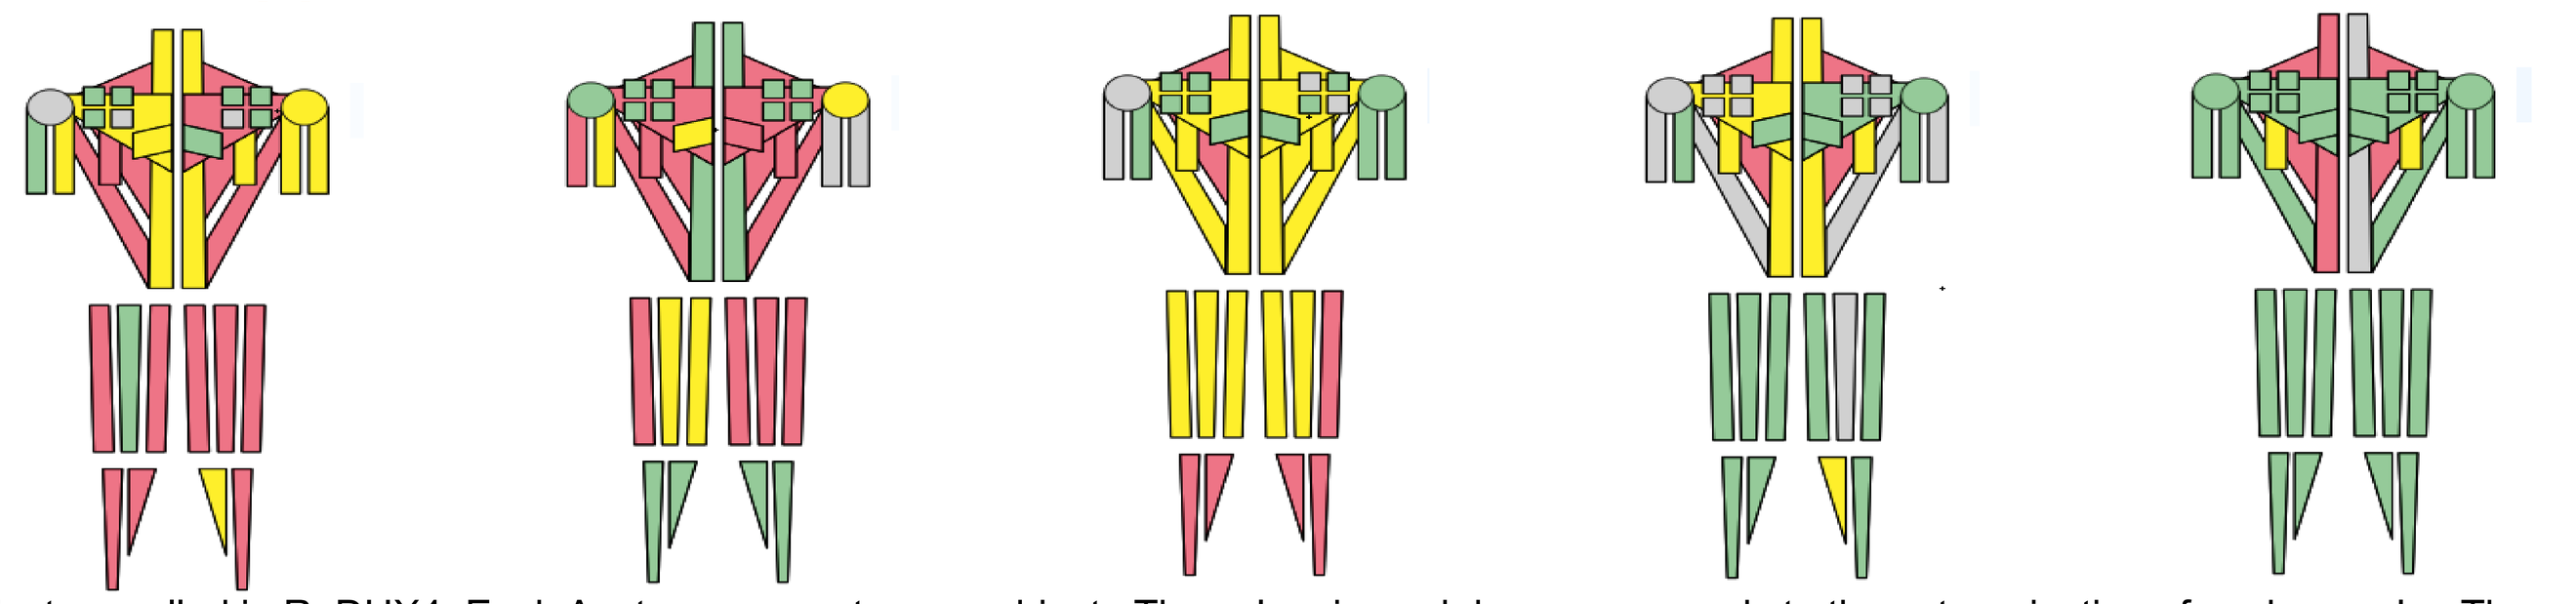

Representation of 5 subjects enrolled in ReDUX4. Each Avatar represents one subject. The colors in each box corresponds to the categorization of each muscle. The pattern of muscle involvement is heterogeneous with the rotator cuff muscles spared, as expected, in most cases, supporting the validity of this WB-MSK MRI quantitative muscle analysis.

Statistical Analysis of Composite Measures

A mixed-effects model for repeated measures (MMRM) was used to analyze the change from baseline in each composite MRI score (MFI_{total}, MFF_{total}, and LMV_{total}), with repeat number category, treatment group, visit, and treatment-by-visit interactions as fixed effects and baseline value of the parameter as a covariate. Within-group LS mean changes from baseline, the associated SEs and 2-sided 95% CIs, treatment differences in LS mean changes from baseline at Weeks 12 and 48 and the associated 2-sided 95% CIs and 2-sided p-values were derived from the MMRM.

Losmapimod Treated Participants Showed Significantly Less Muscle Fat Infiltration (MFI) vs Placebo in Intermediate Muscles

Muscle Fat Infiltration (MFI)

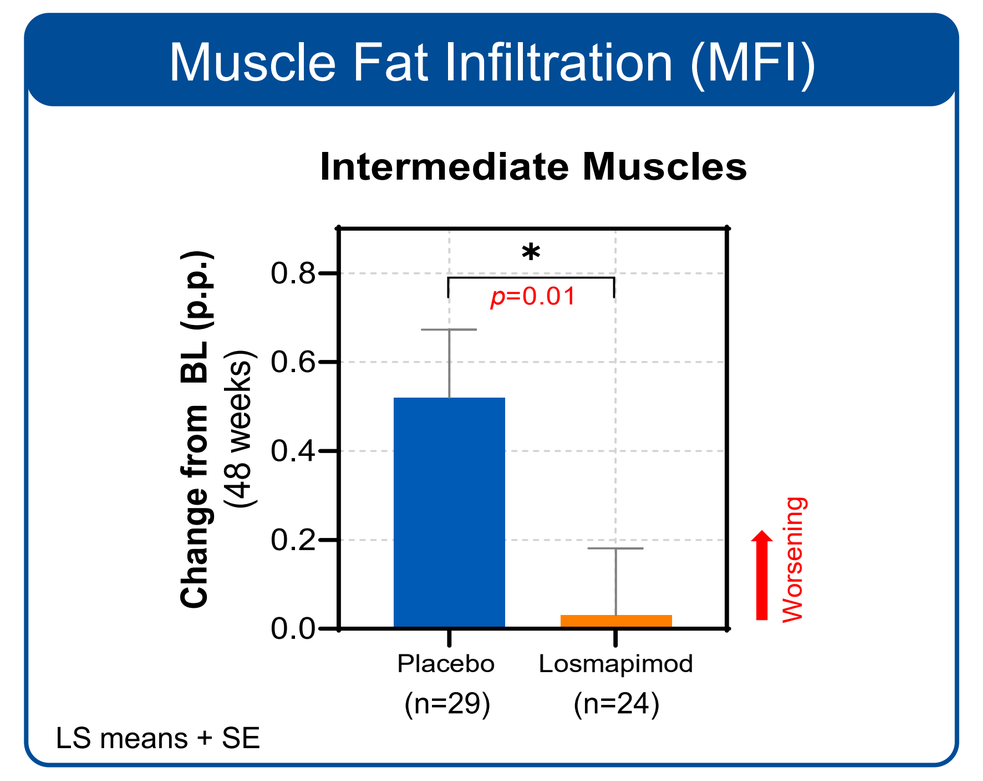
Intermediate Muscles
Change from BL (p.p.) (48 weeks)
*
p=0.01
Worsening
Placebo (n=29)
Losmapimod (n=24)
LS means + SE

Muscle Fat Fraction (MFF)

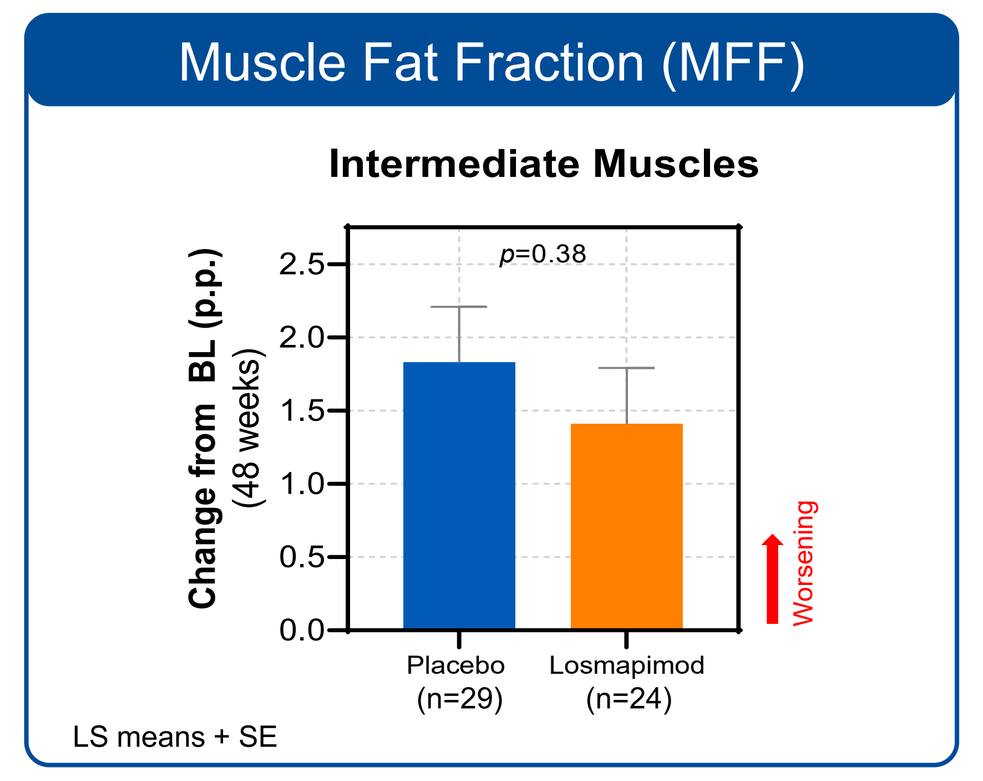
Intermediate Muscles
Change from BL (p.p.) (48 weeks)
p=0.38
Worsening
Placebo (n=29)
Losmapimod (n=24)
LS means + SE

Lean Muscle Volume (LMV)

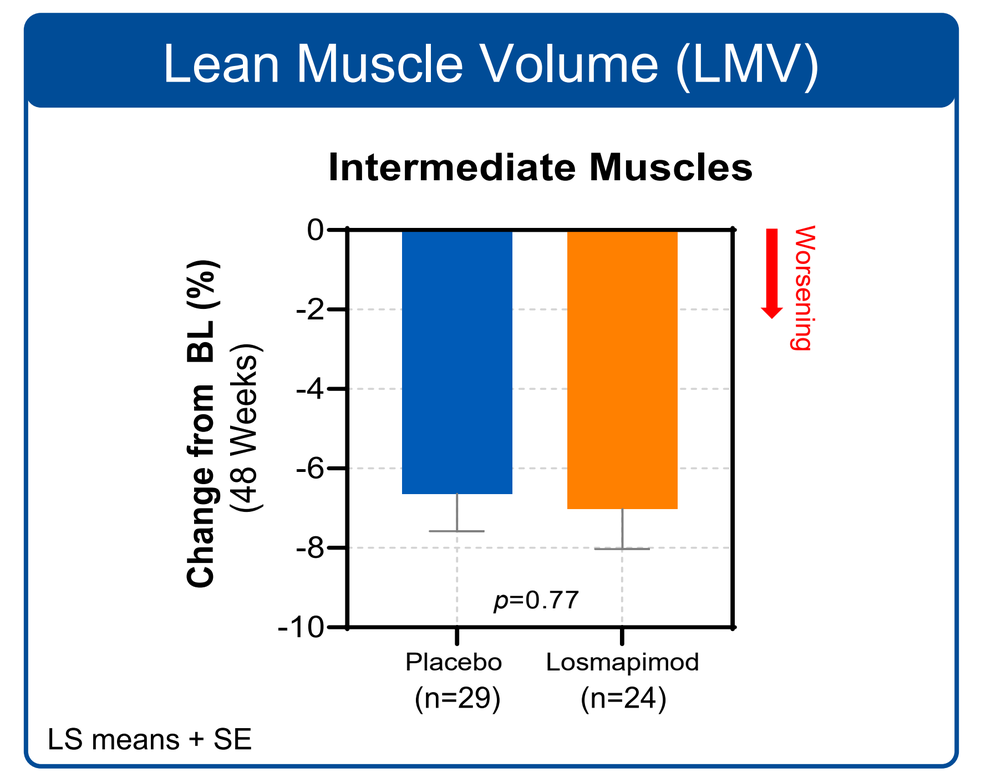
Intermediate Muscles
Change from BL (%) (48 Weeks)
Worsening
p=0.77
Placebo (n=29)
Losmapimod (n=24)
LS means + SE

Normal-Appearing Muscles Appear Preserved With Losmapimod vs Placebo*

Muscle Fat Infiltration (MFI)

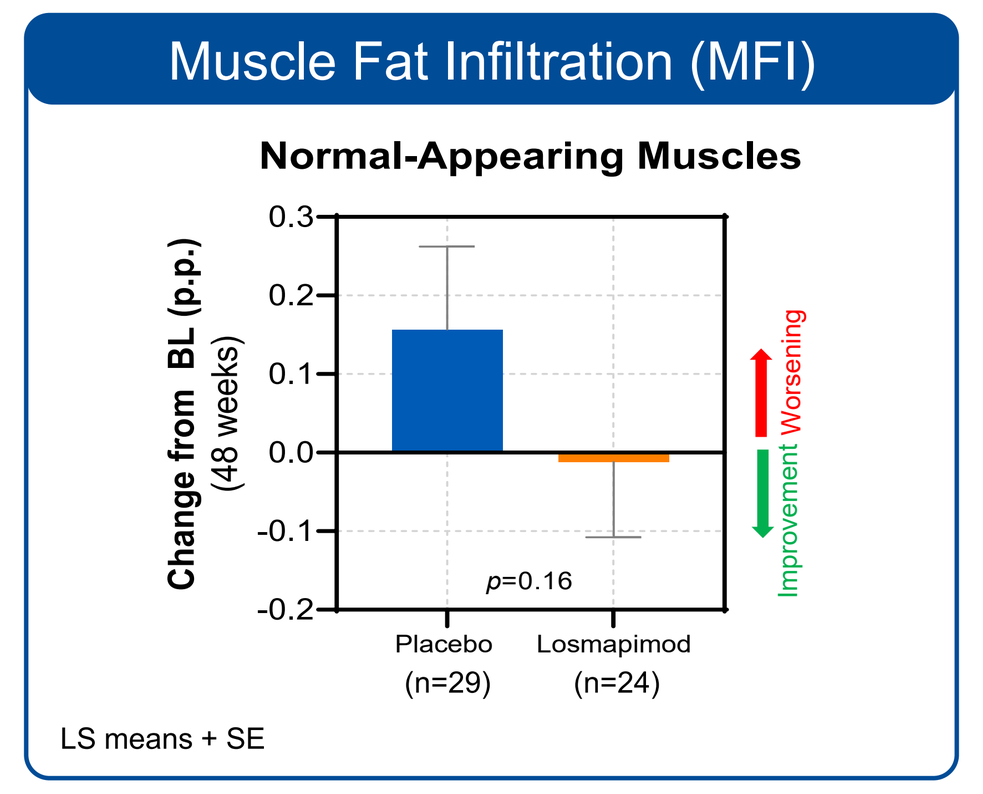
Normal-Appearing Muscles
Change from BL (p.p.) (48 weeks)
p=0.16
Worsening
Improvement
Placebo (n=29)
Losmapimod (n=24)
LS means + SE

Muscle Fat Fraction (MFF)

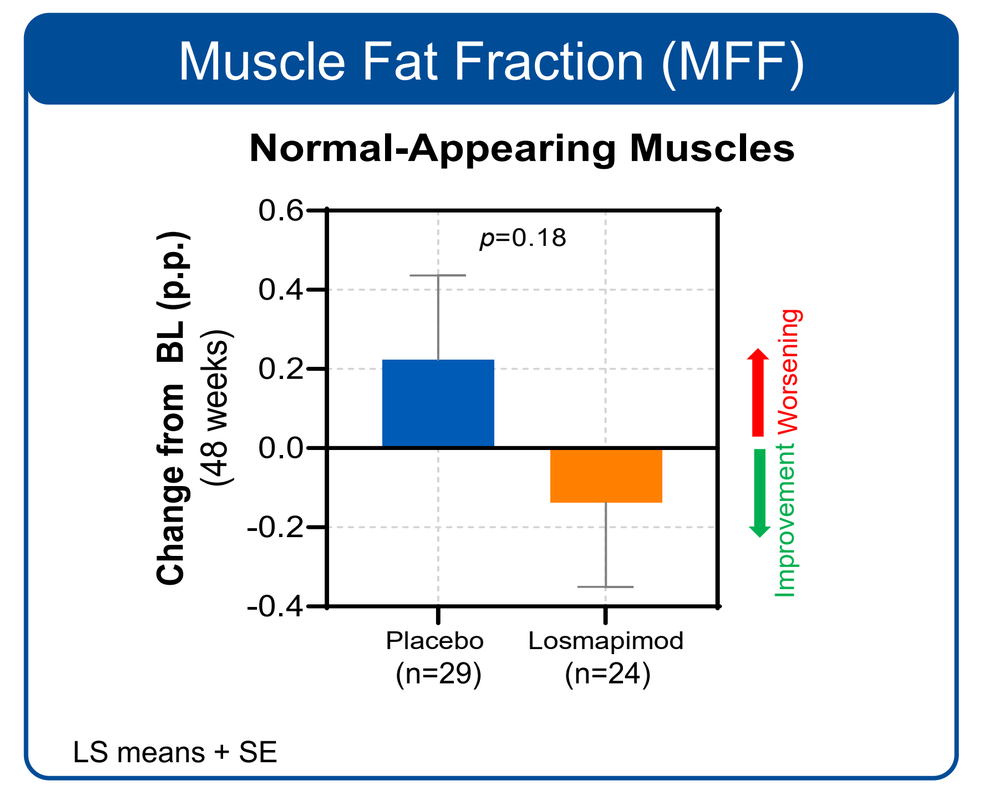
Normal-Appearing Muscles
Change from BL (p.p.) (48 weeks)
p=0.18
Worsening
Improvement
Placebo (n=29)
Losmapimod (n=24)
LS means + SE

Lean Muscle Volume (LMV)

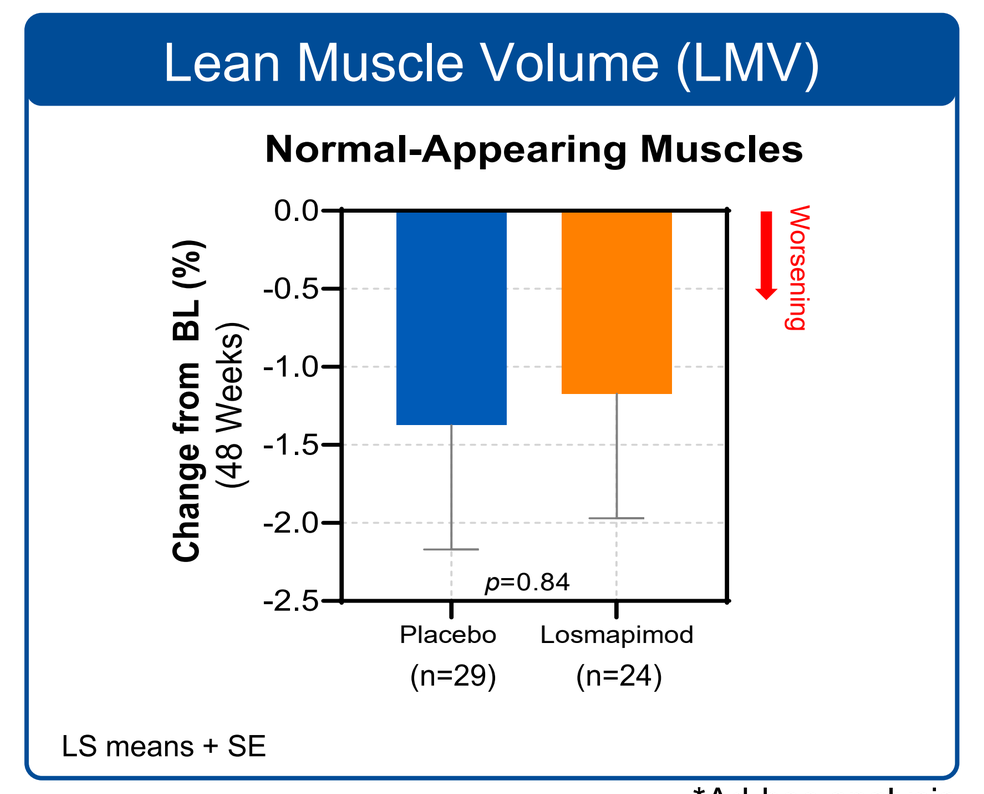
Normal-Appearing Muscles
Change from BL (%) (48 Weeks)
Worsening
p=0.84
Placebo (n=29)
Losmapimod (n=24)
LS means + SE

*Ad-hoc analysis

Moderate and Strong Cross-Sectional Correlations of Regional Composites with COA at week 48 Regardless of Treatment Arm

	MFI		MFF		LMV	
	LOS	**PBO**	**LOS**	**PBO**	**LOS**	**PBO**
Dominant Total RSA with weight p-value N	-0.63 (0.0018) 22	-0.11 (0.5659) 28	-0.66 (0.0008) 22	-0.36 (0.0603) 28	0.74 (0.0001) 22	0.67 (0.0001) 28
Non-Dominant Total RSA with weight p-value N	-0.70 (0.0003) 22	-0.33 (0.0851) 28	-0.77 (<.0001) 22	-0.55 (0.0023) 28	0.62 (0.0023) 22	0.61 (0.0006) 28
TUG p-value N	0.38 (0.0800) 22	0.69 (0.0001) 28	0.48 (0.0229) 22	0.68 (0.0001) 28	-0.42 (0.0492) 22	-0.60 (0.0007) 28
FSHD TUG p-value N	0.60 (0.0054) 20	0.51 (0.0049) 29	0.70 (0.0007) 20	0.70 (<.0001) 29	-0.51 (0.0218) 20	-0.62 (0.0004) 29

RSA= relative surface area

Conclusions

- The WB-MSK MRI quantitative muscle assessment demonstrates a decrease or arrest of fat accumulation in muscles at high risk of progression and normal appearing muscles, suggesting that losmapimod is a potential treatment for FSHD
- After 48 weeks, reduction in the progression of MFI and MFF in A and B muscles suggests losmapimod has an impact on fat accumulation in muscles that have not yet reached end-stage which has been previously reported to have little remaining functional capacity.
 – Detectable MFI changes may be related to the sensitivity of the measure to the earliest detectable pathology of myofiber fat infiltration. Significant changes in MFF and LMV might take more time to be observed.
- WB-MSK-MRI captured the heterogeneity of the disease and provided important information about disease severity as it correlates with FSHD-relevant clinical endpoints.
 – MRI efficacy composites demonstrated sensitivity to disease progression.
 – The clinical relevance of these MRI assessments is supported by the strong cross-sectional correlations with FSHD relevant COAs

References / Acknowledgements

1. Dahlqvist JR. MRI in Neuromuscular Diseases: An Emerging Diagnostic Tool and Biomarker for Prognosis and Efficacy. Ann Neurol 2020, 00:1-13. DOI: 10.1002/ana.25804
2. Rojas LA, Valentine E, Accorsi A, et al. P38α Regulates Expression of DUX4 in a Model of Facioscapulohumeral Muscular Dystrophy. 2019. doi:https://doi.org/10.1101/700195
3. Wagner K. Facioscapulohumeral Muscular Dystrophies. Continuum 2019; 25:1662-1677.

We thank the patients and their families for their participation in this study; the staff at our study sites for their contributions to the care of the patients. We also thank our collaborators and supporting patient organizations for their support.